Potent Parathyroid Hormone 1 Receptor Antagonists Suppress Receptor Activation and Downstream Pathways in Human Primary Osteoblasts

Jonathan Shintaku, PhD; Elizabeth Rico-Bautista, PhD; Joseph Pontillo, PhD; Shimiao Wang, MS; Pablo Arroyo, MS; Anel A. Castellanos Gonzalez, BS; Julie B Nguyen, BS; Kelsey Retting, PhD; Beth Fleck, MS; Sameer Urgaonkar, PhD; Stephen F. Betz, PhD; Stacy Markison, PhD
Crinetics Pharmaceuticals, Inc., San Diego, CA, USA

SAT-539

BACKGROUND AND OBJECTIVES

- Parathyroid hormone (PTH) regulates calcium homeostasis by direct and indirect targeting of bone, kidney, and intestine. In bone, PTH stimulates cAMP generation and protein kinase A (PKA) signaling, which alter expression of genes such as *RANKL* (encoding receptor activator of nuclear factor kappa B ligand) and *OPG* (encoding osteoprotegerin). RANKL secretion from osteoblasts activates osteoclasts to promote bone resorption and calcium release from bone.

- Primary hyperparathyroidism (PHPT) is diagnosed in about 100,000 people in the United States every year and is characterized by over-secretion of PTH, usually due to parathyroid adenomas or hyperplasia. Medical intervention seeks to normalize blood calcium levels, reduce bone resorption to lower the risk of bone fractures, and reduce the risk of kidney stones.

- While the first line therapy of partial to total parathyroidectomy is sufficient to treat most individuals, surgery is either declined or insufficient in 10-15% of symptomatic PHPT patients. Alternatives to surgery include calcimimetics which reduce circulating calcium without an effect on bone mineral density (BMD), or bisphosphonates which can improve BMD but do not improve circulating calcium.

- We hypothesize that blocking PTH action via a PTH 1 receptor (PTH1R) antagonist may improve the current therapeutic landscape for treating PHPT by concurrently treating both BMD and circulating calcium.

Primary hyperparathyroidism indirectly promotes bone loss via osteoblast secretion of RANKL

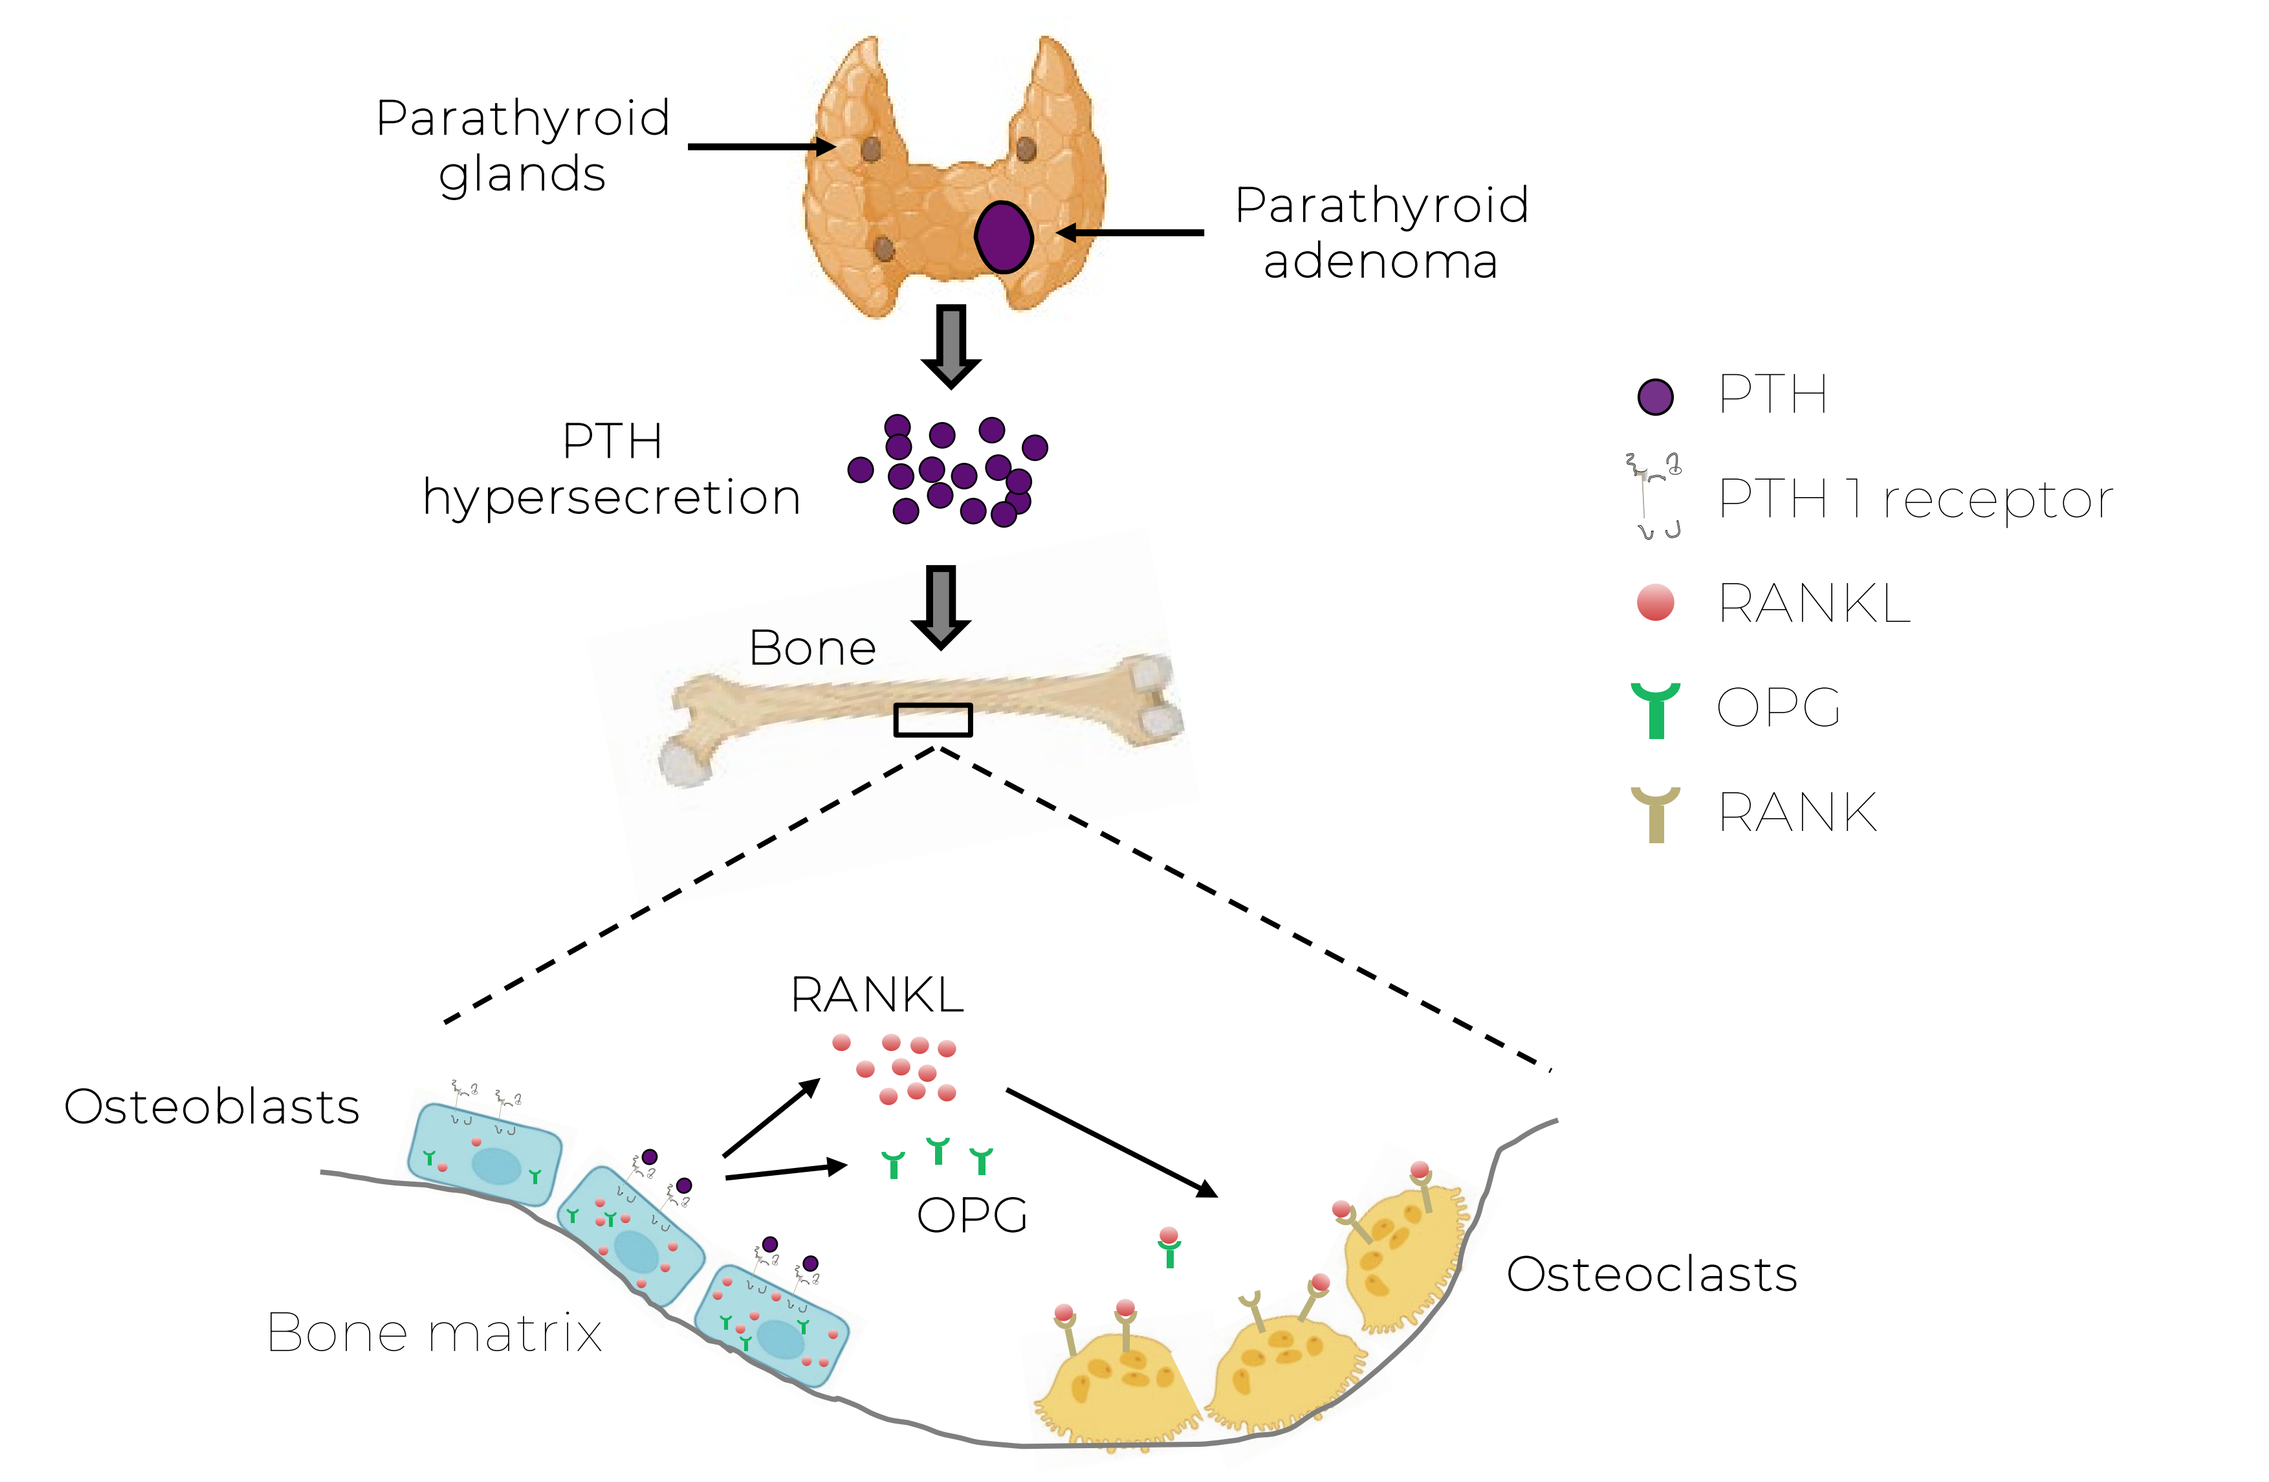
Parathyroid glands
Parathyroid adenoma
PTH hypersecretion
Bone
PTH
PTH 1 receptor
RANKL
OPG
RANK
Osteoblasts
RANKL
OPG
Bone matrix
Osteoclasts

