Use of the Acromegaly Symptom Diary (ASD) in a Phase 3, Placebo-Controlled Study of Once-Daily, Oral Paltusotine in Patients With Acromegaly Switched From Injected Octreotide or Lanreotide (PATHFNDR-1 Study)

MON-156

Mônica R. Gadelha, MD, PhD[1]; Alessandra Casagrande, MD, PhD[2]; Christian J. Strasburger, MD[3]; Martin Bidlingmaier, MD[4]; Peter J. Snyder, MD[5]; Mirtha A. Guitelman, MD[6]; Cesar L. Boguszewski, MD, PhD[7]; Michael Buchfelder, MD, PhD[8]; Ilan Shimon, MD[9]; Gerald Raverot, MD, PhD[10]; Miklós Tóth, MD, PhD, DSci[11]; Emese Mezősi, MD, PhD, DSc[12]; Mirjana Doknic, MD[13]; Xiaolin Fan, PhD[2]; David Clemmons, MD[14]; Michael Keeley, PhD[2]; Peter J. Trainer, MD[2]; R. Scott Struthers, PhD[2]; Alan Krasner, MD[2]; Beverly M.K. Biller, MD[15]

BACKGROUND

- Paltusotine is the first once-daily, oral SST2 receptor agonist in development for the treatment of acromegaly and carcinoid syndrome[1]
- Collecting symptom data directly from patients on a daily basis in a placebo-controlled study provides unique opportunity to evaluate symptoms as well as biochemical response[2]

METHODS

- Patients with acromegaly previously controlled (IGF-I ≤1.0 × ULN) on injected octreotide or lanreotide; switched to oral paltusotine (or placebo)

Study Design: PATHFNDR-1

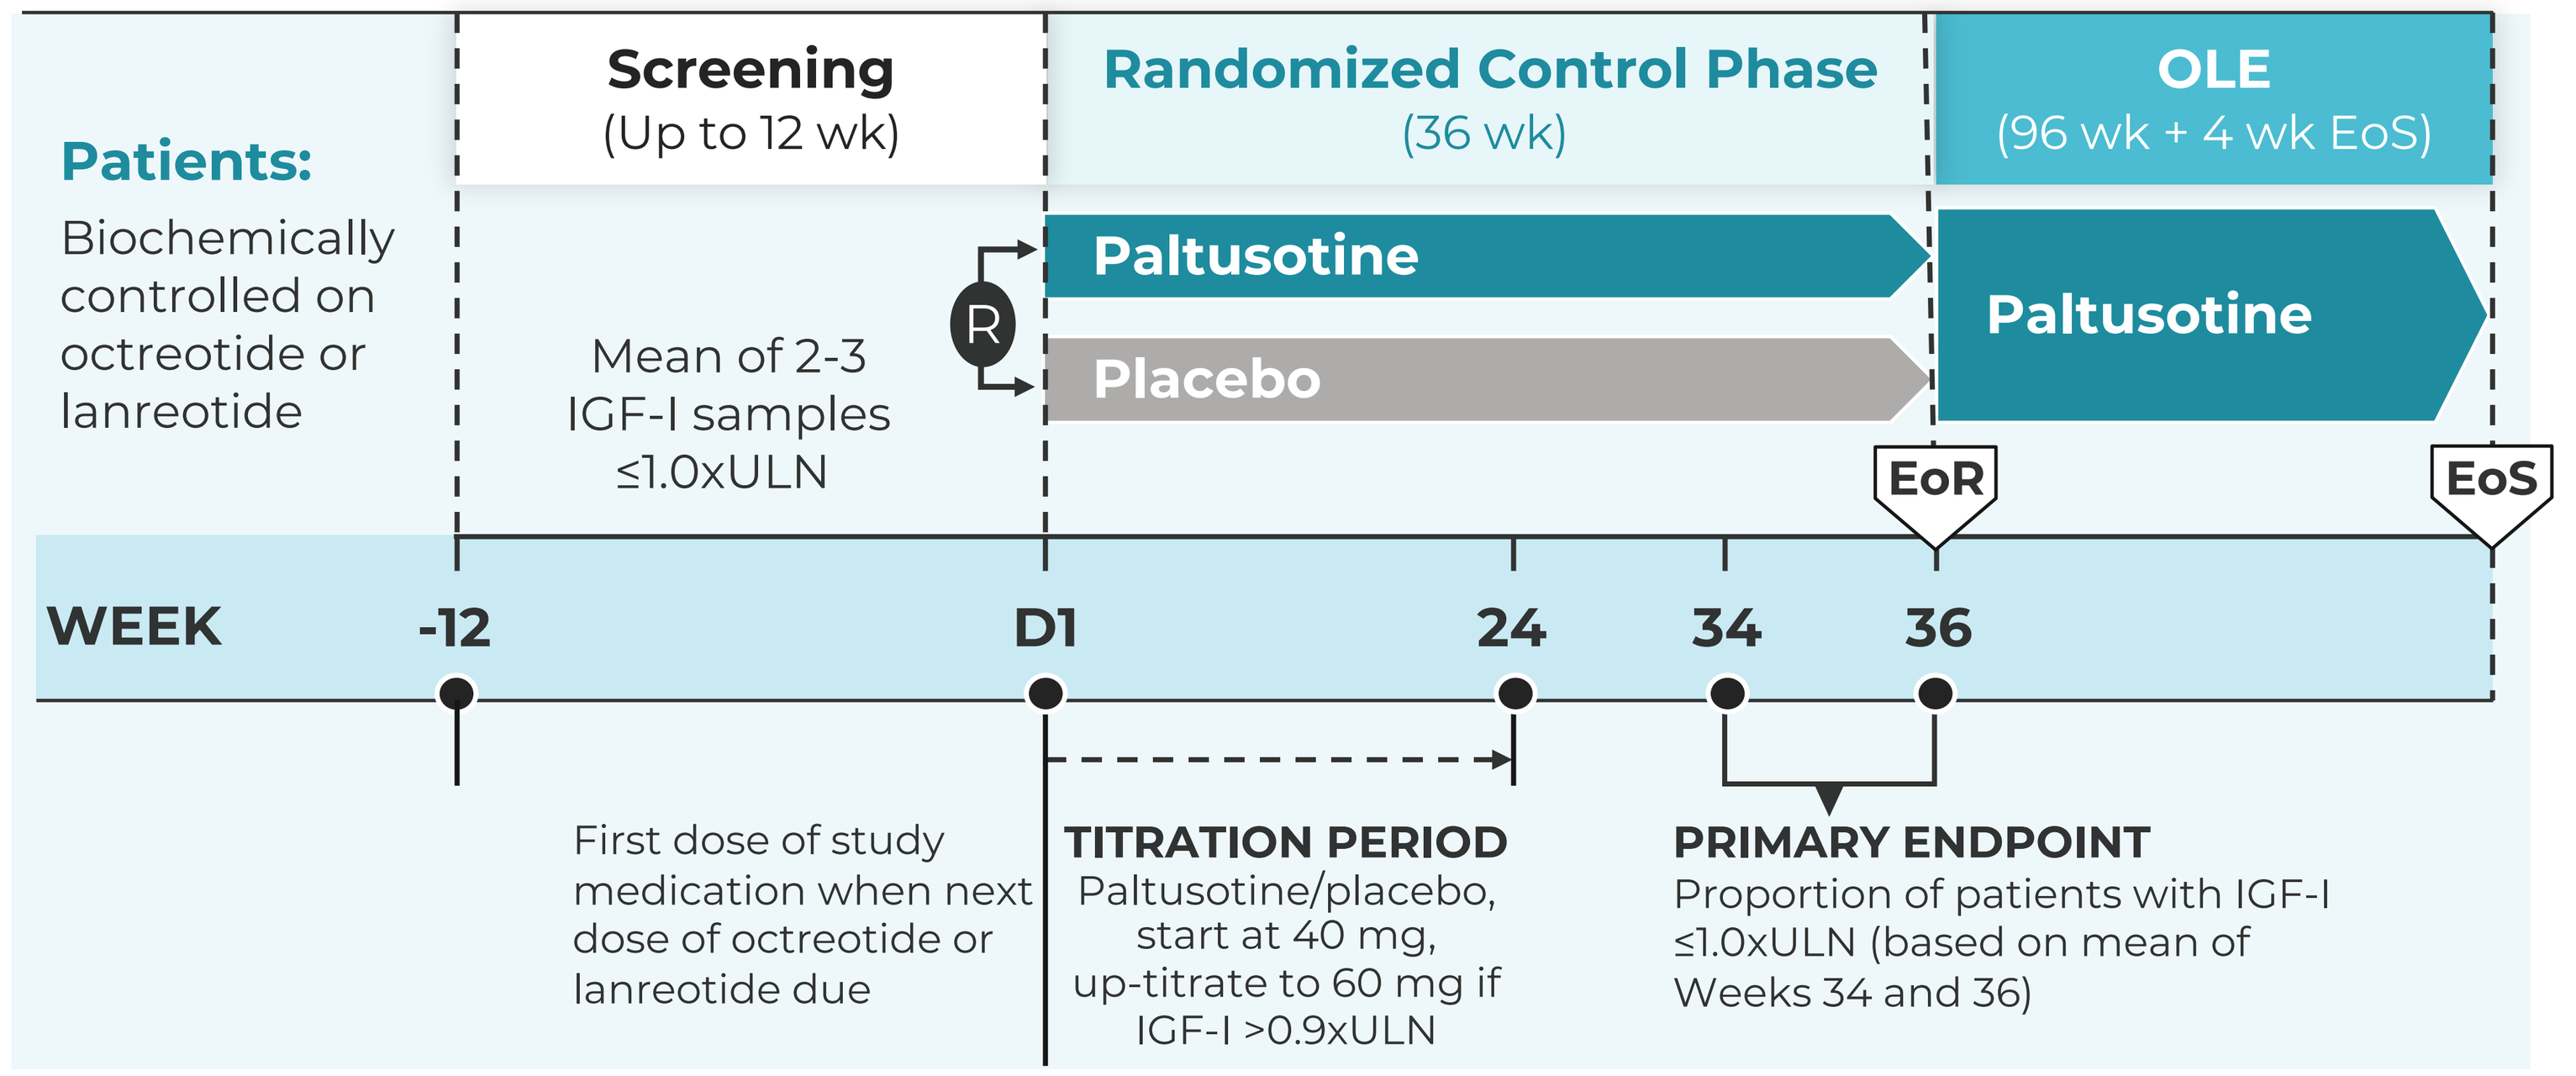
Patients:
Biochemically controlled on octreotide or lanreotide
Screening
(Up to 12 wk)
Mean of 2-3 IGF-I samples ≤1.0xULN
Randomized Control Phase
(36 wk)
R
Paltusotine
Placebo
OLE
(96 wk + 4 wk EoS)
Paltusotine
EoR
EoS
WEEK
-12
D1
24
34
36
First dose of study medication when next dose of octreotide or lanreotide due
TITRATION PERIOD
Paltusotine/placebo, start at 40 mg, up-titrate to 60 mg if IGF-I >0.9xULN
PRIMARY ENDPOINT
Proportion of patients with IGF-I ≤1.0xULN (based on mean of Weeks 34 and 36)

