Reductions in Adrenal Volume in Patients With Congenital Adrenal Hyperplasia Receiving Once-Daily Oral Atumelnant (CRN04894): Interim Results From a 12-Week, Phase 2, Open-Label Study

SAT-452

Tania A.S.S. Bachega, MD, PhD[1]; Jamie Marko, MD[2]; Dario Bruera, MD[3]; Alejandro Ayala, MD[4]; Mônica R. Gadelha, MD, PhD[5]; Nicole Reisch, MD[6]; Yang Wu, PhD[4]; Eduardo De la Torre Ames, MD[4]; Alan Krasner, MD[4]; Richard J. Auchus, MD, PhD[7]; Umasuthan Srirangalingam, MD, PhD[8]

BACKGROUND

- Congenital adrenal hyperplasia (CAH) is a rare disease of adrenocorticotropic hormone (ACTH)–excess due to inadequate cortisol synthesis and/or aldosterone synthesis[1]
- Excess stimulation by ACTH, exerting a trophic effect over the lifetime of the individual, causes enlargement of the adrenal glands in patients with CAH;[2] thus, measurement of adrenal volume may provide insight into the degree of ACTH suppression and its potential beneficial effect on adrenal hyperplasia in patients with CAH[3]
- Atumelnant (CRN04894) is a first-in-class, once-daily, oral, nonpeptide, melanocortin type 2 receptor (MC2R) antagonist that selectively and specifically inhibits ACTH-mediated adrenal steoidogenesis and is being developed for the treatment of CAH and ACTH-dependent Cushing's syndrome[4]
- In a Phase 2, open-label, dose-finding study of atumelnant (40 mg, 80 mg, or 120 mg) in adults with classic CAH (21-hydroxylase deficiency) (NCT05907291), treatment with atumelnant has demonstrated rapid and marked reductions in primary endpoint, morning androstenedione (A4), within 2 weeks of treatment that were maintained for 12 weeks of treatment[4]

OBJECTIVE

- To present results from an exploratory endpoint of the ongoing Phase 2 study: change in adrenal volume from baseline to Week 12

Study Design and Methods

Key Eligibility Criteria

- Male or female participants age ≥18 to 75 years (≥16 years in USA)
- Classic CAH (21-hydroxylase deficiency)
- On ≥15 mg hydrocortisone equivalent daily dose for ≥6 months (stable for duration of study)
- Serum A4 >1.5 × ULN

Cohorts

- 40 mg once daily (n=11)
- 80 mg once daily (n=11)
- 120 mg once daily (n=6)

Study Endpoints

Primary: Change from baseline in serum A4

Secondary: Change from baseline in serum 17-OHP

Safety: TEAEs

Exploratory:
- Change from baseline in serum steroid levels and PD biomarkers
- PK profile of atumelnant
- Impact of atumelnant on medical outcomes in participants with CAH including change from baseline in adrenal volume

Assessment Schedule (Week)

BL · 2 · 6 · 12 · 14 · 16[a]

Adrenal glands MRI (BL) · Adrenal glands MRI (12)

17-OHP; 17-hydroxyprogesterone; A4, androstenedione; BL, baseline; CAH, congenital adrenal hyperplasia; MRI, magnetic resonance imaging; PD, pharmacodynamic; PK, pharmacokinetic; TEAE, treatment-emergent adverse events; ULN, upper limit of normal.
[a]Week 16 was scheduled as a phone visit.

- Change in adrenal gland size from baseline to Week 12 was an exploratory endpoint
- Adrenal gland size and morphology were assessed via magnetic resonance imaging (MRI) following a standardized image acquisition protocol at baseline (during screening and prior to atumelnant dosing on Day 1) and Week 12
- All MRI assessments were read by a single central radiologist; total volume was derived as the sum of the left and right adrenal gland volumes. If only one side has evaluable volume, then that is set to the total volume

ENDO 2025 • July 12-15, 2025 • San Francisco, CA

RESULTS

- As of January 14, 2025, 19 participants (40 mg, n=5; 80 mg, n=9; and 120 mg, n=5) had evaluable adrenal MRI data at baseline and Week 12 and were included in this analysis

Parameters	40 mg cohort (n=5)	80 mg cohort (n=9)	120 mg cohort (n=5)	All (N=19)
Age, years, mean (range)	34.4 (27-45)	32.6 (22-42)	32.4 (22-47)	33.0 (22-47)
Female, n (%)	3 (60)	7 (78)	2 (40)	12 (63)
GC dose, mg/day, mean (range)[a]	28.7 (20-34)	29.9 (20-40)	24.0 (20-30)	28.0 (20-40)
Median (range) serum A4, ng/dL	1180 (409-2600)	1125 (116-2755)	778 (383-2025)	1049 (116-2755)

A4, androstenedione; GC, glucocorticoid.
[a]GC dose in hydrocortisone equivalents.