- PTH over-secretion due to a parathyroid gland adenoma hyperactivates PTH1R on osteoblasts
- PTH stimulates osteoblast expression of RANKL and increases the ratio of expressed RANKL/OPG
- RANKL promotes osteoclast formation, activation, and survival, which increases bone resorption
- OPG is a decoy receptor for RANKL, inhibiting RANKL binding to the RANK receptor
- Primary hyperparathyroidism results in hypercalcemia and loss of bone tissue

STUDY DESIGN

- Crinetics used an iterative medicinal chemistry approach to identify potent nonpeptide PTH1R antagonists
- In CHO-K1 cells with heterologous expression of human PTH1R, novel compounds demonstrated good potency in a functional antagonist assay (IC_{50}<100 nM)
- We utilized normal human primary osteoblasts to investigate the therapeutic potential of antagonists to block PTH1R activation and downstream effectors mediating PHPT in bone

RESULTS

Maturation of primary osteoblasts in differentiation conditions

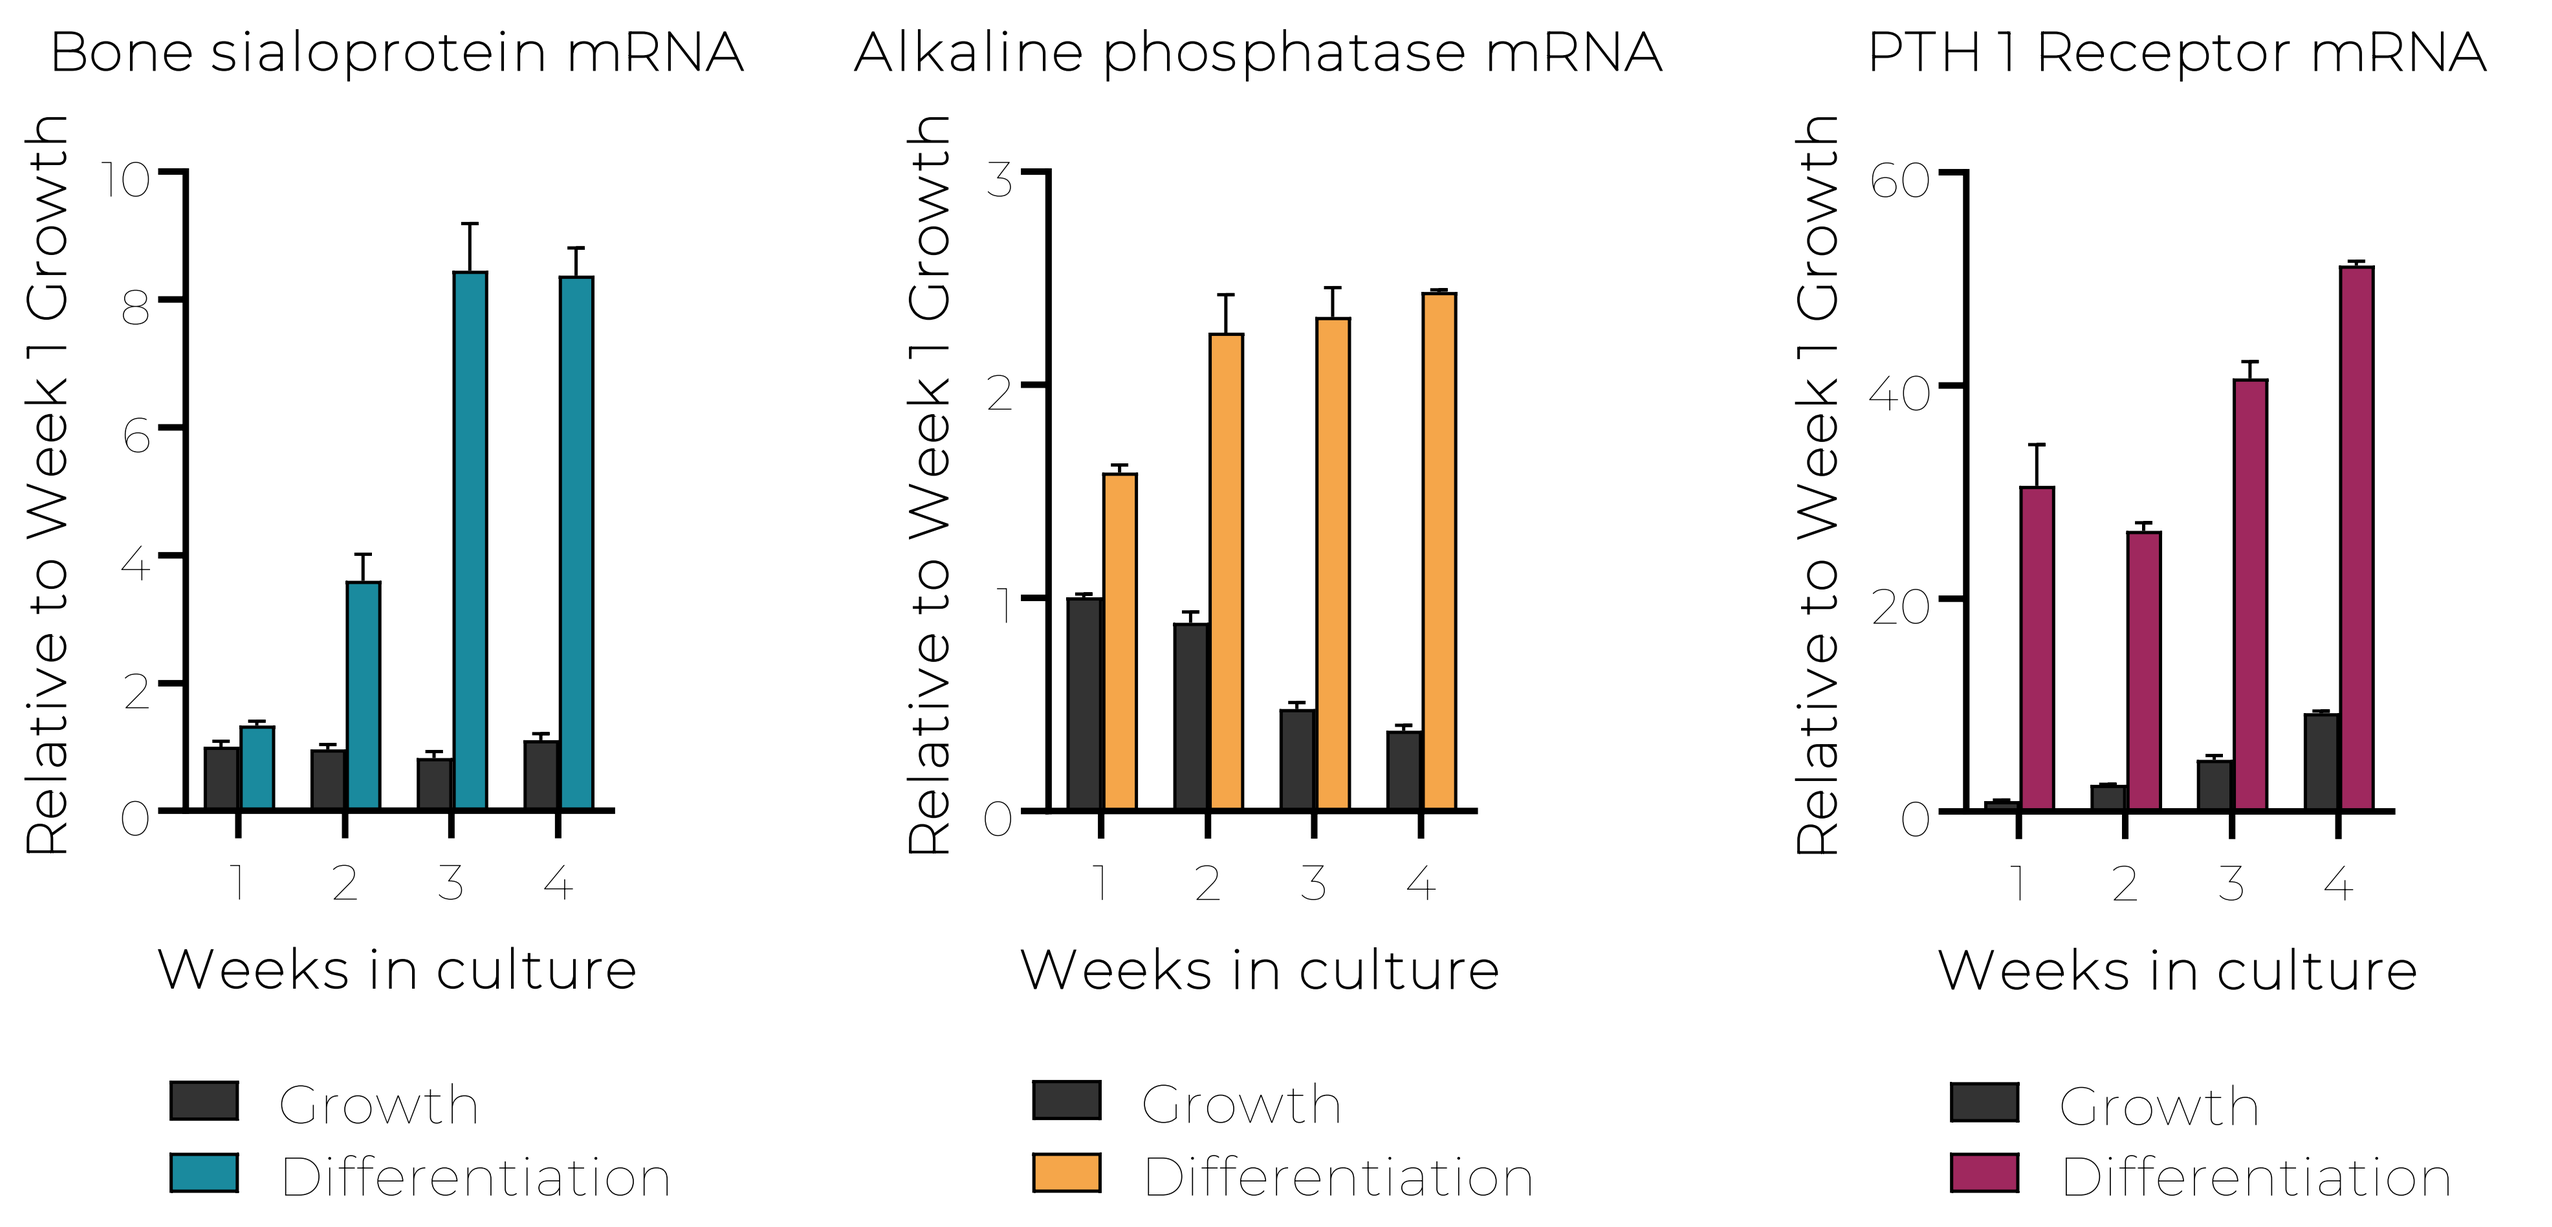
Bone sialoprotein mRNA
Alkaline phosphatase mRNA
PTH 1 Receptor mRNA
Relative to Week 1 Growth
Weeks in culture
Growth
Differentiation

Figure 1. Primary human osteoblasts increase mRNA expression of genes indicative of mature osteoblasts. Normal human primary osteoblasts (donor 1), were obtained from Lonza (#CC-2538) and cultured in the vendor-recommended Growth Media. Differentiation media included Growth Media supplemented with 10 mM β-glycerophosphate and 400 nM hydrocortisone-21-hemisuccinate. Expression of mRNA was measured with real-time RT-PCR and normalized to β-actin. Bars represent mean±SEM. n=3 technical replicates.