EoR = end of the randomized control phase; EoS = end of study; OLE = open-label extension; R = randomization.

- Paltusotine dose range: 20 to 60 mg based on IGF-I level and tolerability
- IGF-I and GH measured centrally using IDS iSYS immunoassays
- Acromegaly Symptom Diary completed daily during screening (on injected SRL) and on paltusotine/placebo[2]
 - 7 core items: headache, joint pain, sweating, fatigue, leg weakness, swelling, numbness/tingling
 - Score range: 0-70; higher scores → greater symptom burden
 - 2 additional items: sleep difficulty, memory difficulty
- Tumor volume assessed on MRI scans, read centrally
- Statistical methods: exact logistic regression models for primary endpoint and maintenance of GH, analysis of covariance for change from baseline in IGF-I and ASD scores
- Exploratory analysis: intrapatient comparison of breakthrough acromegaly symptom exacerbations while treated with stable dose paltusotine versus injected SRLs

RESULTS

Patient Characteristics

Parameters	Paltusotine (n=30)	Placebo (n=28)
Age, years, mean (SD)	55.9 (14.6)	53.9 (12.9)
Female sex, n (%)	15 (50.0)	17 (60.7)
Duration of acromegaly, n (%)*		
<10 years	5 (16.7)	15 (53.6)
≥10 to <20 years	19 (63.3)	11 (39.3)
≥20 years	6 (20.0)	2 (7.1)
Previous pituitary surgery	26 (86.7)	24 (85.7)
Prior injected SRL, n (%)†		
Octreotide	18 (60.0)	16 (57.1)
Lanreotide	12 (40.0)	12 (42.9)
Baseline IGF-I, ×ULN, mean (SD)	0.83 (0.14)	0.82 (0.16)
Screening GH, ng/mL, mean (SD)‡	0.92 (1.02)	0.89 (0.83)

*Time since diagnosis. †Randomization stratified by prior medication. ‡Mean of 5 samples; screening value served as baseline.

Primary Endpoint Met

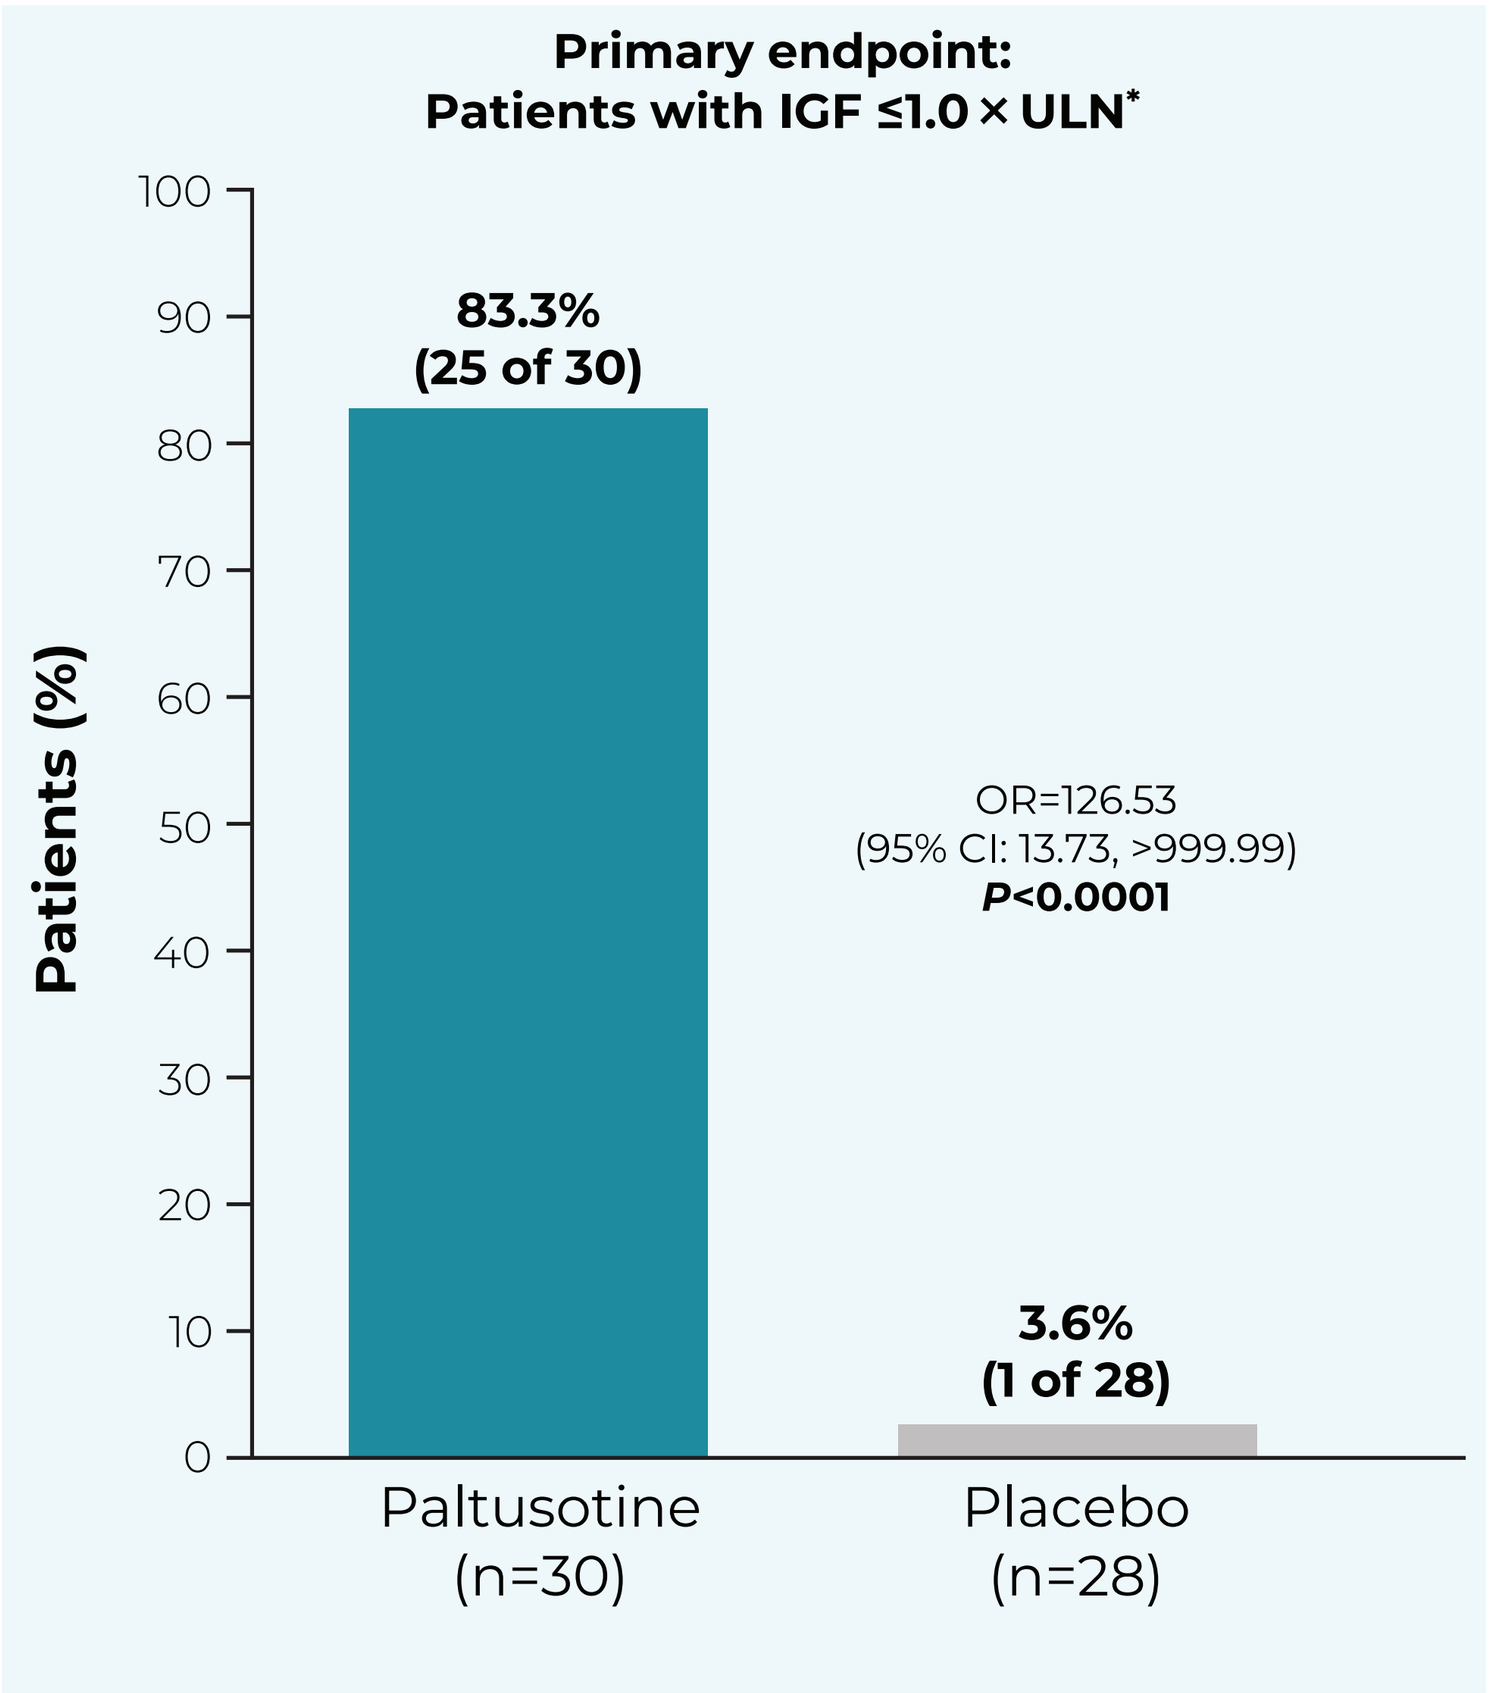
Primary endpoint:
Patients with IGF ≤1.0 × ULN*
Patients (%)
83.3%
(25 of 30)
3.6%
(1 of 28)
OR=126.53
(95% CI: 13.73, >999.99)
P<0.0001
Paltusotine
(n=30)
Placebo
(n=28)

