CRN04777 an Oral, Nonpeptide SST5-Selective Somatostatin Agonist Dose Dependently Suppresses Basal and Stimulated Insulin Secretion

Christine T. Ferrara-Cook[1], Rosa Luo[1], Eduardo De la Torre[1], Yang Wang[1], Stephen F. Betz[1], Ajay Madan[1], R. Scott Struthers[1], Ulrike Hövelmann[2], Tim Heise[2], Alan Krasner[1]
[1] Crinetics Pharmaceuticals®, San Diego, USA; [2] Profil, Neuss, Germany

PSUN304

BACKGROUND

- Pathophysiology of congenital hyperinsulinism (HI)
 - Most common cause of hypoglycemia in neonates, infants and children
 - Mutations within the pancreatic beta-cell result in excess insulin secretion
 - Current treatments are limited, highly burdensome, and not effective in all patients
 - SST5 receptor activation inhibits insulin secretion downstream of known beta-cell mutations
 - CRN04777 acting at the SST5 receptor is the only oral therapy with the potential to suppress insulin secretion in patients with congenital HI regardless of the genetic cause

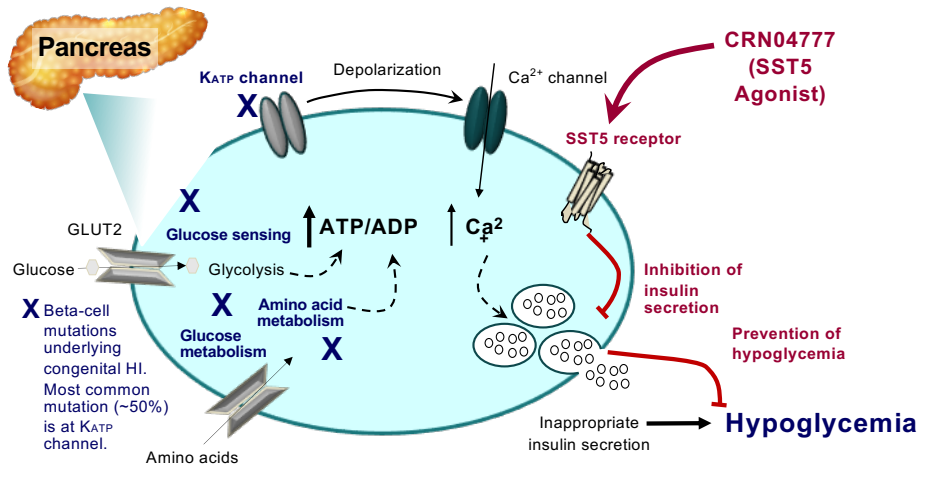
Pancreas
KATP channel
Depolarization
Ca2+ channel
CRN04777 (SST5 Agonist)
SST5 receptor
X
Glucose sensing
ATP/ADP
Ca2+
GLUT2
Glucose
Glycolysis
Amino acid metabolism
Glucose metabolism
Inhibition of insulin secretion
Prevention of hypoglycemia
X Beta-cell mutations underlying congenital HI. Most common mutation (~50%) is at KATP channel.
Amino acids
Inappropriate insulin secretion
Hypoglycemia

METHODS

- CRN04777 oral dosing in 80 healthy volunteers
 - Single ascending (0.5-120 mg; 6 active: 2 Pbo)
 - Multiple ascending (30-120 mg x10d; 6 active: 3 Pbo)
- Evaluate safety, tolerability, and pharmacokinetics
- Pharmacodynamic assessments
 - Intravenous glucose tolerance test (IVGTT)
 - Sulfonylurea (SU) challenge

AUC: area under the curve, GIR: glucose infusion rate, HI: hyperinsulinism, HV: healthy volunteer, m: number if occurrences, MAD: multiple ascending dose, n: incidence, Pbo: placebo, PG: plasma glucose, PK: pharmacokinetics, SAD: single ascending dose, SST5: somatostatin subtype 5, SU: sulfonylurea, t_{max}: time to maximum plasma concentration, $t_{1/2}$: half-life.