Differentiated osteoblasts demonstrate a cAMP response to PTH

A

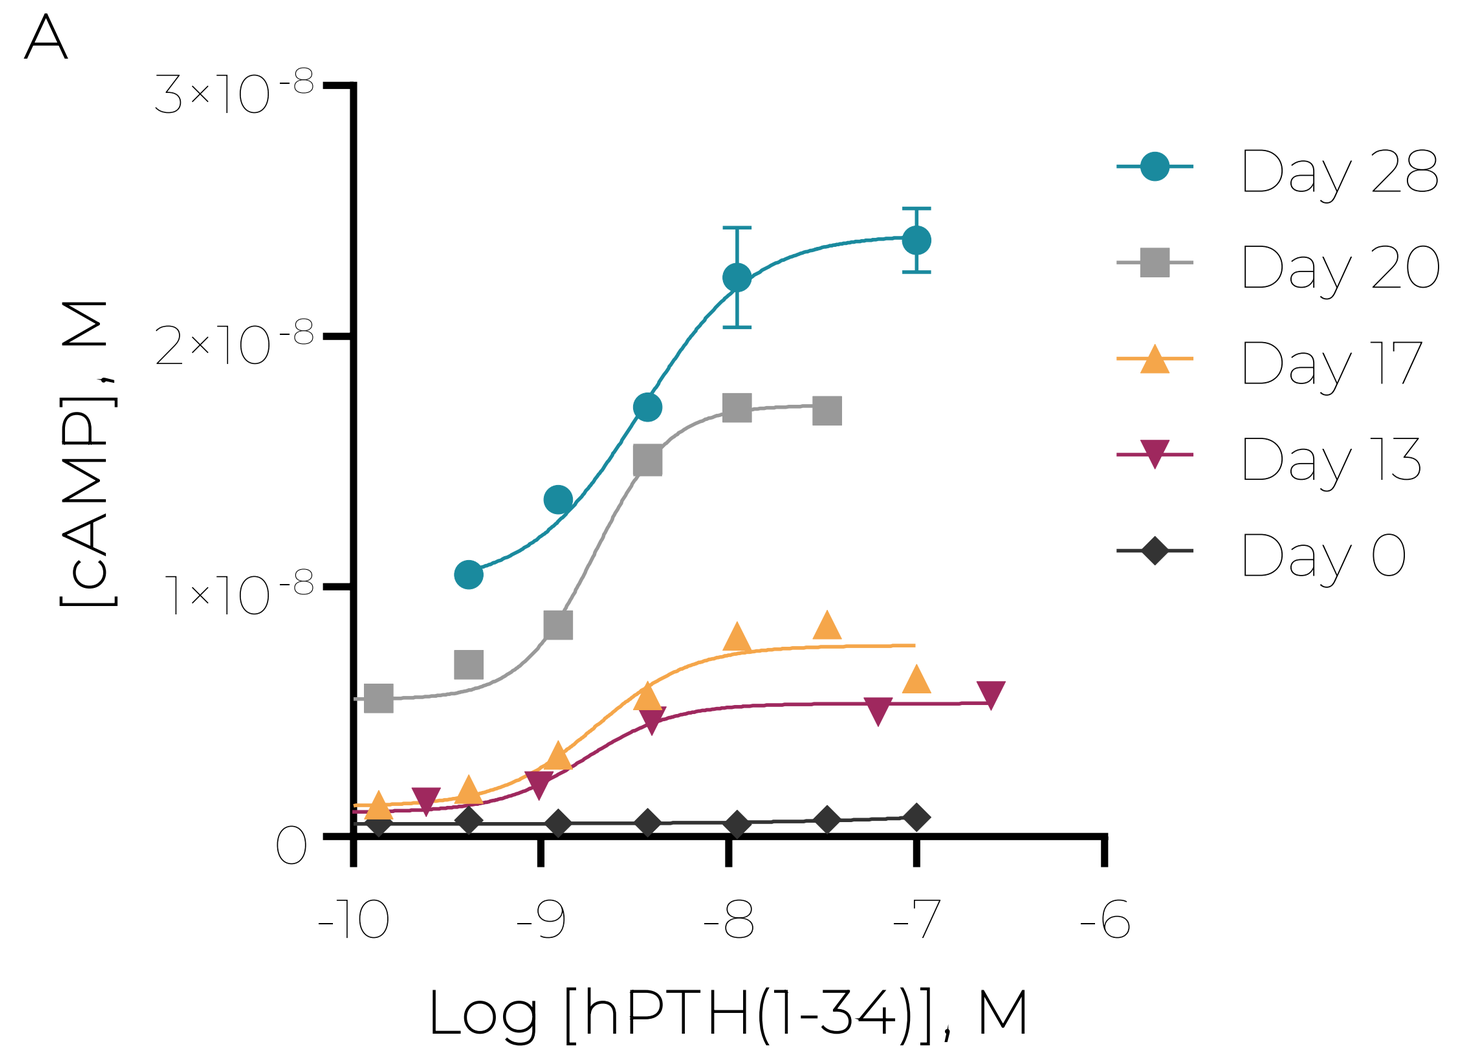
[cAMP], M
Log [hPTH(1-34)], M
Day 28
Day 20
Day 17
Day 13
Day 0

B

Day	EC_{50} (nM)	Dynamic range (nM)
0	-	0.5 – 0.7
13	1.7	0.9 – 5.6
17	2.0	1.2 – 7.6
20	1.8	5.5 – 17
28	3.4	10 - 24

Figure 2. cAMP accumulation, in response to PTH1R stimulation, was more robust with osteoblast differentiation. (A) cAMP concentrations were measured (Cisbio, #62AM4PEB) from differentiating osteoblasts stimulated for 1 hour with hPTH(1-34). Points represent mean±SEM. n=3 technical replicates. (B) EC_{50} and cAMP dynamic range were calculated from hPTH(1-34) agonist curves.

RESULTS (cont.)

Primary osteoblasts from multiple donors respond to PTH

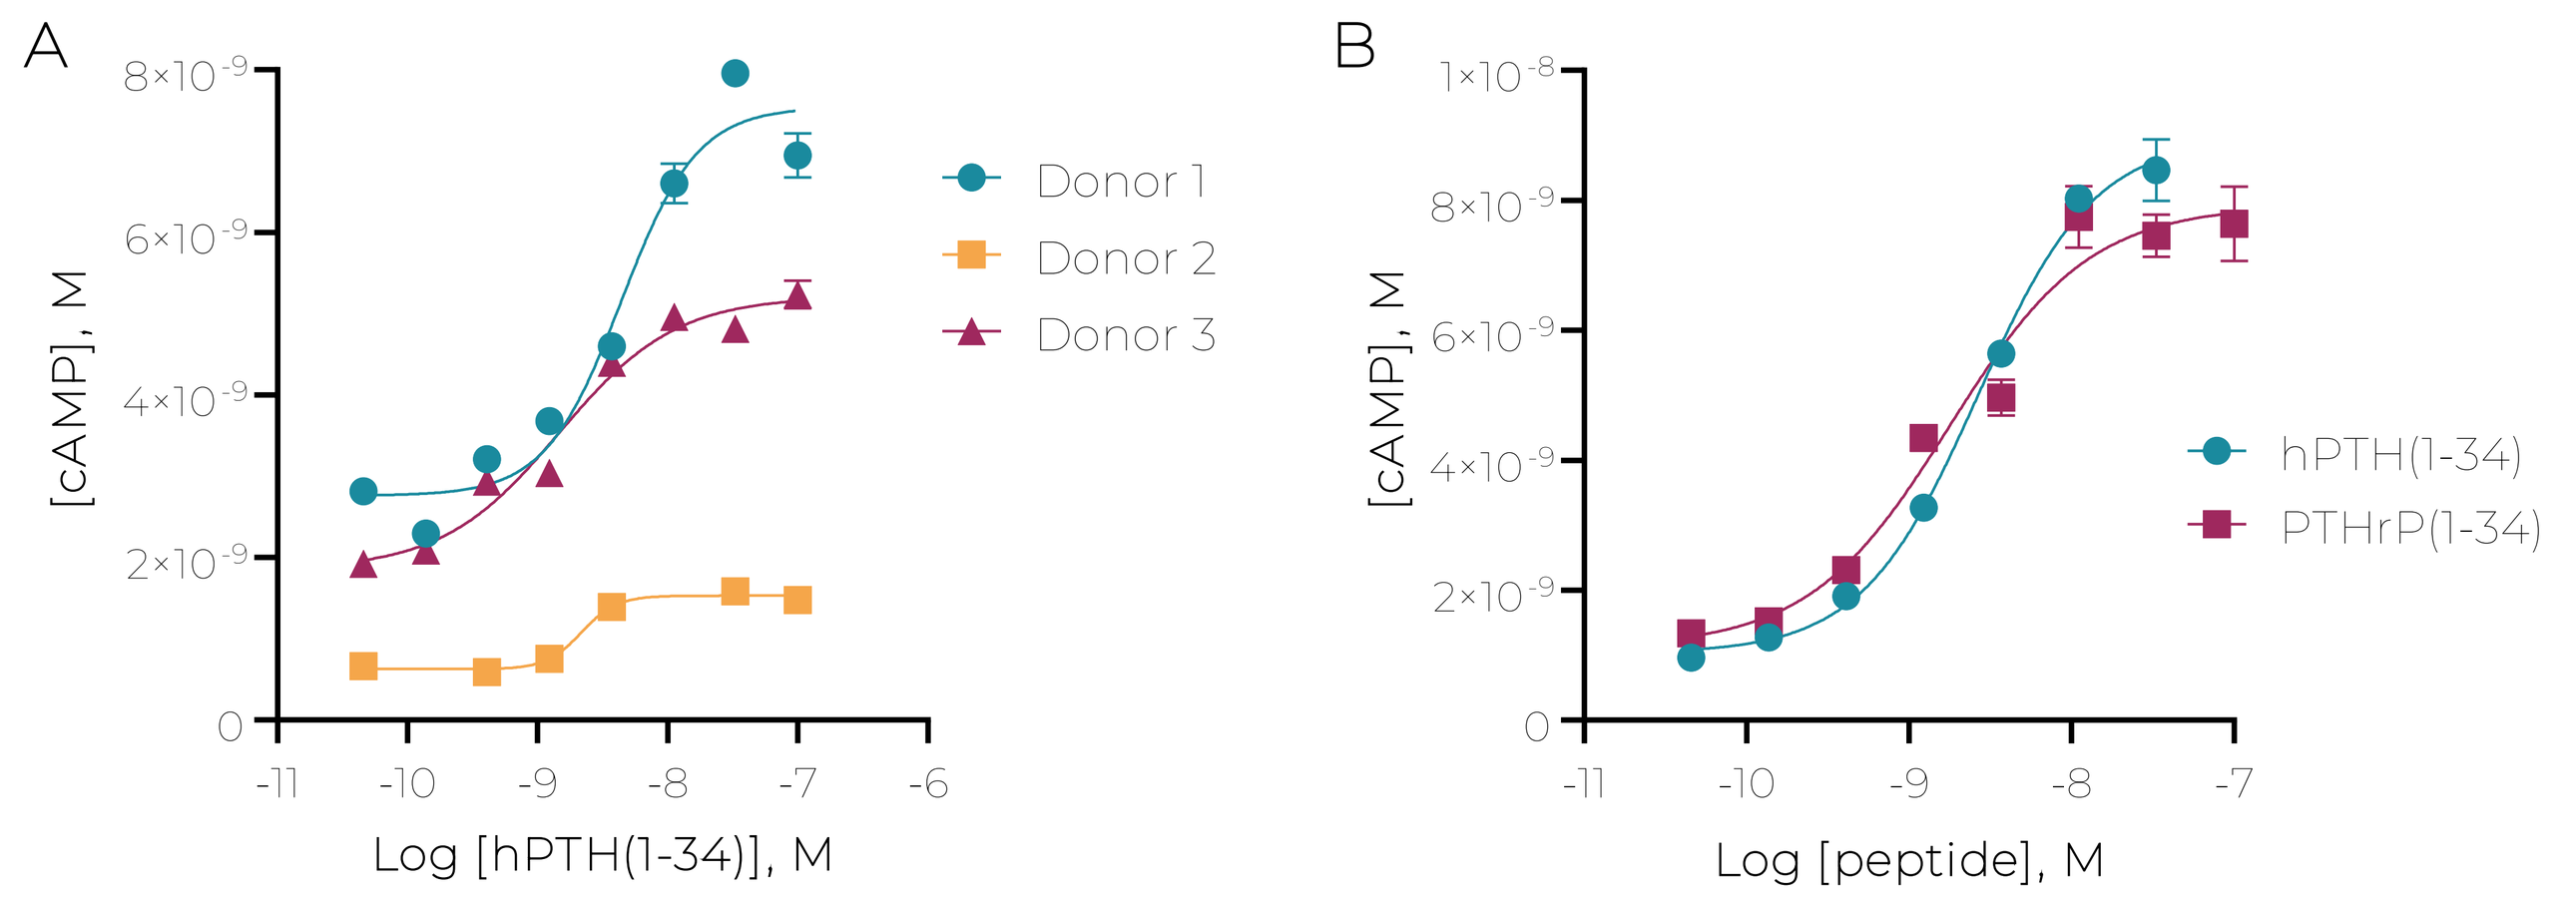
A
[cAMP], M
Log [hPTH(1-34)], M
Donor 1
Donor 2
Donor 3
B
[cAMP], M
Log [peptide], M
hPTH(1-34)
PTHrP(1-34)