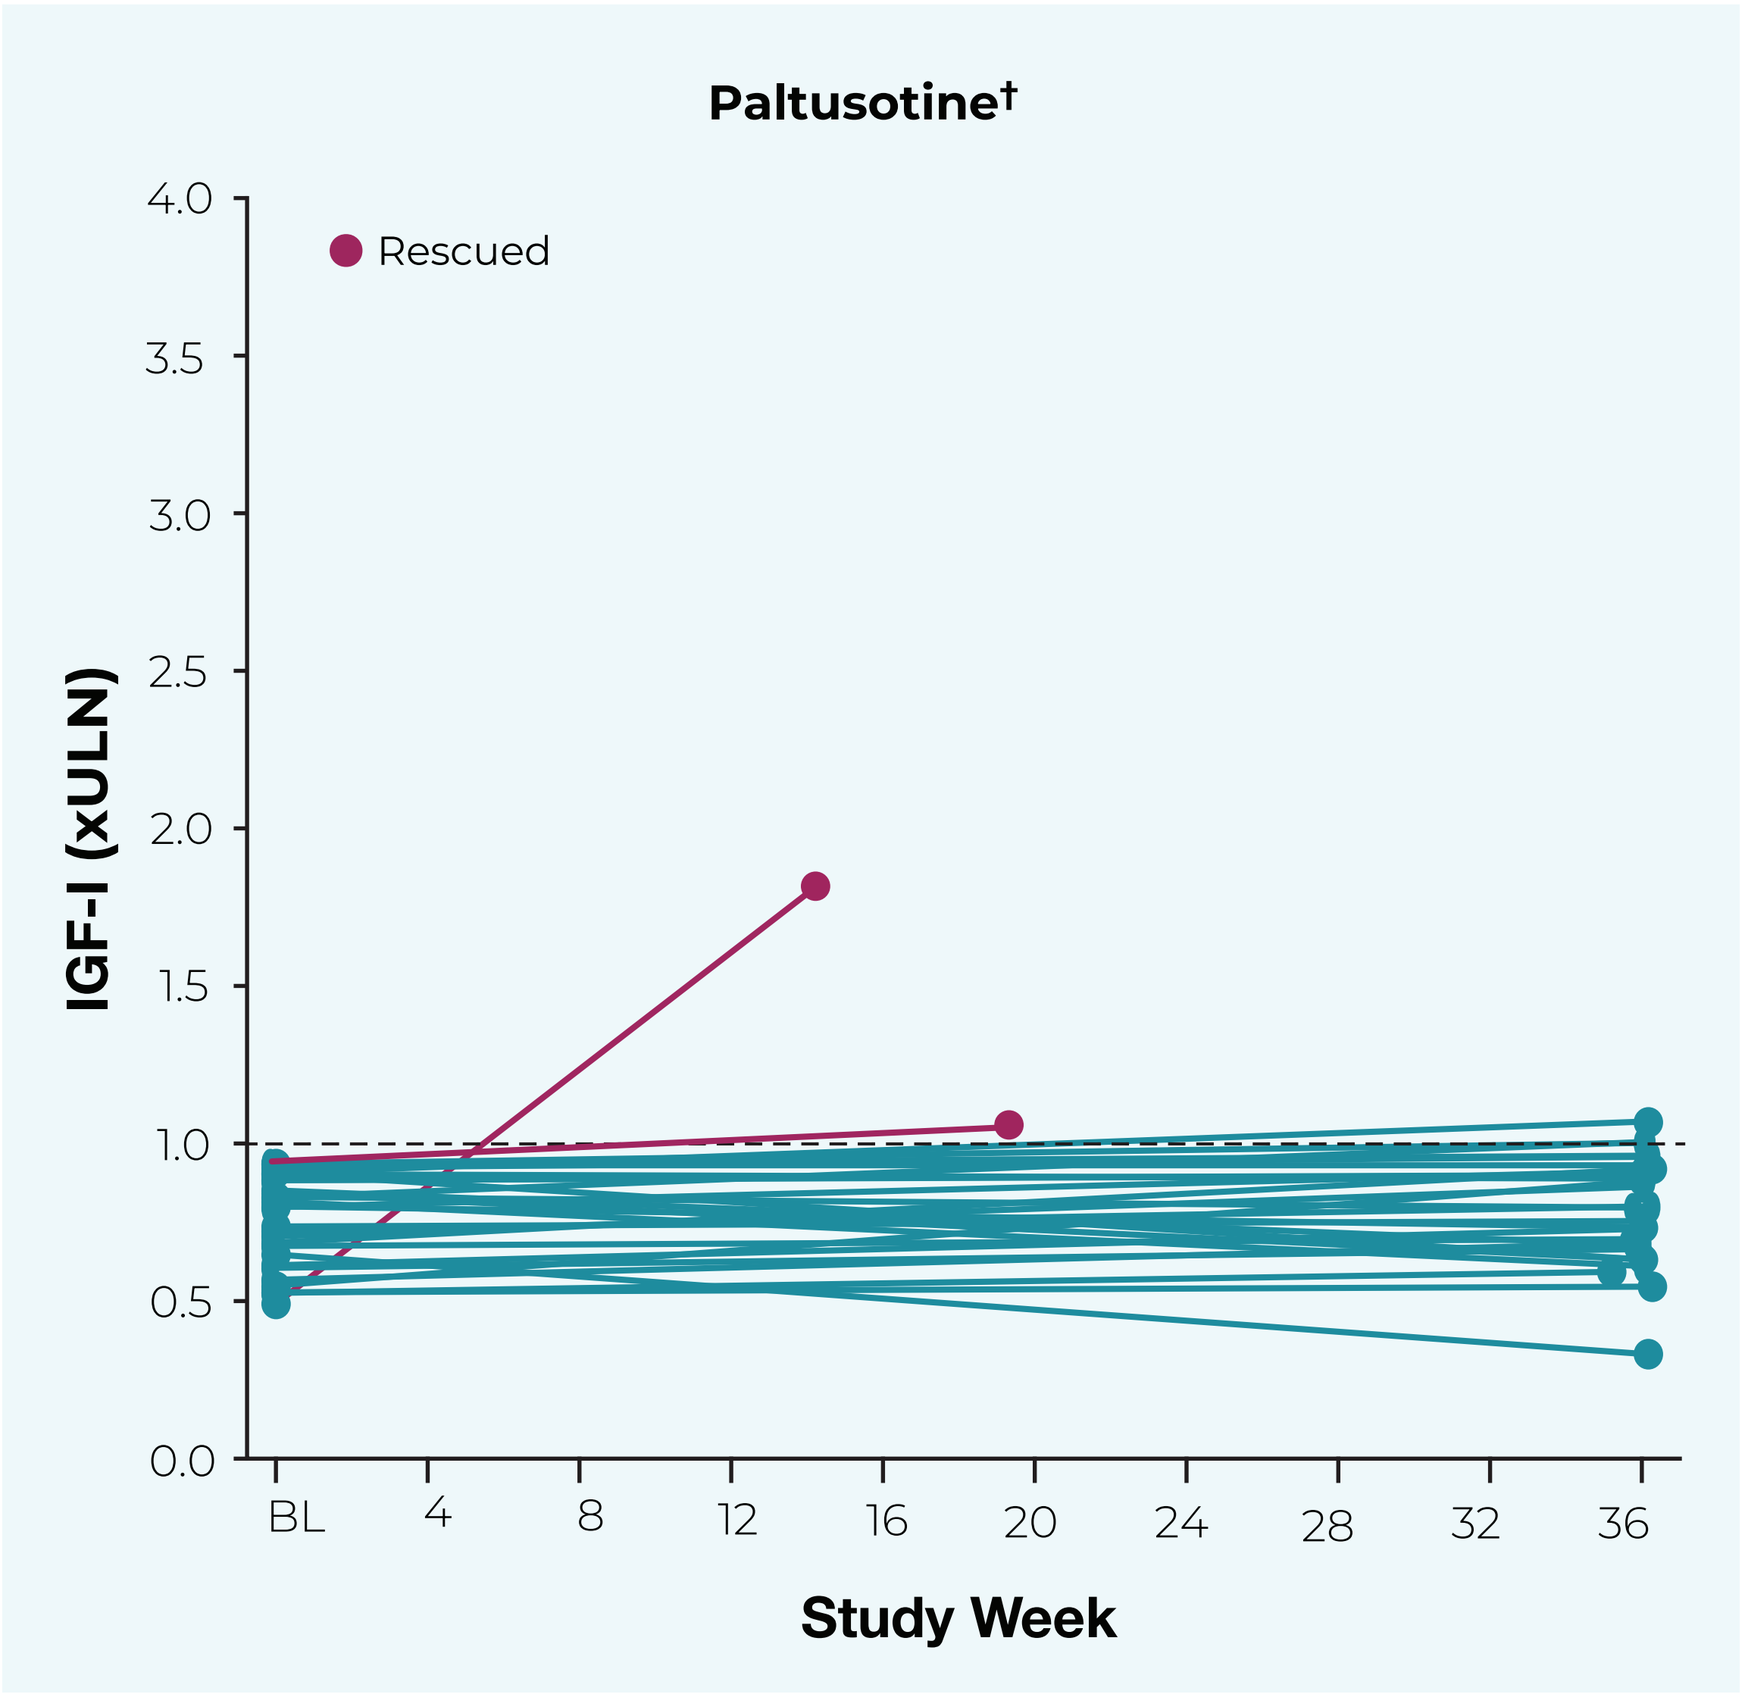
Paltusotine†
Rescued
IGF-I (xULN)
Study Week