RESULTS

SAFETY

Single and multiple doses up to 120mg are well tolerated with no discontinuations or modifications

	Placebo N=29 n (%) m	Total 4777 (SAD + MAD) N=78 n (%) m
Nausea	0	15 (19.2) 17
Vomiting	0	7 (9.0) 9
Diarrhea	0	5 (6.4) 5
Headache	0	5 (6.4) 5
Chills	0	3 (3.8) 3
Hypoglycemia	0	3 (3.8) 3
Abdominal pain	0	2 (2.6) 2
Nasopharyngitis	0	2 (2.6) 2
Phlebitis	4 (13.8) 4	1 (1.3) 1
Skin irritation	2 (6.9) 2	1 (1.3) 1

Table 1: Treatment-Emergent Adverse Events by Preferred Term, n=incidence, m=events; GI AEs followed soon after treatment initiation, and resolved without drug discontinuation

PHARMACOKINETICS

- Rapid oral absorption(t_{max} 1-3 hours)
- $t_{1/2}$ of ~40 hours supports once daily dosing

Single Dose (SAD; Day 1)

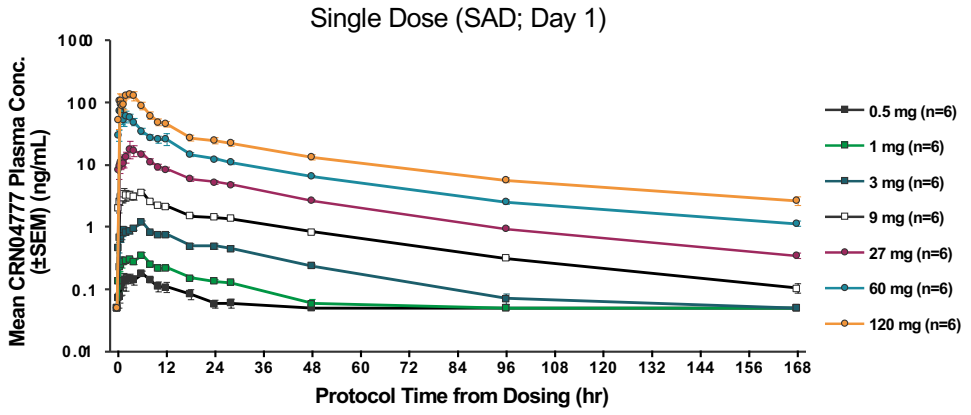
Mean CRN04777 Plasma Conc. (±SEM) (ng/mL)
Protocol Time from Dosing (hr)
0.5 mg (n=6)
1 mg (n=6)
3 mg (n=6)
9 mg (n=6)
27 mg (n=6)
60 mg (n=6)
120 mg (n=6)

Steady State (MAD; Day 9)

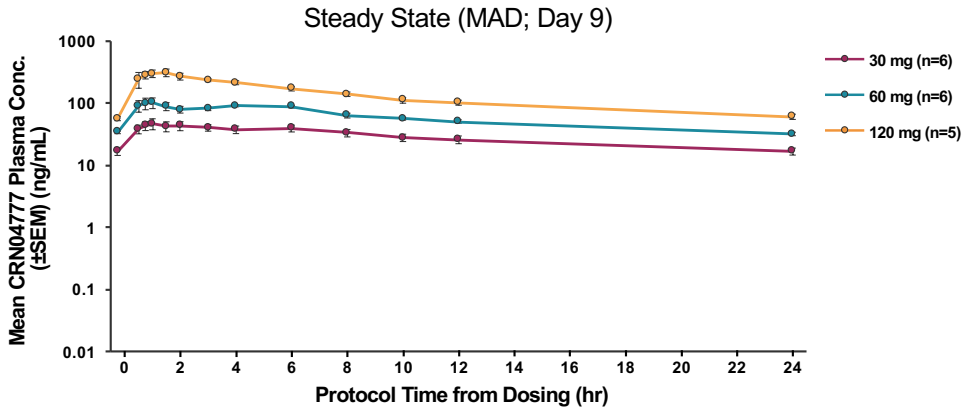
Mean CRN04777 Plasma Conc. (±SEM) (ng/mL)
Protocol Time from Dosing (hr)
30 mg (n=6)
60 mg (n=6)
120 mg (n=5)

Figure 1. Concentration time profiles from after a single dose and at steady state.