Donor	1	2	3
EC_{50} (nM)	4.4	2.2	1.5

Peptide	hPTH(1-34)	PTHrP(1-34)
EC_{50} (nM)	2.7	1.8

Figure 3. cAMP accumulation in human primary osteoblasts in response to hPTH(1-34) and PTHrP(1-34). (A) Primary osteoblasts from Lonza (#CC-2538, donors 1 and 3) and Millipore Sigma (#406-05A, donor 2) were cultured 21 days in differentiation media. PTH1R activation was evaluated as described in Figure 2. (B) Donor 1 osteoblasts responded similarly to hPTH(1-34) and PTHrP(1-34).

PTH increases *RANKL* and *OPG* mRNA expression in primary osteoblasts

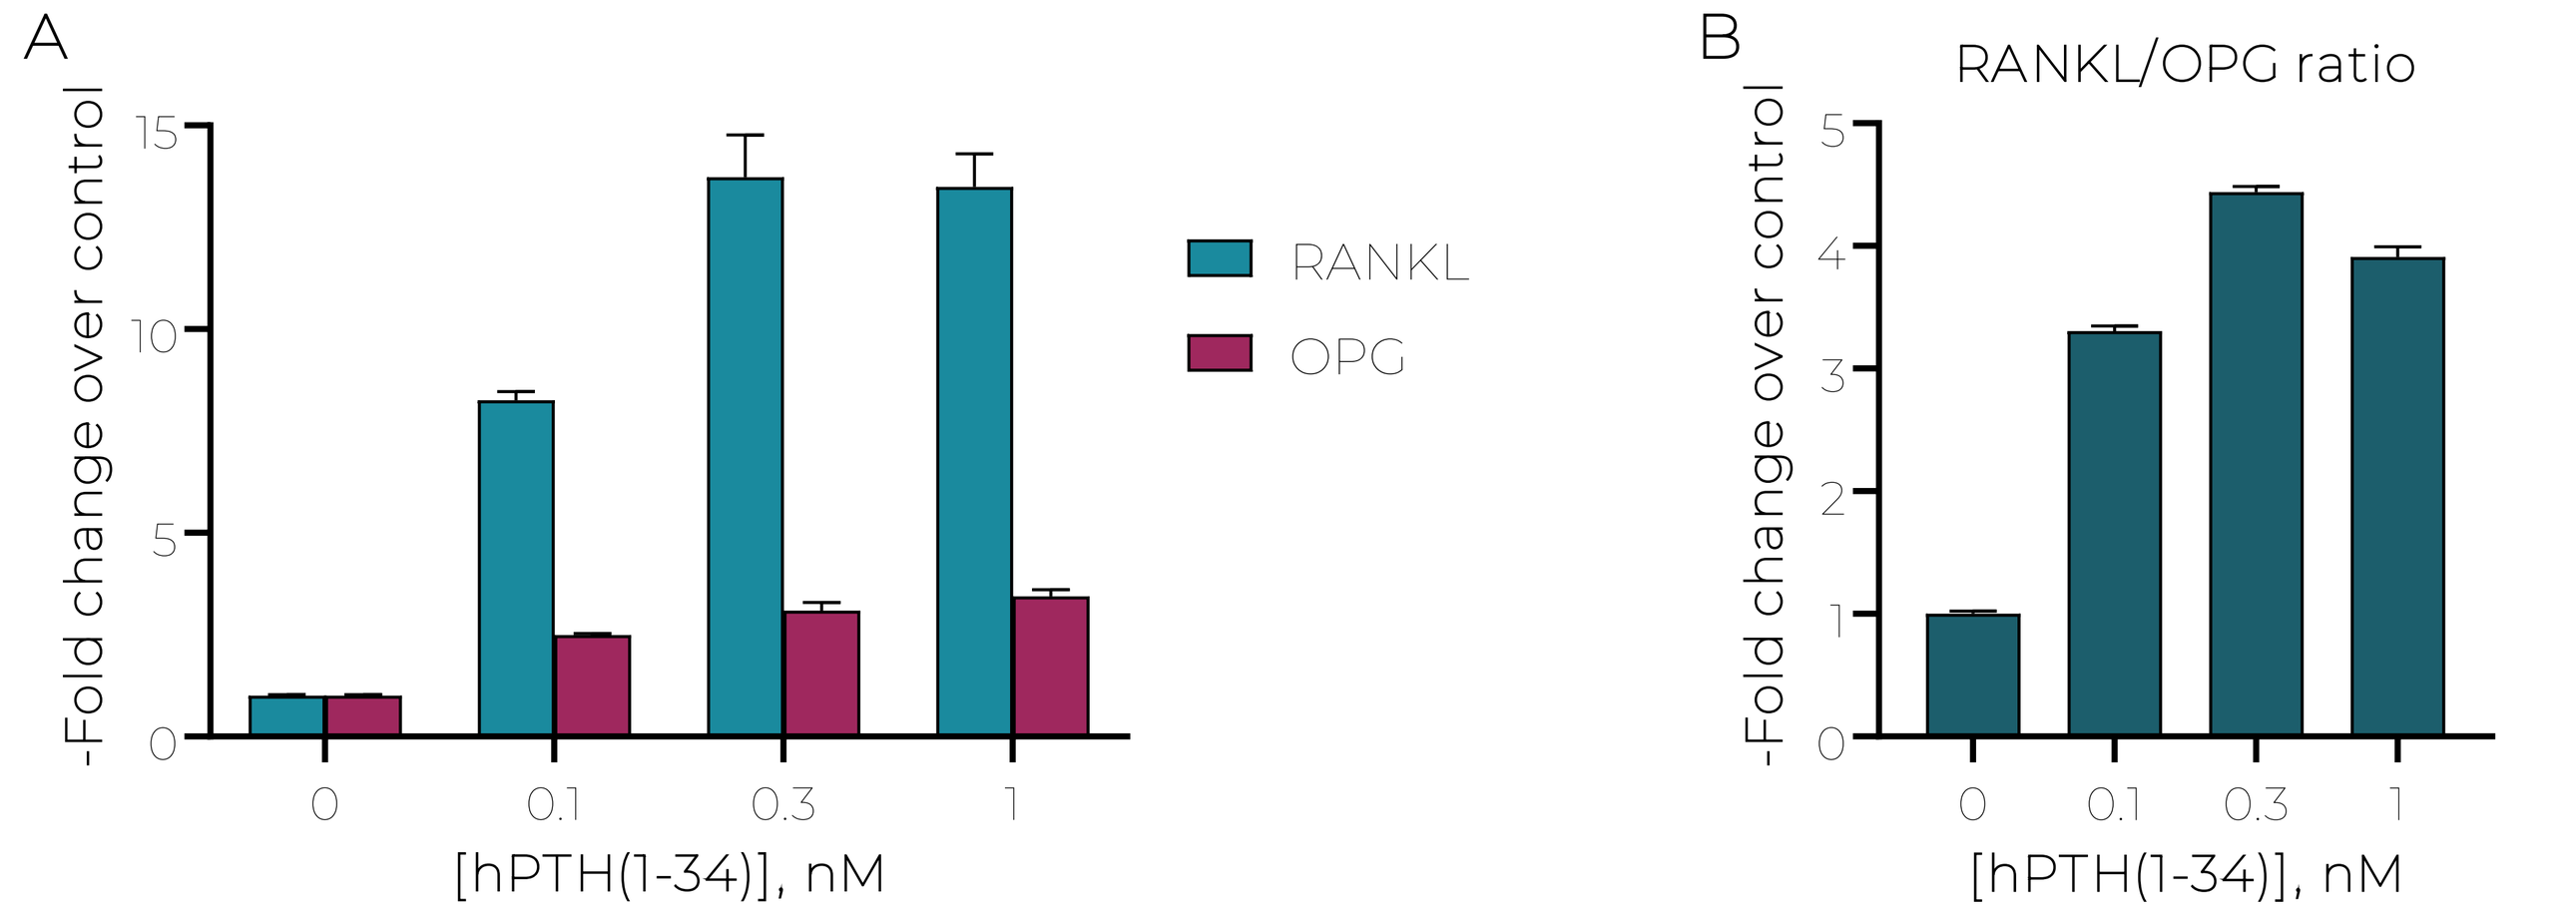
A
-Fold change over control
[hPTH(1-34)], nM
RANKL
OPG
B
RANKL/OPG ratio
-Fold change over control
[hPTH(1-34)], nM