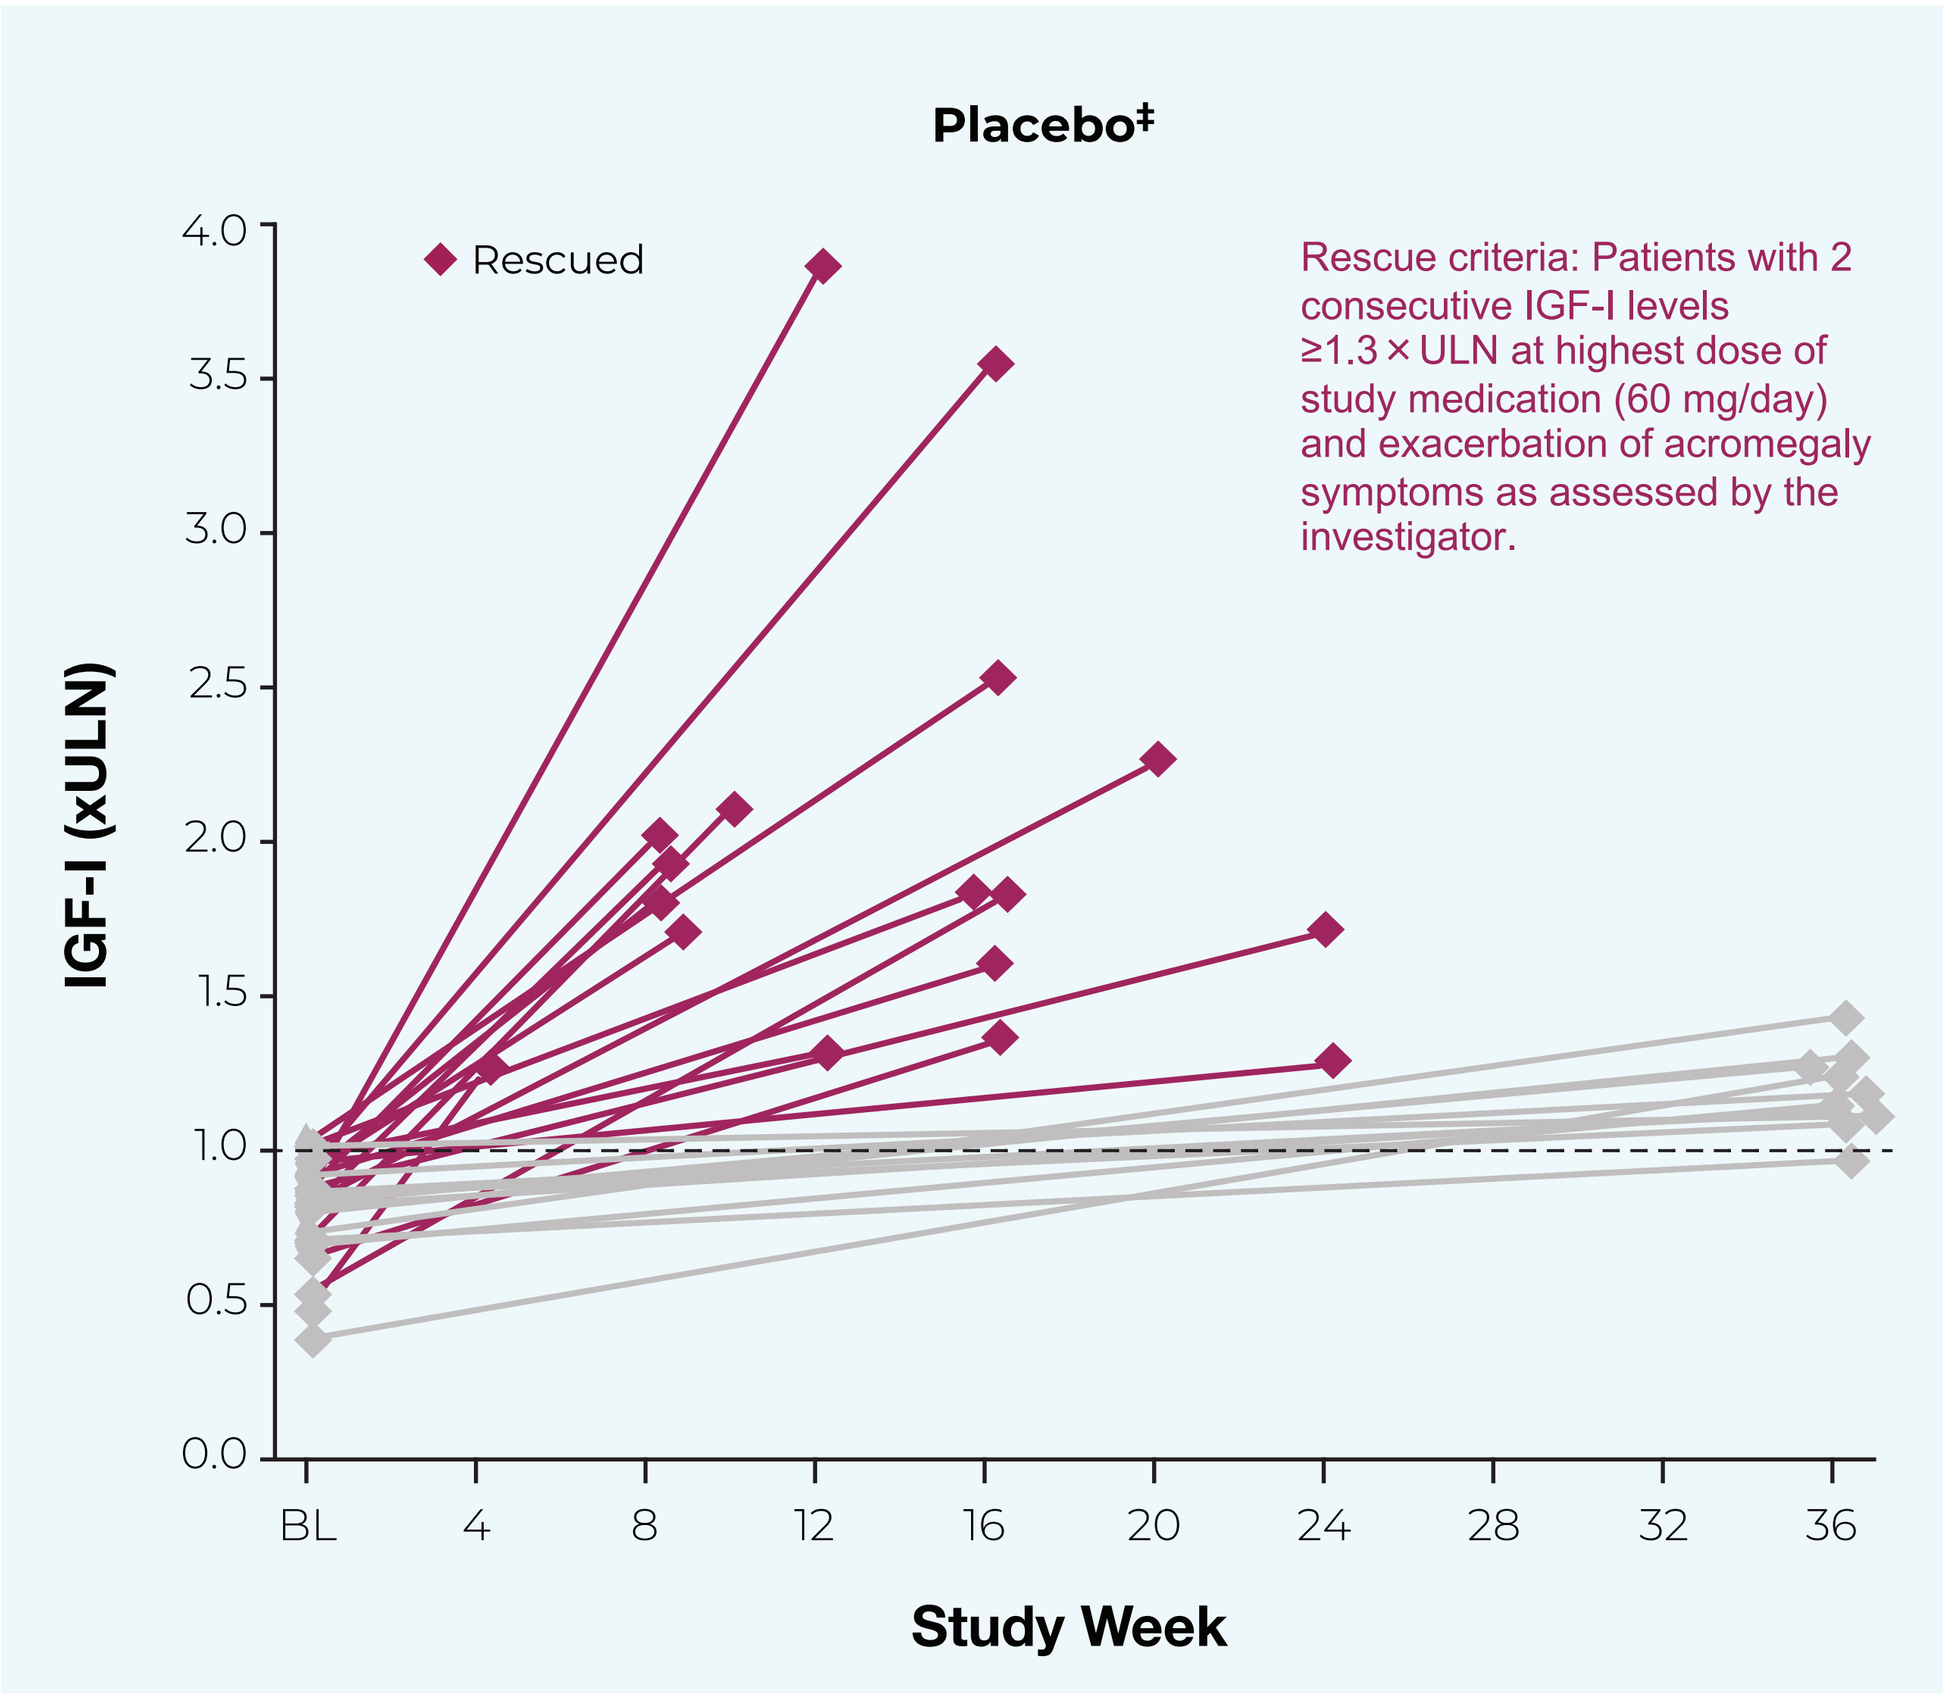
Placebo‡
Rescued
Rescue criteria: Patients with 2 consecutive IGF-I levels ≥1.3 × ULN at highest dose of study medication (60 mg/day) and exacerbation of acromegaly symptoms as assessed by the investigator.
IGF-I (xULN)
Study Week