INTRAVENOUS GLUCOSE TOLERANCE TEST

CRN04777 dose-dependently suppressed glucose stimulated insulin secretion

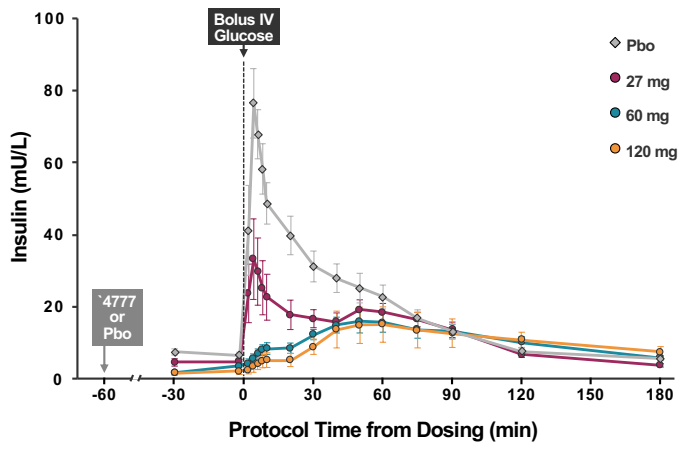
Bolus IV Glucose
Insulin (mU/L)
`4777 or Pbo
Pbo
27 mg
60 mg
120 mg
Protocol Time from Dosing (min)

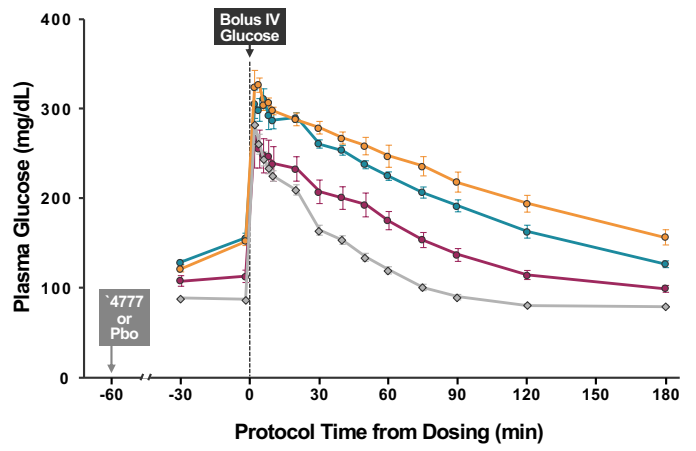
Bolus IV Glucose
Plasma Glucose (mg/dL)
`4777 or Pbo
Protocol Time from Dosing (min)

Figure 2: Insulin and glucose data measured during intravenous glucose tolerance test (IVGTT) in single dose cohorts. IV glucose bolus 300mg/kg was administered 1 hour after CRN04777 dosing. Glucose stimulated insulin secretion during the IVGTT (plasma insulin AUC) was reduced dose-dependently, to a maximum of approximately 50% with a parallel doubling of plasma glucose AUC following 120 mg of CRN04777. Data represent mean ± SEM

SULFONYLUREA CHALLENGE: PHARMACOLOGIC MODEL OF CONGENITAL HI

SST5 agonism by CRN04777 dose-dependently reversed sulfonylurea induced insulin secretion