Figure 4. Primary osteoblasts demonstrate hPTH(1-34) concentration-dependent increases in *RANKL*, *OPG*, and *RANKL/OPG* ratio. Osteoblasts differentiated 28 days were treated 8 hours with hPTH(1-34). Relative mRNA was normalized to *β-actin*. (A) *RANKL* mRNA and *OPG* mRNA relative to vehicle control. Bars represent mean±SEM. n=3 technical replicates. (B) *RANKL/OPG* mRNA ratio was calculated from (A).

Novel antagonists block PTH1R activation in primary osteoblasts

A

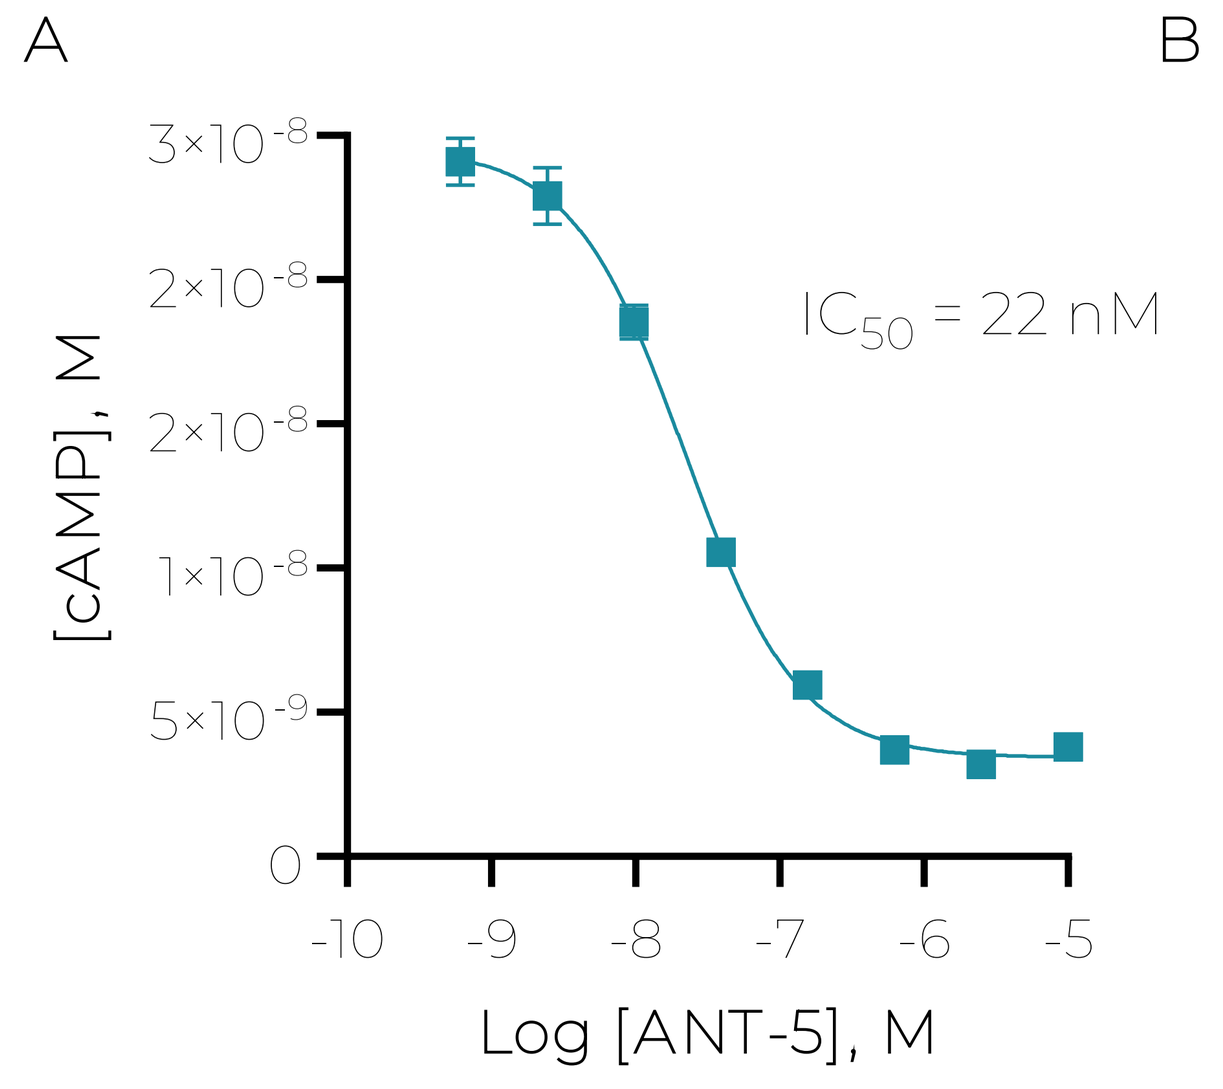
[cAMP], M
IC_{50} = 22 nM
Log [ANT-5], M

B

Antagonist	Osteoblasts Day 14 IC_{50} (nM)	CHO-K1-hPTH1R IC_{50} (nM)
ANT-5	26	5
ANT-6	51	4
ANT-7	54	10
ANT-8	17	6
ANT-9	19	26
ANT-10	8	29
ANT-11	3	9
ANT-12	79	10
ANT-13	162	27
ANT-14	32	12

Figure 5. hPTH(1-34)-dependent cAMP accumulation in primary osteoblasts is inhibited by novel PTH1R antagonists. (A) Osteoblasts differentiated 14 days were stimulated with 3.5 nM hPTH(1-34) (~EC_{80}) and ANT-5 for 1 hour, followed by measurement of cAMP. Graph is a representative experiment from mean values presented in (B). Points represent mean±SEM. n=3 technical replicates. (B) PTH1R antagonist potencies are presented as means of 1-5 independent experiments in primary osteoblasts differentiated 14 days or CHO-K1 cells with heterologous expression of human PTH1R.