- At baseline, total bilateral adrenal volume (reference range, 8-10 mL) was >10 mL in 18/19 participants (median [range] 22 [9.8-943.6] mL)
- Following 12 weeks of atumelnant treatment, 15/19 participants had a decline in total adrenal volume

Change From Baseline in Total Adrenal Volume Per Patient[a]

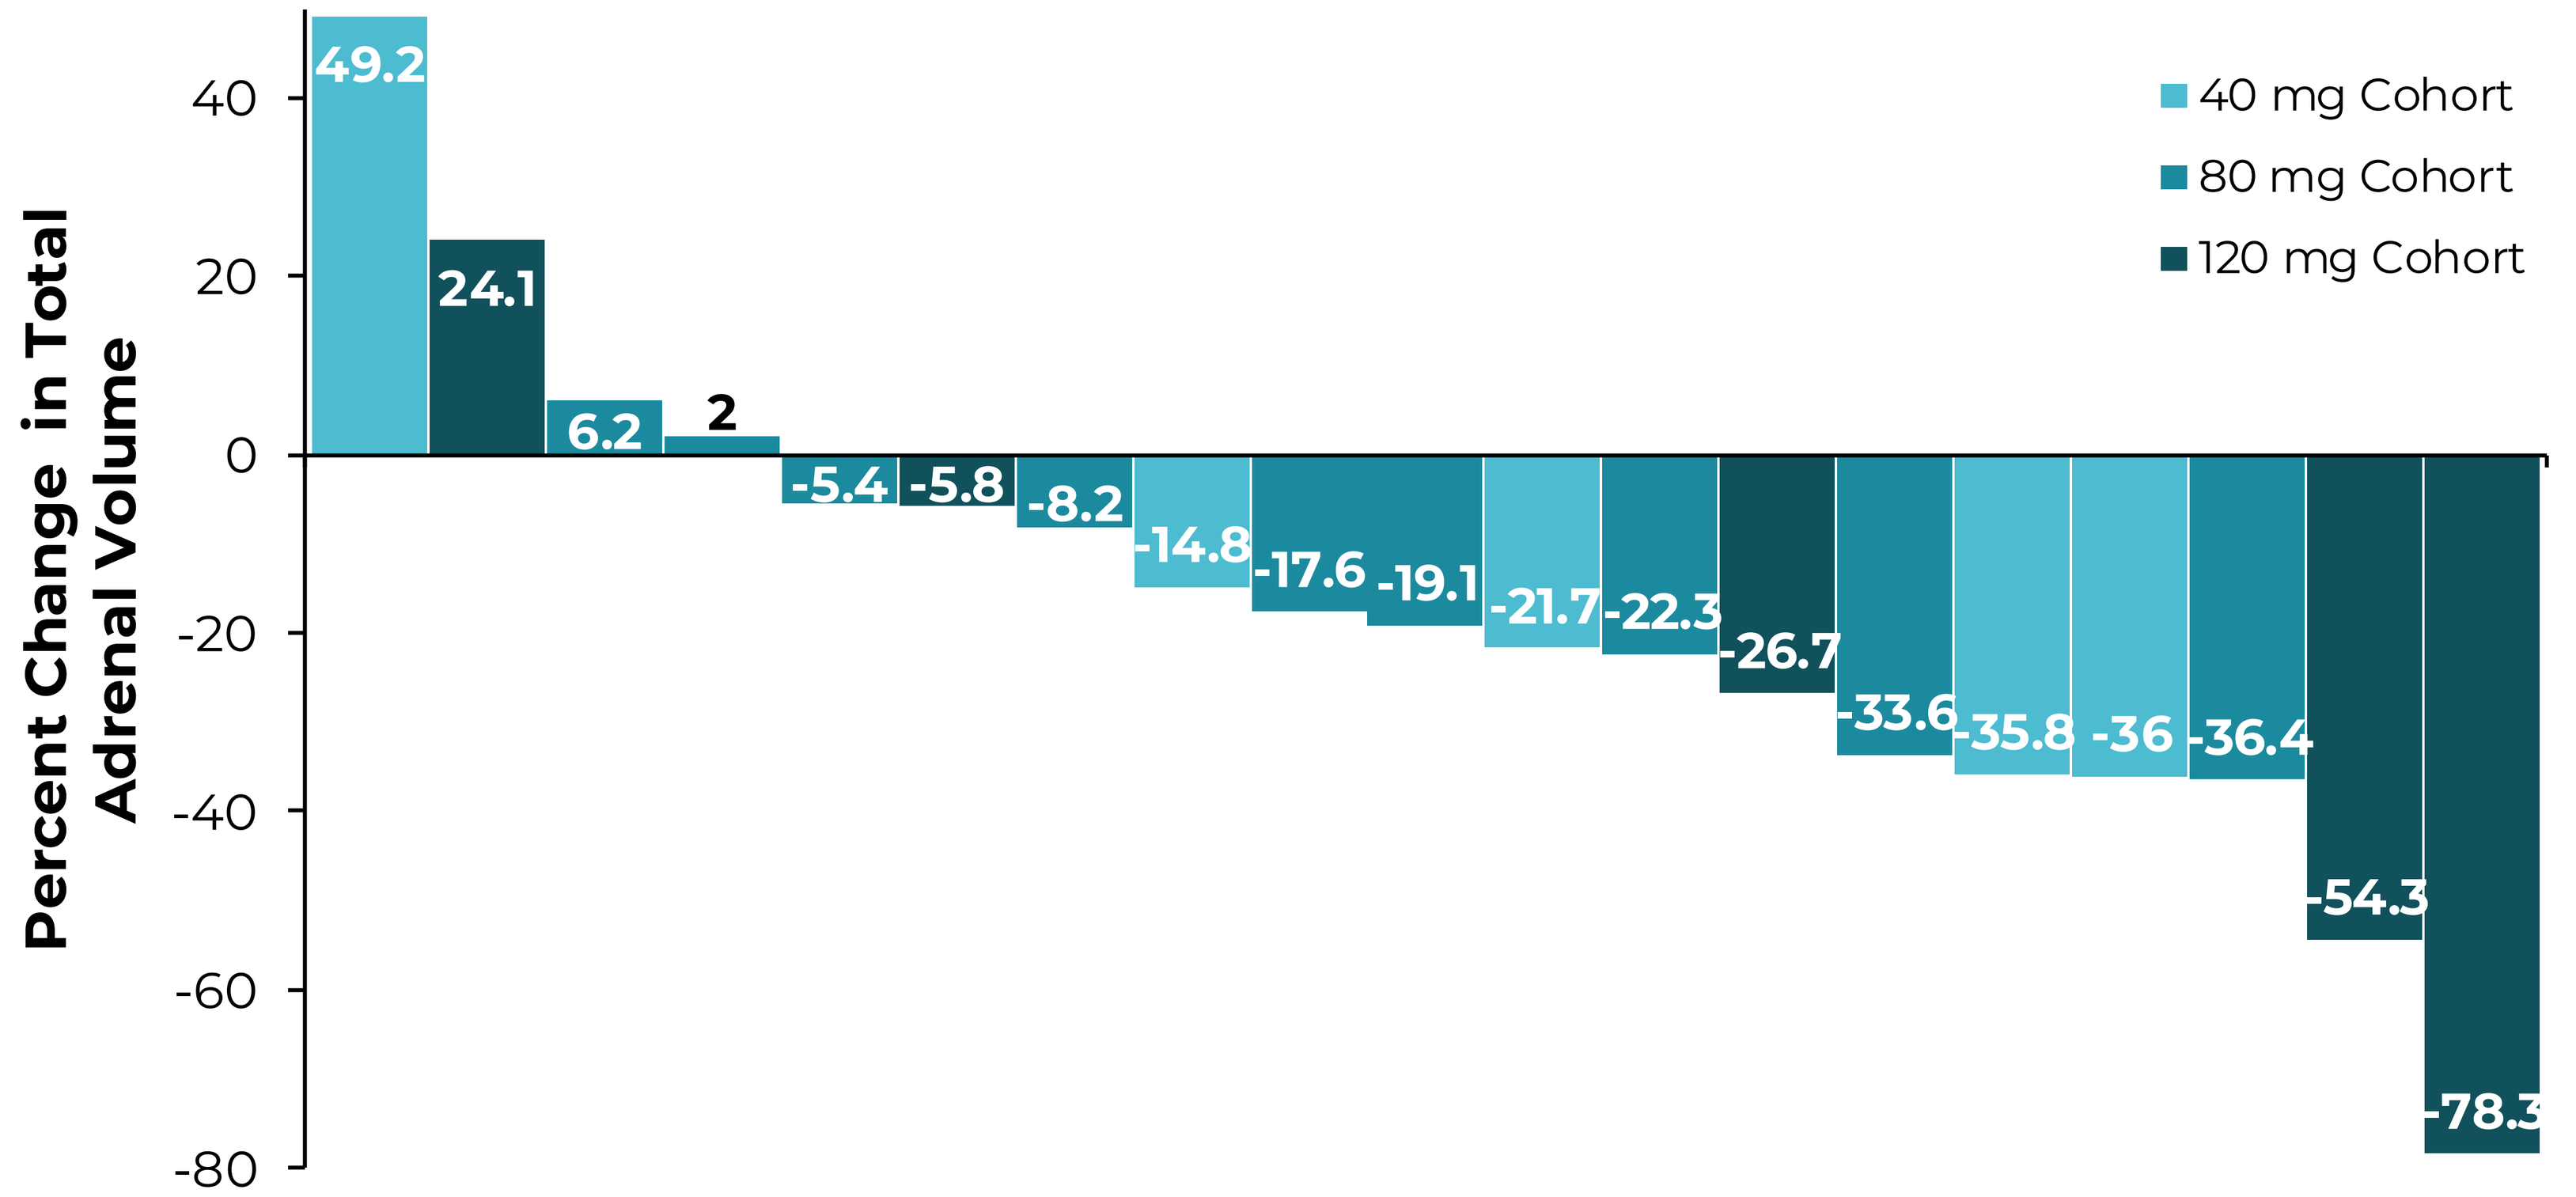
Percent Change in Total Adrenal Volume
40
20
0
-20
-40
-60
-80
49.2
24.1
6.2
2
-5.4
-5.8
-8.2
-14.8
-17.6
-19.1
-21.7
-22.3
-26.7
-33.6
-35.8
-36
-36.4
-54.3
-78.3
40 mg Cohort
80 mg Cohort
120 mg Cohort