*Based on mean of 2 measurements (Weeks 34 and 36) in the randomized treatment period. Patients considered to have failed to meet the primary outcome if IGF-I >1.0 × ULN based on the mean of the last 2 measurements, IGF-I missing at both Weeks 34 and 36, treatment discontinued (for any reason) prior to EOR, other acromegaly treatment administered, or uptitration in dose after Week 24. †2 of 30 patients received rescue medication (1 per protocol and 1 who discontinued study treatment due to adverse events). ‡17 of 28 patients received rescue medication (per protocol).

Acromegaly Symptoms

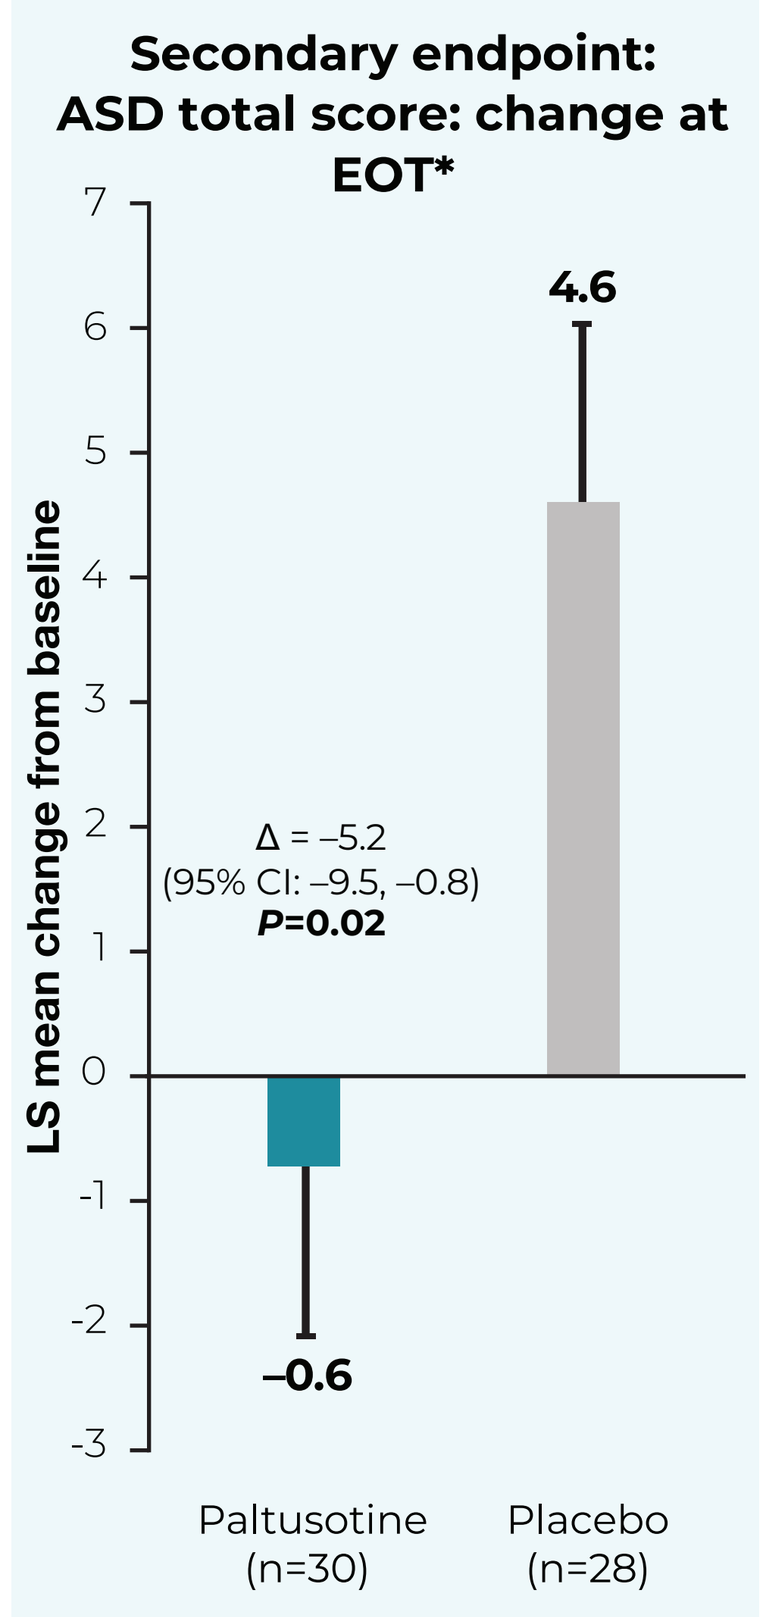
Secondary endpoint:
ASD total score: change at EOT*
LS mean change from baseline
4.6
Δ = –5.2
(95% CI: –9.5, –0.8)
P=0.02
–0.6
Paltusotine
(n=30)
Placebo
(n=28)