RESULTS (cont.)

ANT-5 has no cytotoxic effects on primary osteoblasts after 24 hours

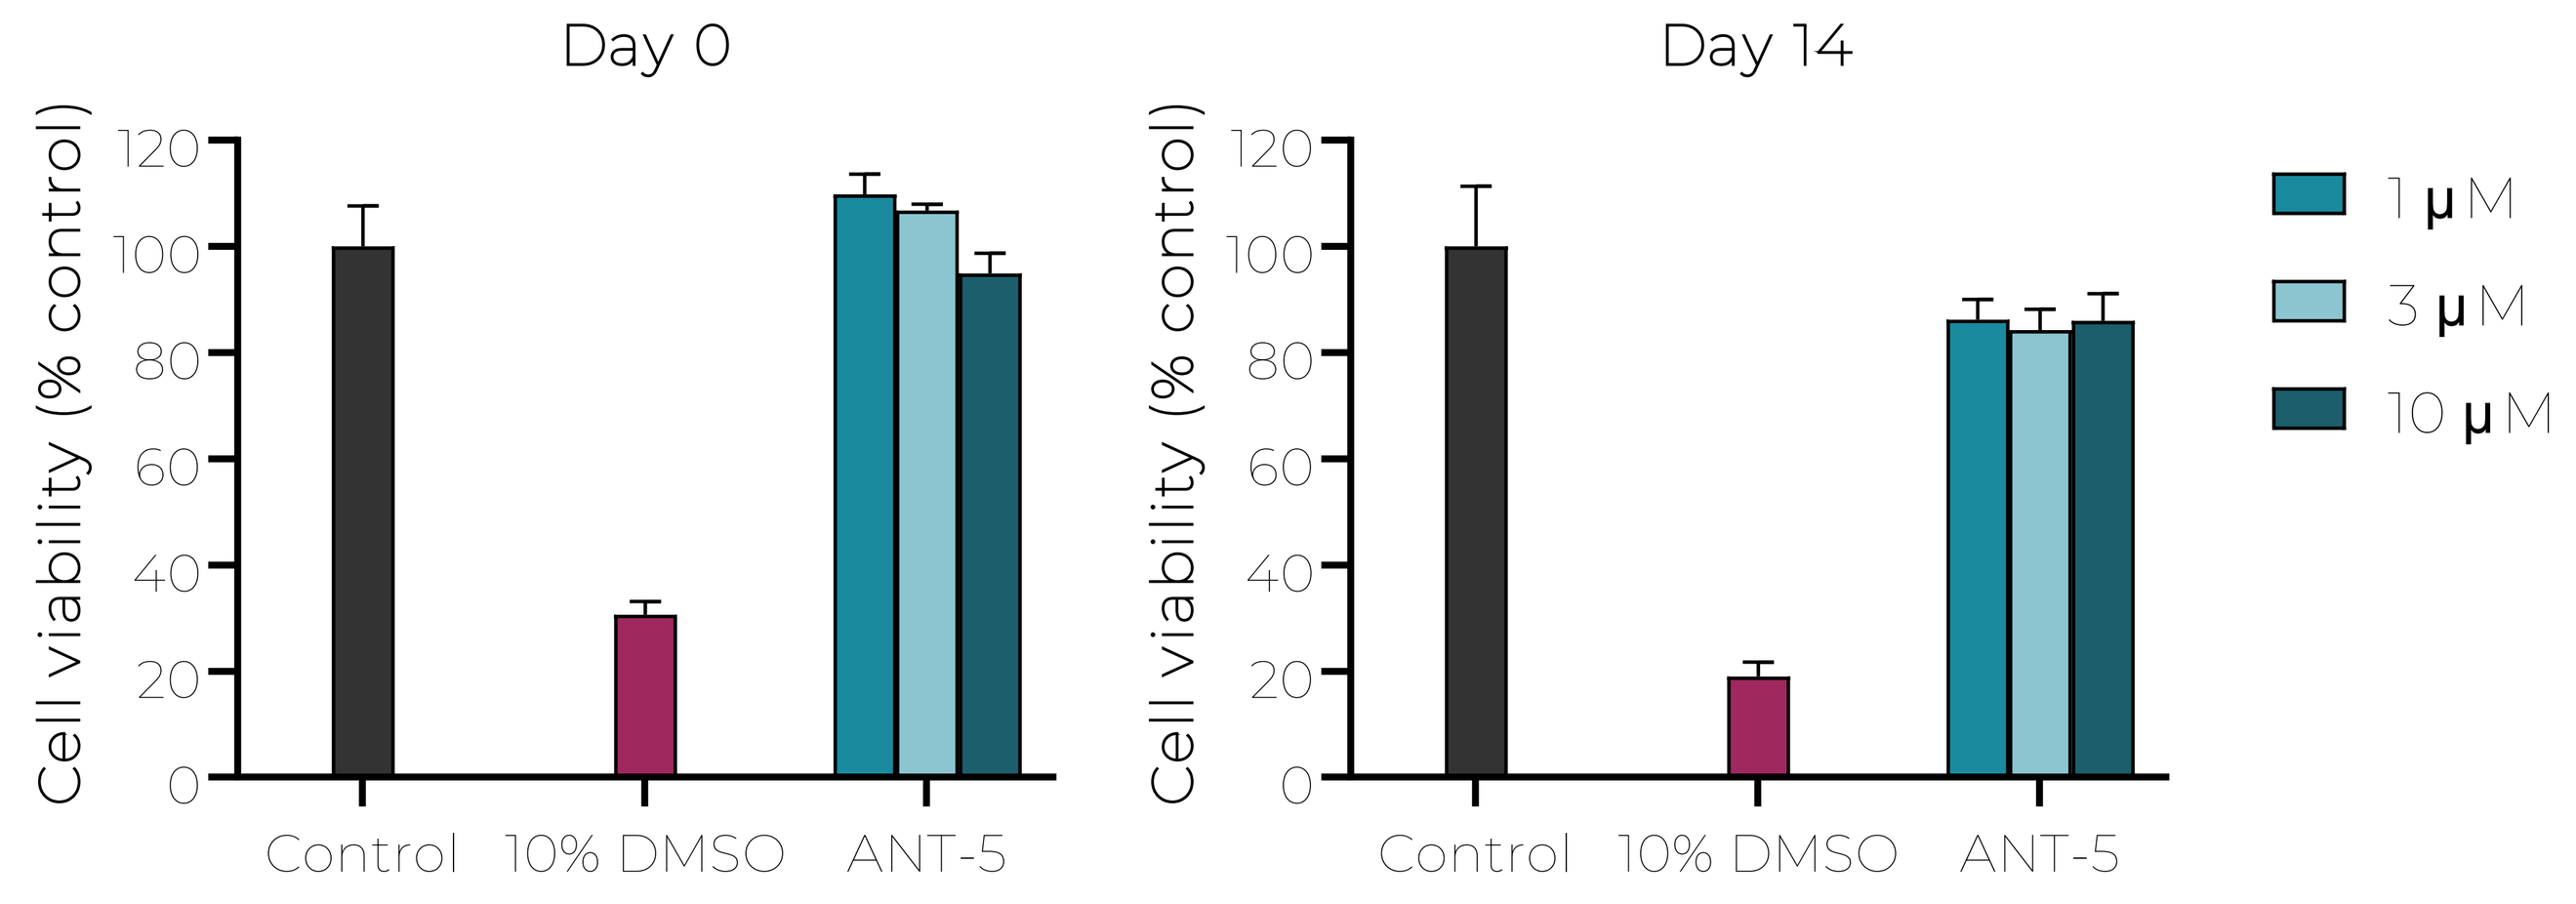
Day 0
Day 14
Cell viability (% control)
Control
10% DMSO
ANT-5
1 μM
3 μM
10 μM

Figure 6. Primary osteoblasts differentiated 0 or 14 days remain healthy after 24 hours of exposure to ANT-5. Viability was measured using the CyQUANT XTT Cell Viability Assay (Thermo Fisher #X12223). Vehicle control included 0.25% DMSO. Bars represent mean±SEM. n=3 technical replicates.