[a]Four participants had increased total bilateral adrenal volume. Baseline adrenal volumes were underestimated due to inadequate field of view in two participants. One participant had suboptimal imaging for interpretation at baseline and at the end of the study. The participant with a 2% increase had optimal imaging quality; the increase was in the right adrenal only and was negligible.

- Consistent decrease in adrenal volume was seen across dose cohorts
- For all cohorts combined, median (range) change from baseline in total adrenal volume was -5.2 (-77.5 to 9.1) mL, or -19.1% (-78.3% to 49.2%)

Total Adrenal Volume Percent Reduction From Baseline to Week 12

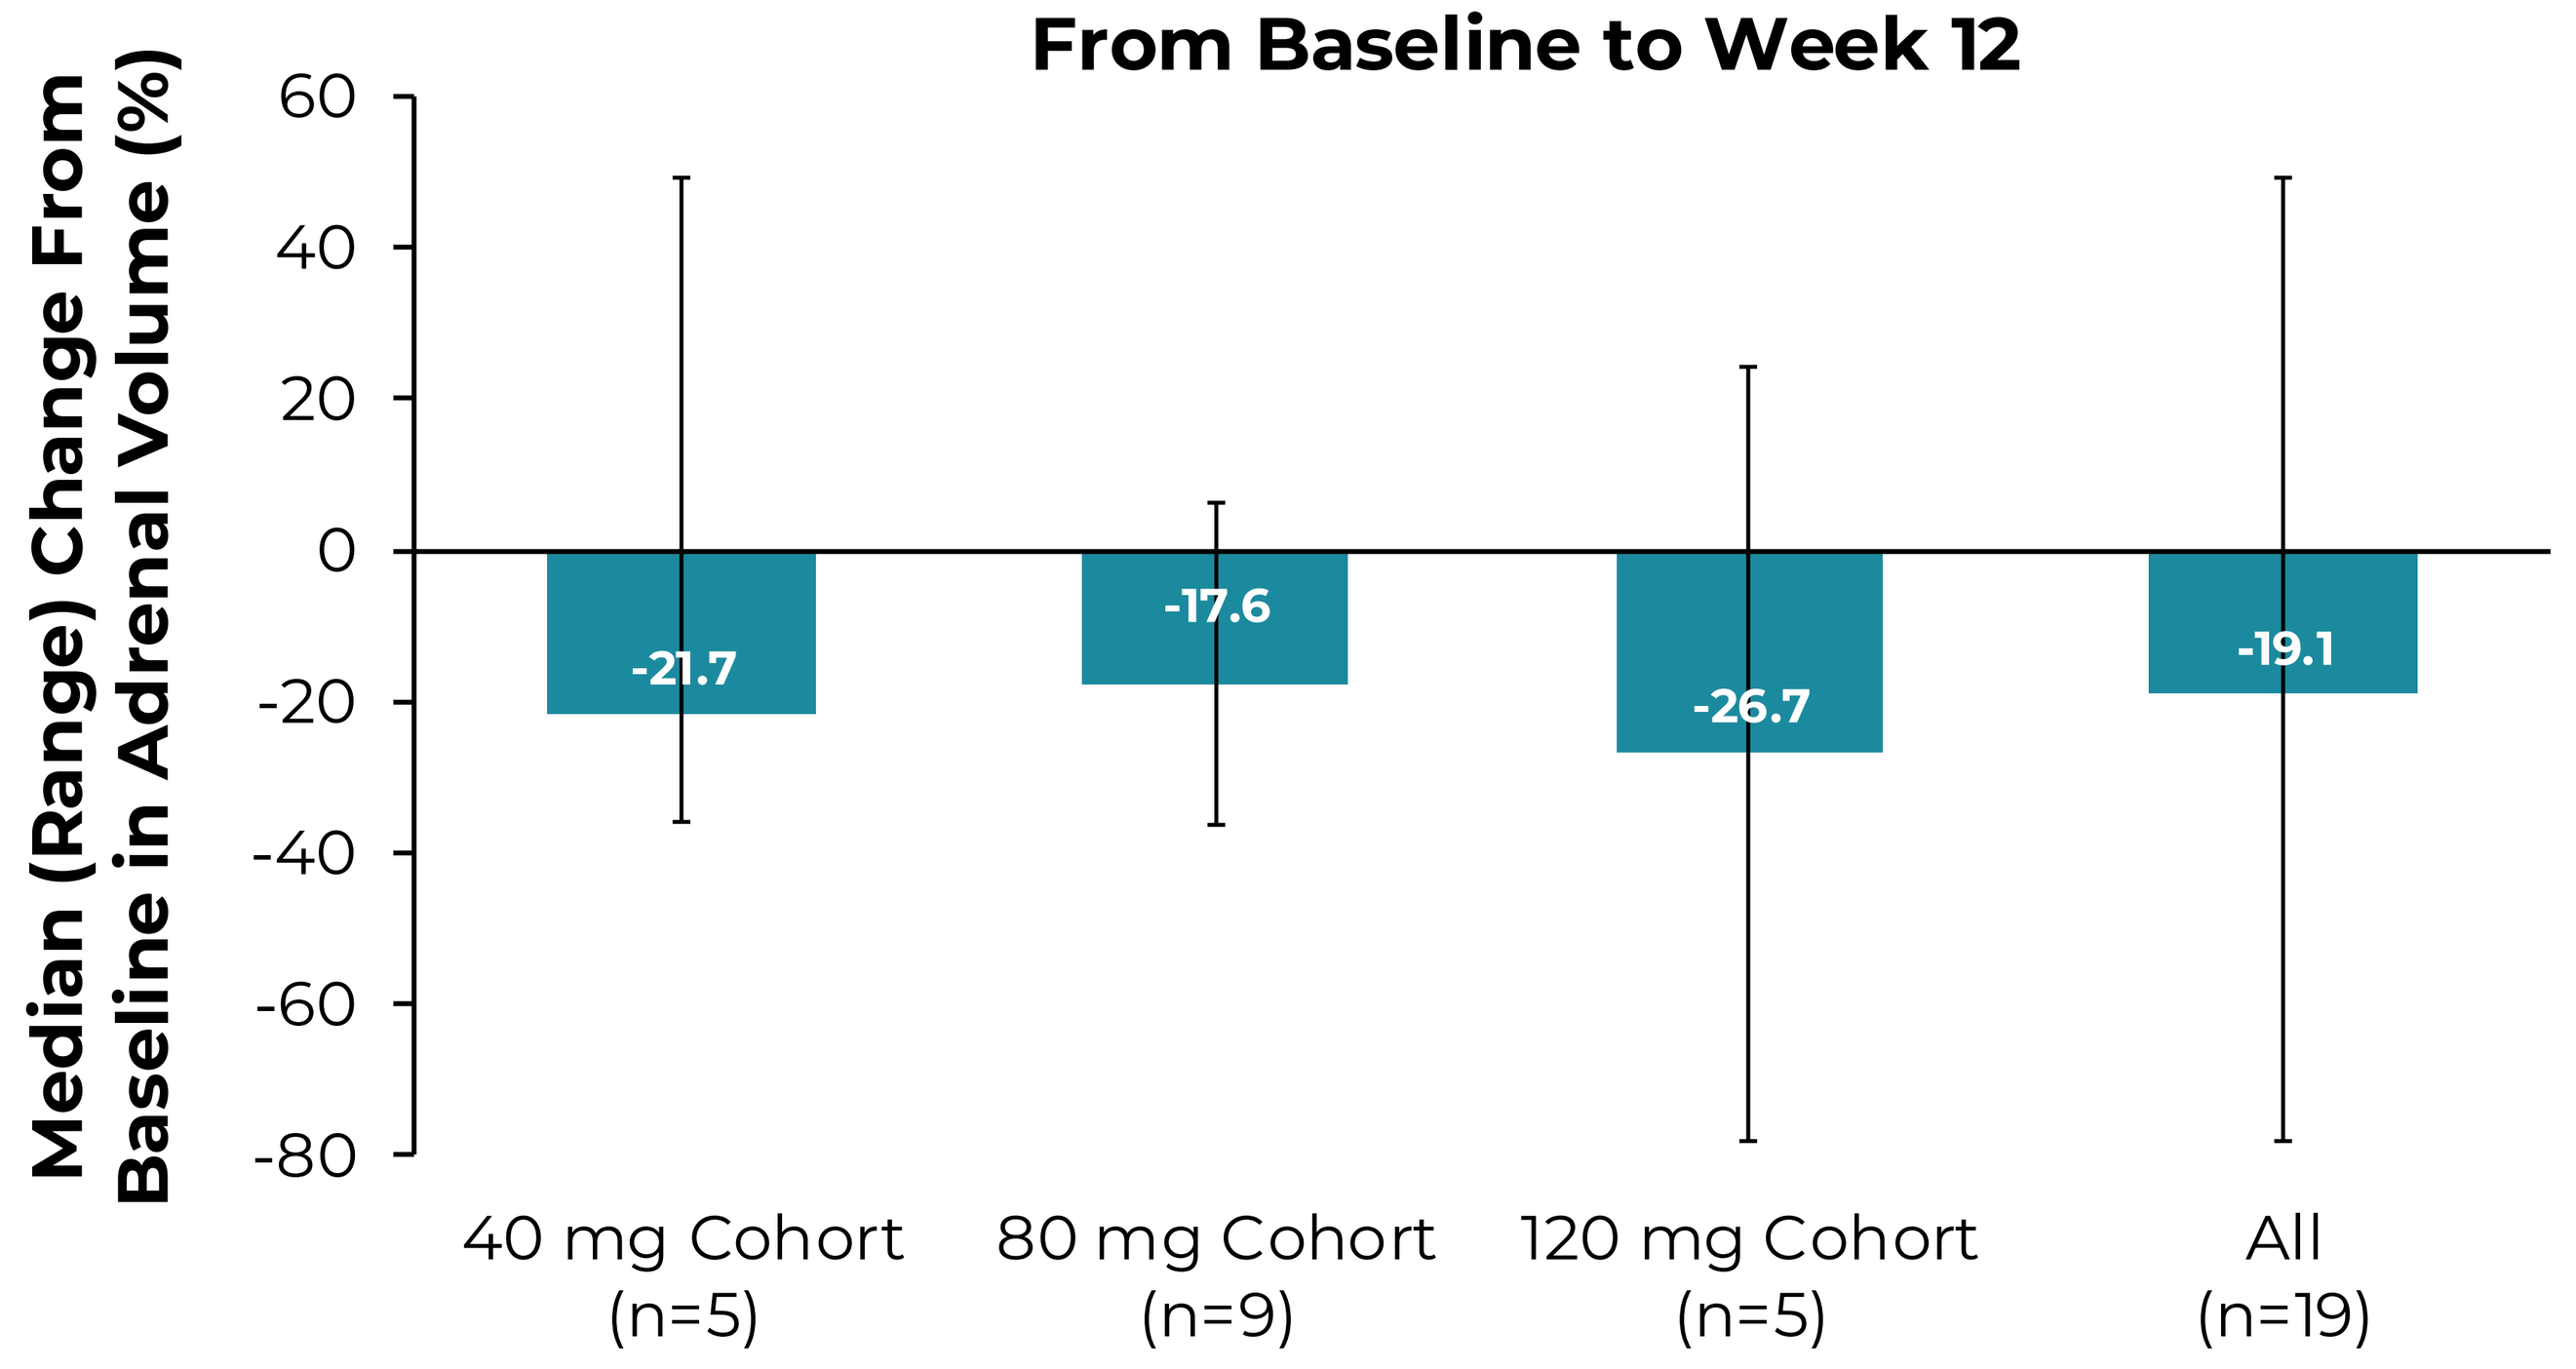
Median (Range) Change From Baseline in Adrenal Volume (%)
60
40
20
0
-20
-40
-60
-80
-21.7
-17.6
-26.7
-19.1
40 mg Cohort (n=5)
80 mg Cohort (n=9)
120 mg Cohort (n=5)
All (n=19)