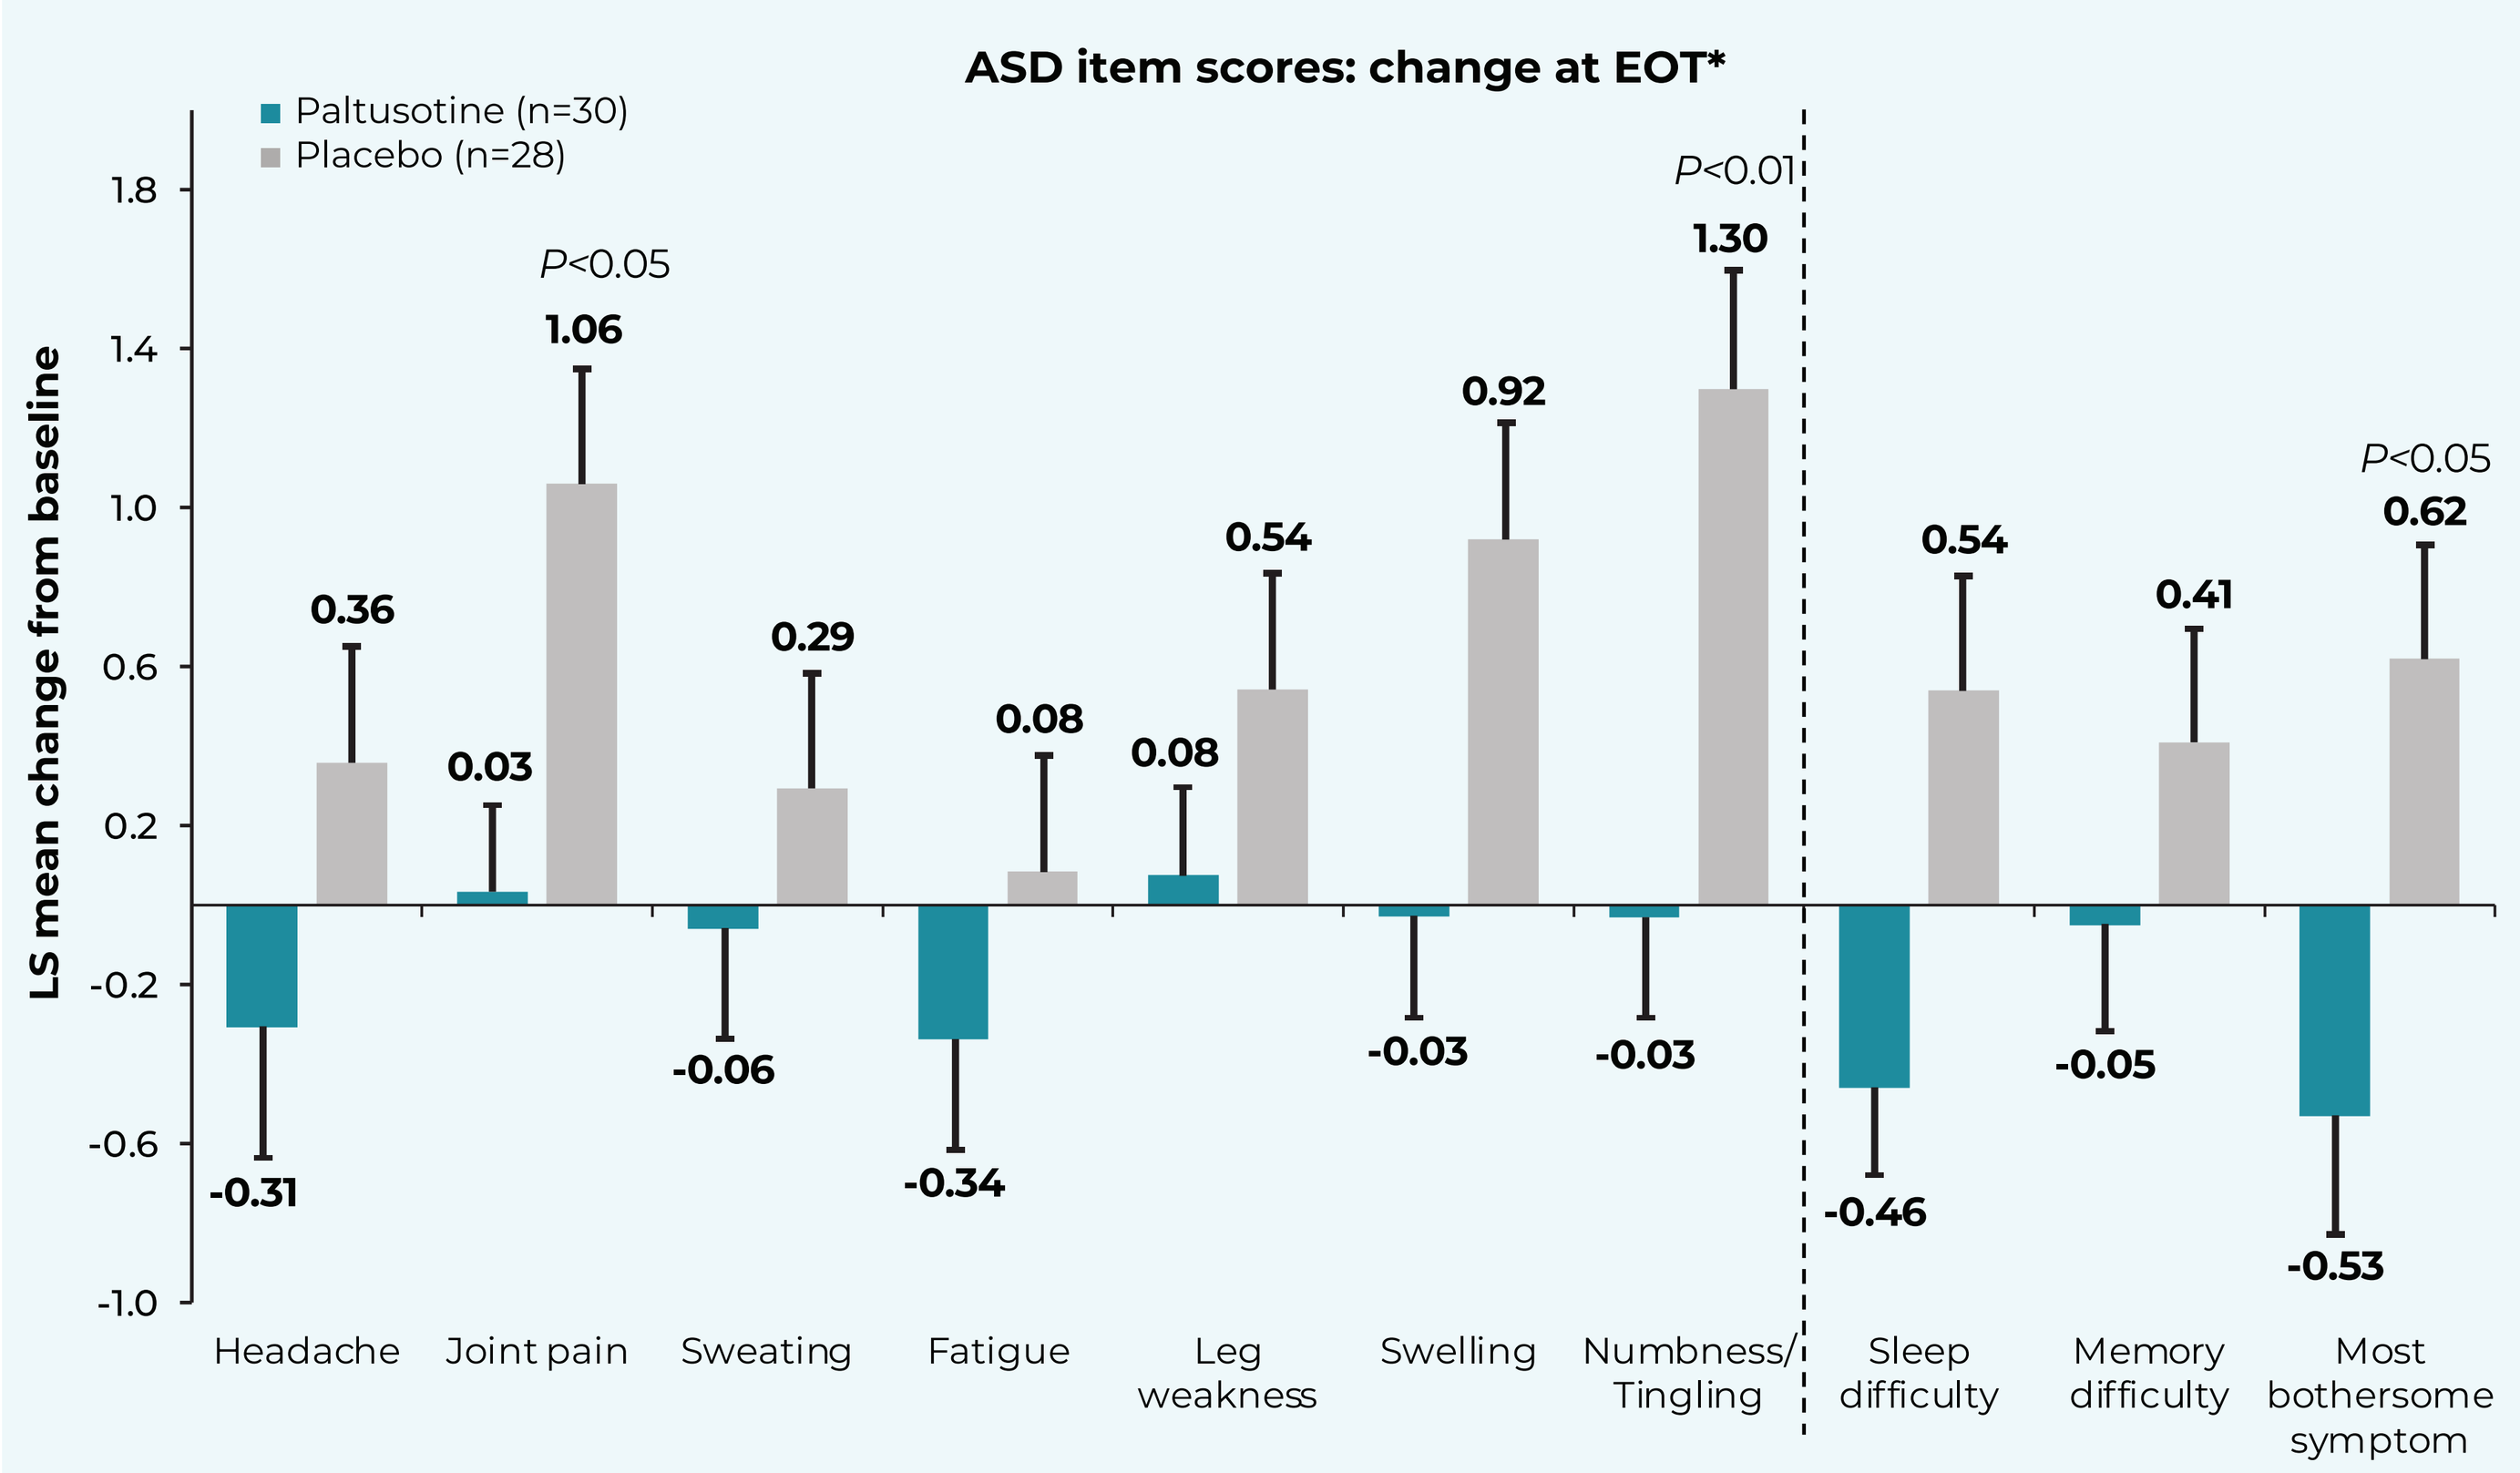
ASD item scores: change at EOT*
Paltusotine (n=30)
Placebo (n=28)
LS mean change from baseline
Headache -0.31 0.36
Joint pain 0.03 1.06 P<0.05
Sweating -0.06 0.29
Fatigue -0.34 0.08
Leg weakness 0.08 0.54
Swelling -0.03 0.92
Numbness/Tingling -0.03 1.30 P<0.01
Sleep difficulty -0.46 0.54
Memory difficulty -0.05 0.41
Most bothersome symptom -0.53 0.62 P<0.05