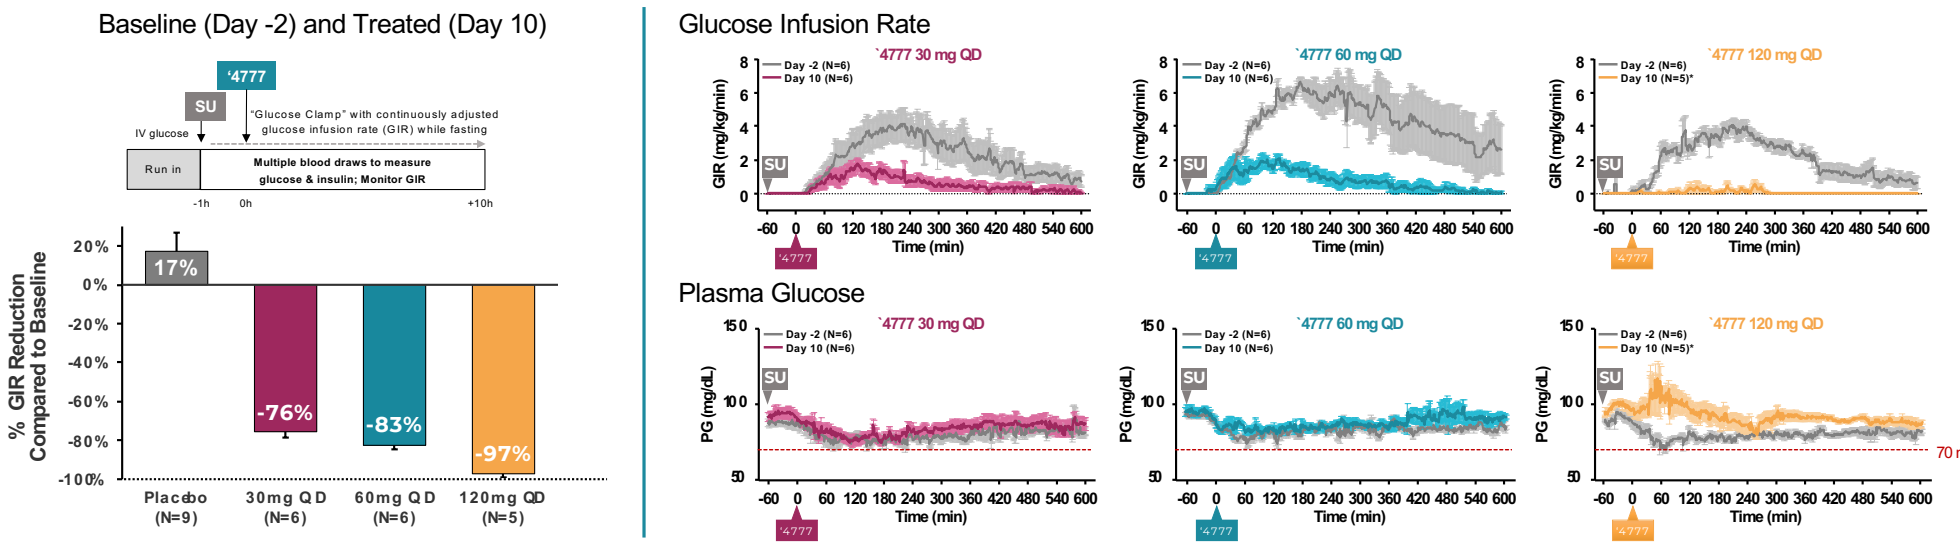
Baseline (Day -2) and Treated (Day 10)
'4777
SU
IV glucose
"Glucose Clamp" with continuously adjusted glucose infusion rate (GIR) while fasting
Run in
Multiple blood draws to measure glucose & insulin; Monitor GIR
-1h
0h
+10h
% GIR Reduction Compared to Baseline
17%
-76%
-83%
-97%
Placebo (N=9)
30mg QD (N=6)
60mg QD (N=6)
120mg QD (N=5)
Glucose Infusion Rate
`4777 30 mg QD
`4777 60 mg QD
`4777 120 mg QD
Day -2 (N=6)
Day 10 (N=6)
Day 10 (N=5)*
GIR (mg/kg/min)
Time (min)
Plasma Glucose
PG (mg/dL)
70 mg/dL

Figure 3. SU challenge (10mg glibenclamide/glyburide) was conducted in multiple ascending dose cohorts at baseline and Day 10 under automated euglycemic clamp conditions (ClampArt®). CRN04777 was administered one hour after oral SU administration, followed by measurement of IV glucose infusion rate (GIR) and plasma glucose (PG) over 10 hours; Solid line: mean, shaded: SEM. Glucose infusion rate over clamp duration (total AUC_{GIR}) on Day 10 compared to baseline showed dose-dependent reductions. Data represent mean ± SEM.

CONCLUSION

Crinetics Pharmaceuticals has developed oral SST5 agonist that suppresses insulin secretion in basal and stimulated conditions. Given the mechanism of action, CRN04777 has the potential to be universally effective in all patients with congenital HI.

Crinetics® Pharmaceuticals