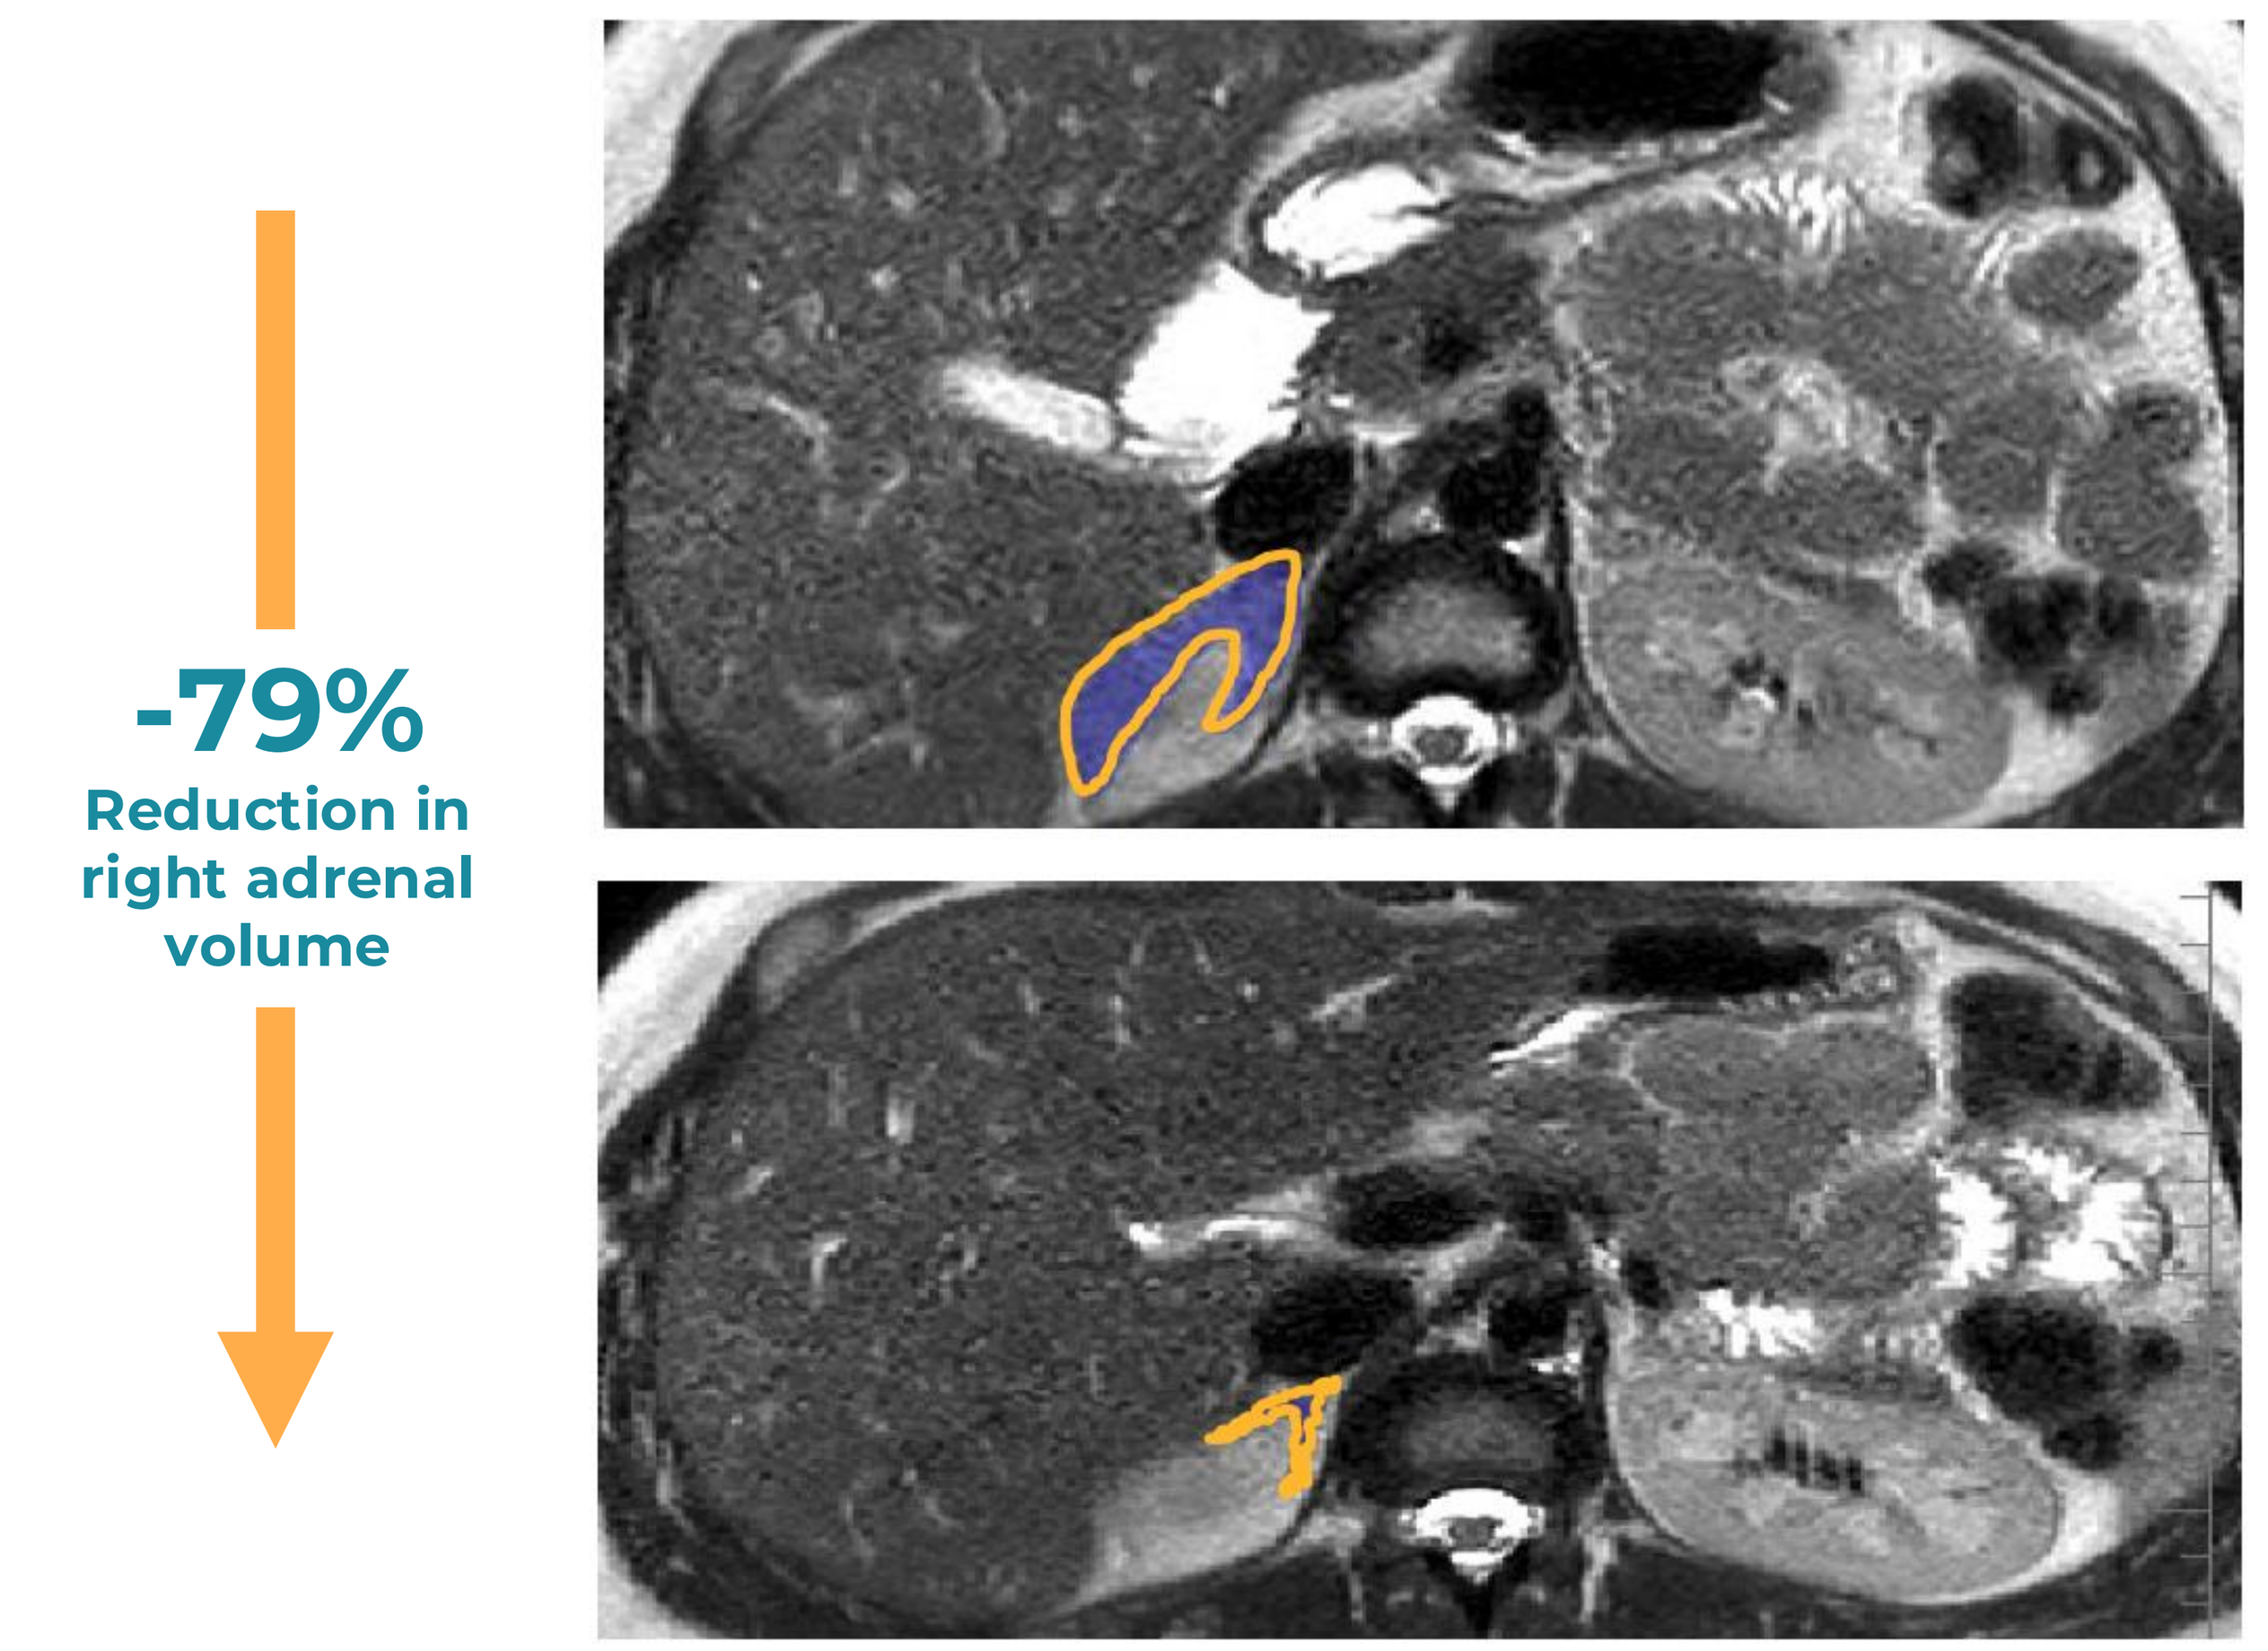
-79%
Reduction in right adrenal volume

- Right adrenal imaging from a male participant with CAH taking 30 mg of daily hydrocortisone whose serum A4 was 2025 ng/dL at baseline and 173.5 ng/dL after 12 weeks of 120 mg atumelnant
- Right adrenal volume was 11.7 mL at baseline. After 12 weeks of treatment with 120 mg atumelnant, right adrenal volume was reduced to 2.4 mL. Left adrenal volume findings were similar.

CONCLUSIONS

- 12 weeks of treatment with atumelnant, a first-in-class, once daily, oral, non-peptide MC2R antagonist, consistently reduced adrenal size in participants with CAH
- These results suggest the plasticity of adrenal tissue in adults with long-standing CAH and that ongoing adrenal hyperplasia is dependent on continued exposure to excess ACTH

- Primary, secondary, safety, and additional exploratory endpoints of the Phase 2 study oral session presentation: July 12, 2025, 1:45-3:15 pm (OR07-06)

REFERENCES

1. Yang M, White PC. *J Clin Endocrinol Metab*. 2025;110(Supplement_1):S1-S12.
2. Mallappa A, Merke DP. *Nat Rev Endocrinol*. 2022;18(6):337-352.
3. Kim TM, et al. *Endocrinol Metab*. 2022;37:124-137.
4. Auchus RJ, et al. *J Endocr Soc*. 2024;8 (Supplement_1):bvae163.249.

ACKNOWLEDGMENTS