ANT-5 restores *RANKL* and *OPG* mRNA expression to basal levels in the presence of hPTH(1-34)

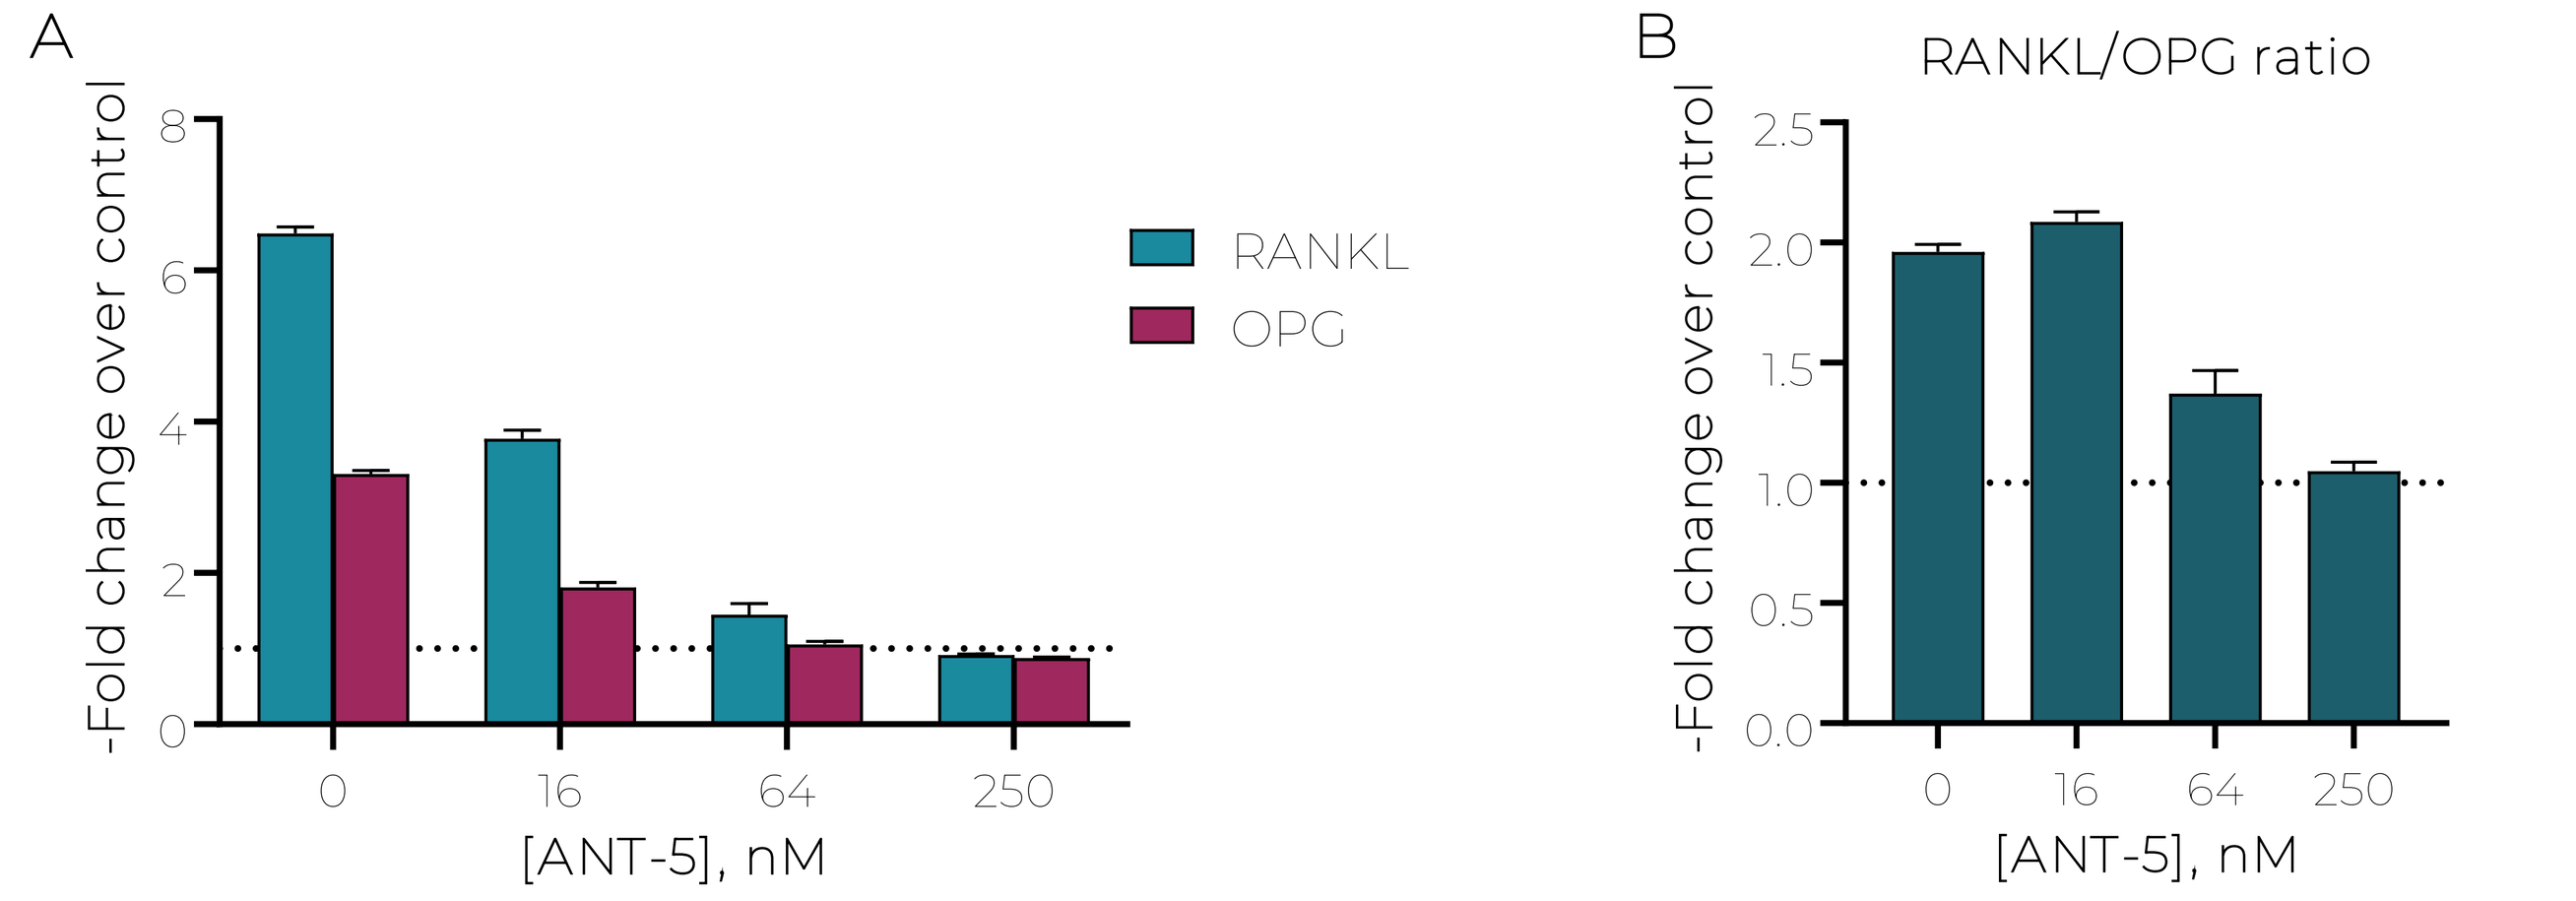
A
-Fold change over control
[ANT-5], nM
RANKL
OPG
B
RANKL/OPG ratio
-Fold change over control
[ANT-5], nM

Figure 7. PTH stimulation of gene expression was inhibited by ANT-5. (A) Primary osteoblasts differentiated 28 days were treated for 8 hours with 1 nM hPTH(1-34) and the indicated concentrations of ANT-5. Dotted line indicates the relative expression of control osteoblasts only exposed to vehicle (0.25% DMSO). Expression of mRNA was measured by real-time RT-PCR and normalized to *β-actin*. Bars represent mean±SEM. n=3 technical replicates. (B) *RANKL/OPG* mRNA radio was calculated from (A).

CONCLUSIONS

- We developed and validated a model of human primary osteoblasts to evaluate PTH1R activation through:
 - cAMP accumulation
 - *RANKL* and *OPG* mRNA expression
- Our novel nonpeptide antagonists blocked PTH1R activation in human primary osteoblasts, as indicated by:
 - Inhibition of PTH1R-mediated cAMP accumulation
 - Restoration of *RANKL* and *OPG* mRNA expression to basal levels in the presence of PTH
- Concentrations of ANT-5 as high as 10 μM did not cause cytotoxicity
- These data strongly suggest that our novel PTH1R antagonists would produce a beneficial effect on bone homeostasis in PHPT patients

ACKNOWLEDGMENTS
This study was funded by Crinetics Pharmaceuticals, Inc.

DISCLOSURES
All authors are employees of Crinetics Pharmaceuticals, Inc.

ASBMR 2023 • October 13-16, 2023 • Vancouver, BC, Canada