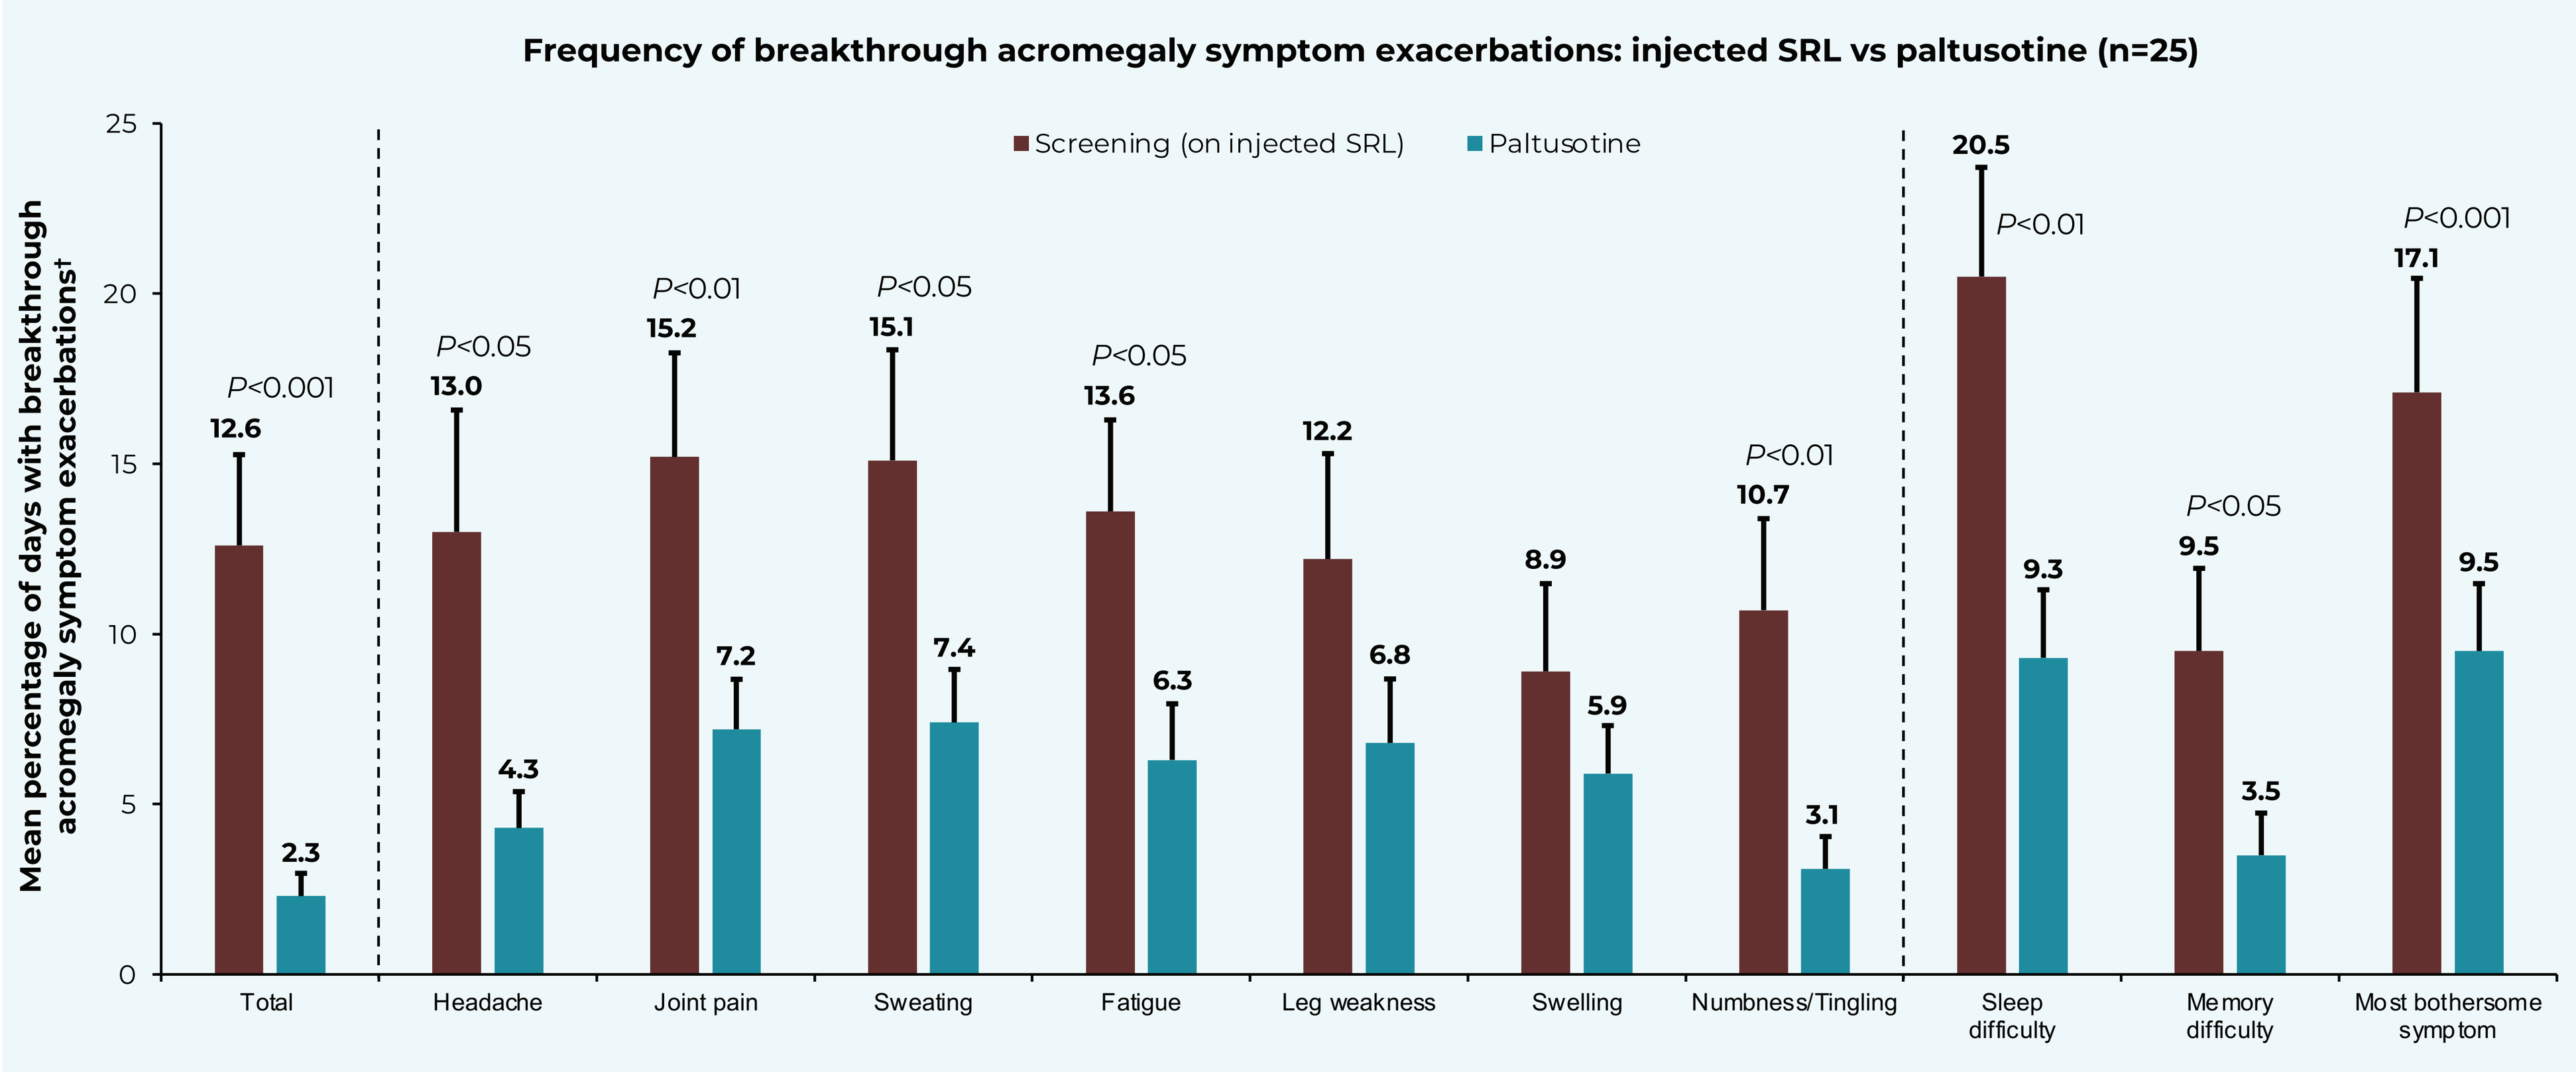
Frequency of breakthrough acromegaly symptom exacerbations: injected SRL vs paltusotine (n=25)
Screening (on injected SRL)
Paltusotine
Mean percentage of days with breakthrough acromegaly symptom exacerbations†
Total 12.6 2.3 P<0.001
Headache 13.0 4.3 P<0.05
Joint pain 15.2 7.2 P<0.01
Sweating 15.1 7.4 P<0.05
Fatigue 13.6 6.3 P<0.05
Leg weakness 12.2 6.8
Swelling 8.9 5.9
Numbness/Tingling 10.7 3.1 P<0.01
Sleep difficulty 20.5 9.3 P<0.01
Memory difficulty 9.5 3.5 P<0.05
Most bothersome symptom 17.1 9.5 P<0.001