Technical editorial and medical writing assistance were provided under the direction of the authors by Ryan Avenatti, PhD, Crinetics Pharmaceuticals, and Heather Caballes, PhD, CMPP, from Citrus Health Group (Chicago, Illinois), and were funded by Crinetics Pharmaceuticals, Inc. (San Diego, California). This study was funded by Crinetics Pharmaceuticals, Inc. (San Diego, CA).

For author affiliations and disclosures, please use the QR code.

Crinetics Pharmaceuticals

AUTHOR AFFILIATIONS

[1]Laboratorio de Hormonios e Genetica Molecular-LIM 42, da Faculdade de Medicina da Universidade de Sao Paulo, Sao Paulo, Brazil; [2]Department of Radiology and Radiological Sciences, Uniformed Services University of Health Sciences, Bethesda, MD, USA; [3]Hospital Misericordia, Córdoba, Argentina; [4]Crinetics Pharmaceuticals, Inc., San Diego, CA, USA; [5]Centro de Pesquisa Clínica, Instituto Estadual do Cérebro Paulo Niemeyer, Rio de Janeiro, RJ, Brazil; [6]Medizinische Klinik and Poliklinik IV, Klinikum der Universität München, LMU München, Munich, Germany; [7]Division of Metabolism, Endocrinology, and Diabetes and the Departments of Internal Medicine and Pharmacology, University of Michigan, Ann Arbor, MI, USA; [8]Endocrinology & Diabetes, University College London Hospitals NHS Foundation Trust, London

AUTHOR DISCLOSURES

TASS Bachega is a principal investigator for Crinetics Pharmaceuticals and Spruce Biosciences and has received consulting fees from Novo Nordisk.
J Marko and **D Bruera** have nothing to disclose.
A Ayala, **Y Wu**, **EDl Torre Ames**, and **A Krasner** are employees of Crinetics Pharmaceuticals and own stocks and shares in Crinetics Pharmaceuticals.
MR Gadelha has received speaker fees from Camarus, Ipsen, Novo Nordisk, and Recordati; and has attended advisory boards for Crinetics Pharmaceuticals, Novo Nordisk, and Recordati.
N Reisch has received consulting fees from Crinetics Pharmaceuticals, Diurnal Ltd, Lundbeck A/S, Neurocrine Biosciences, and Spruce Biosciences; and received conference travel support from Recordati Rare Diseases.
RJ Auchus contracted research support and consulting fees from Neurocrine Biosciences, Diurnal Ltd, Corcept Therapeutics, Recordati Rare Diseases, and Crinetics Pharmaceuticals; contracted research support from Adrenas Therapeutics and Spruce Biosciences; and received consulting fees from Quest Diagnostics, Xeris Pharmaceuticals, Novo Nordisk, H Lundbeck A/S, and Sparrow Pharmaceuticals.
U Srirangalingam received consulting fees from Crinetics Pharmaceuticals, Diurnal Ltd, and H Lundbeck A/S.