Mean completion rate of daily ASD questionnaire, 79.1%. Error bars represent SE. *EOT (end of treatment) defined as Week 36 if no rescue medication administered, or last assessment prior to rescue.

Error bars represent SE. Breakthrough acromegaly symptom exacerbation defined as 4-point increase for ASD total score or 1-point increase for individual item score comparing a 2-day average to the previous 2-day average. Includes all available ASD data from injected SRL (screening) and paltusotine stable dosing (Months 6-9) periods.

- Other secondary endpoints:
 - Mean IGF-I increase from 161.7 ng/mL to 329.2 ng/mL with placebo and 163.3 ng/mL to 173.8 ng/mL with paltusotine (treatment difference = -158.2 ng/mL; 95% CI: -213.0, -103.4; *P*<0.0001)
 - 5-sample mean GH maintained at <1.0 ng/mL in 87.0% of patients on paltusotine versus 27.8% on placebo (OR: 16.61; 95% CI: 2.86, 181.36; *P*<0.001)

CONCLUSIONS

- Once-daily, oral paltusotine maintained biochemical and symptom control in patients with acromegaly switched from SRLs
- Paltusotine was associated with lower frequency of breakthrough acromegaly symptom exacerbations compared with injected SRLs
- Paltusotine was well tolerated with no severe or serious adverse events

Safety

Adverse Events, n (%)*	Paltusotine (n=30)	Placebo (n=28)
Arthralgia	9 (30.0)	16 (57.1)
Diarrhea	7 (23.3)	4 (14.3)
Headache	6 (20.0)	10 (35.7)
Abdominal pain	5 (16.7)	3 (10.7)
Nausea	4 (13.3)	2 (7.1)
Constipation	2 (6.7)	4 (14.3)
Fatigue	2 (6.7)	5 (17.9)
Peripheral swelling	2 (6.7)	10 (35.7)
Asthenia	1 (3.3)	4 (14.3)
Muscle weakness	1 (3.3)	3 (10.7)
Paresthesia	0	7 (25.0)
Hyperhidrosis	0	4 (14.3)
Pain in extremity	0	3 (10.7)

*Adverse events that emerged or worsened during study treatment; including events with onset after rescue medication was administered.

- No severe or serious AEs in the paltusotine group
- No clinically significant changes in pituitary tumor volume

For author affiliations, acknowledgments, and disclosures, please use the QR code.

ENDO 2024 • June 1-4, 2024 • Boston, MA

ACKNOWLEDGMENTS
The authors thank the site investigators, study coordinators/nurses, clinical staff, and patients who participated in the study.

REFERENCES
1. Zhao J, et al. *ACS Med Chem Lett*. 2023;14(1):66-74. **2.** Martin S, et al. *J Patient Rep Outcomes*. 2023;7(1):15.

Crinetics Pharmaceuticals

AUTHOR AFFILIATIONS

[1]Neuroendocrinology Research Center/Endocrinology Division, Medical School and Hospital Universitario Clementino Fraga Filho, Universidade Federal do Rio de Janeiro, Rio de Janeiro, Brazil; [2]Crinetics Pharmaceuticals, Inc., San Diego, CA, USA; [3]Department of Medicine for Endocrinology and Metabolism, Charité Universitaetsmedizin, Campus Mitte, Berlin, Germany; [4]Medizinische Klinik und Poliklinik IV, Klinikum der Universität München, Munich, Germany; [5]Division of Endocrinology, Diabetes and Metabolism, Department of Medicine, University of Pennsylvania, Philadelphia, PA, USA; [6]División Endocrinología, Hospital Carlos G. Durand, Buenos Aires, Argentina; [7]Department of Internal Medicine, Endocrine Division (SEMPR), University Hospital, Federal University of Paraná, Curitiba, Brazil; [8]Neurosurgery Department, University Hospital Erlangen, Erlangen, Germany; [9]Institute of Endocrinology, Rabin-Beilinson Medical Center, Faculty of Medicine, Tel Aviv University, Tel Aviv, Israel; [10]Endocrinology Department, Reference Center for Rare Pituitary Diseases HYPO, "Groupement Hospitalier Est" Hospices Civils de Lyon, Lyon Cedex, France; [11]Department of Internal Medicine and Oncology, Semmelweis University, Budapest, Hungary; [12]Department of Medicine, University of Pécs Medical School, Pécs, Hungary; [13]Neuroendocrine Department, Clinic for Endocrinology, Diabetes and Metabolic Diseases, University Clinical Center of Serbia, Faculty of Medicine, University of Belgrade, Belgrade, Serbia; [14]University of North Carolina at Chapel Hill, Chapel Hill, NC, USA; [15]Neuroendocrine and Pituitary Tumor Clinical Center, Massachusetts General Hospital and Harvard Medical School, Boston, MA, USA

ACKNOWLEDGMENTS

The authors thank the site investigators, study coordinators/nurses, clinical staff, and patients who participated in the study. This study was funded by Crinetics Pharmaceuticals, Inc. The authors thank Michael Monahan, MBA, Crinetics Pharmaceuticals, for his contributions. Technical editorial and medical writing assistance were provided under the direction of the authors by Janetricks Okeyo, PhD, Crinetics Pharmaceuticals, and Synchrony Medical Communications, LLC, West Chester, PA, USA; funding for this support was provided by Crinetics Pharmaceuticals, Inc.

DISCLOSURES

MRG reports being a principal investigator (PI) of research grants from Crinetics and Recordati; occasional consultant for Crinetics, Ipsen, Novo Nordisk, and Recordati; and speaker for Ipsen, Novo Nordisk, and Recordati. AC, XF, MK, PJT, RSS, and AK are employees and stock shareholders of Crinetics Pharmaceuticals. CJS reports being a PI of a research grant from Crinetics to Charité Universitaetsmedizin; and occasional consultant/speaker for Amolyt Pharma, Crinetics, Debiopharm, Novo Nordisk, Pfizer, Recordati, and Sandoz-Hexal. M Bidlingmaier reports being a PI of research grants from Amolyt, Camurus, Chiasma, Crinetics, IDS, Ionis, Lumos, and OPKO; occasional consultant for Crinetics, Ionis, Novo Nordisk, Pfizer, Roche, and Sandoz; and speaker for Euroimmun, Novo Nordisk, and Pfizer. PJS, M Buchfelder, and EM report being a PI of a research grant from Crinetics. MG reports being a PI of research grants from Crinetics; and occasional consultant and speaker for Recordati, Sanofi Aventis, and Varifarma. CLB reports receiving consulting fees, honoraria, and meeting support from Ipsen, Novo Nordisk, and Recordati; and serving on advisory boards for Novo Nordisk and Recordati. IS reports being a PI of a research grant from Crinetics and Debiopharm; and speaker and consultant for Medison and Pfizer. GR reports being a PI of a research grant from Crinetics; occasional consultant for Amolyt, Crinetics, Ipsen, Novo Nordisk, Pfizer, and Recordati; and speaker for Ipsen, Pfizer, and Recordati. MT reports being a PI of a research grant from Crinetics; and speaker and consultant for Ipsen, Lilly, Novartis, and Recordati. MD reports being a PI of a research grant from Crinetics; and advisory board member for Pfizer in CEE. DRC is a consultant for Crinetics. BMKB reports being a PI of research grants from Crinetics and Ionis; and occasional consultant for Amolyt, Amryt, Camurus, Crinetics, and Recordati.