Rapid and Sustained Reduction of 11-Oxygenated Androgens in Adults With Classic Congenital Adrenal Hyperplasia Following Once-Daily Oral Atumelnant (CRN04894): Results From a 12-Week, Phase 2, Open-Label Study

SUN-438

Nicole Reisch, MD[1]; Richard J. Auchus, MD, PhD[2]; Umasuthan Srirangalingam, MD, PhD[3]; Tania A.S.S. Bachega, MD, PhD[4]; Brian Keevil MSc, FRCPath[5]; James M. Hawley MSc, FRCPath[5]; Alejandro Ayala, MD[6]; Yang Wu, PhD[6]; Eduardo De la Torre Ames, MD[5]; Alan Krasner, MD[6]; Lise Kjems, MD[6]; Deborah P. Merke, MS, MD[7]

BACKGROUND

- A hallmark of classic congenital adrenal hyperplasia (CAH) is 21-hydroxylase deficiency (21-OHD), which disrupts normal steroidogenesis pathways, resulting in decreased cortisol and aldosterone levels and excess adrenal androgens[1,2]
 - Traditional biomarkers of disease activity include 17-hydroxyprogesterone (17-OHP) and androstenedione (A4);[3] however, traditional androgen markers may not fully reflect the total androgen burden in CAH
 - The adrenals produce 11β-hydroxyandrostenedione (11-OHA4), which is metabolized to the androgen 11-ketotestosterone (11-KT); 11-oxygenated androgens contribute substantially to the total androgen burden in patients with CAH[4]
 - Assessment of 11-oxygenated androgens may aid in quantification of total androgen load and treatment response
- Atumelnant (CRN04894), a first-in-class, once-daily, oral, nonpeptide, melanocortin 2 receptor antagonist[5,6] that selectively inhibits adrenocorticotropic hormone (ACTH)-mediated adrenal steroidogenesis, is currently under development for the treatment of CAH and ACTH-dependent Cushing's syndrome
 - Rapid and sustained reductions with atumelnant, including change from baseline in A4 of -96% at Week 12 with 80 mg treatment, have been previously reported[7]

OBJECTIVE

- To present results from 2 exploratory endpoints of the ongoing Phase 2 study: change from baseline in morning serum 11-OHA4 and 11-KT over time

STUDY DESIGN AND METHODS

Key Eligibility Criteria

- Male or female participants age ≥18 to 75 years (≥16 years in USA)
- Classic CAH (21-OHD)
- On ≥15 mg hydrocortisone equivalent daily dose
- Serum A4 >1.5 × ULN

Cohorts[a]

- 40 mg once daily (n=11)
- 80 mg once daily (n=11)
- 120 mg once daily (n=6)

Study Endpoints

Primary: Change from baseline in serum A4

Secondary: Change from baseline in serum 17-OHP

Safety: Incidence of TEAEs

Exploratory: Change from baseline in serum steroid levels and PD biomarkers over time
- 11-OHA4
- 11-KT

Assessment Schedule

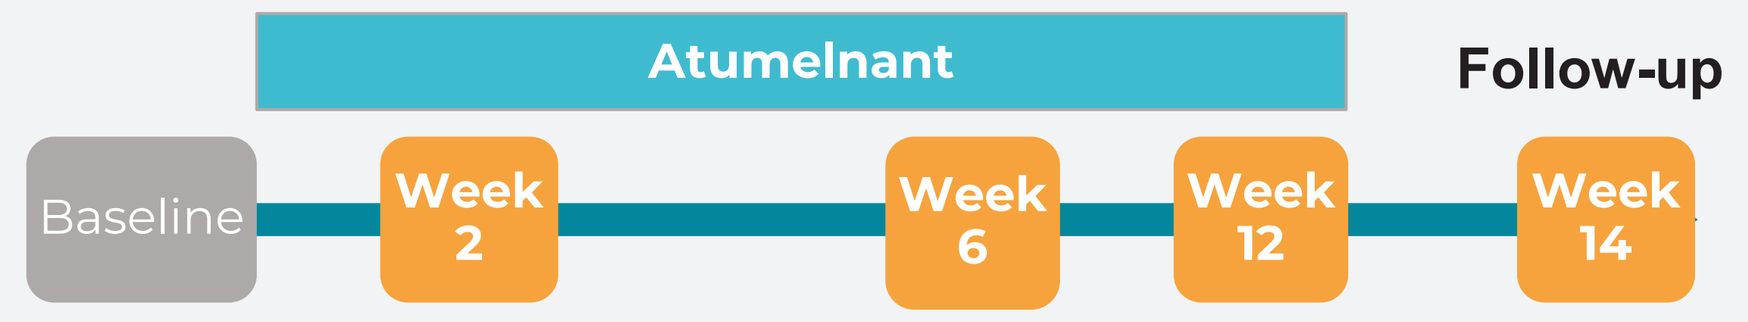

Atumelnant
Follow-up
Baseline
Week 2
Week 6
Week 12
Week 14

11-KT, 11-ketotestosterone; 11-OHA4, 11β-hydroxyandrostenedione; 17-OHP, 17-hydroxyprogesterone; 21-OHD, 21-hydroxylase deficiency; A4, androstenedione; CAH, congenital adrenal hyperplasia; PD, pharmacodynamics; TEAE, treatment-emergent adverse event; ULN, upper limit of normal.
[a]Participants received fixed doses of oral atumelnant administered nightly for 12 weeks.

RESULTS

- As of January 14, 2025, a total of 28 participants (40 mg, n=11; 80 mg, n=11; 120 mg, n=6) completed treatment
- Baseline 11-OHA4 and 11-KT levels in participants with CAH were considerably higher than previously published ranges of circulating levels in healthy individuals[2]

Participant Demographics and Baseline Characteristics

Parameters	Atumelnant 40 mg (n=11)	Atumelnant 80 mg (n=11)	Atumelnant 120 mg (n=6)	All participants (N=28)
Age, mean (range), y	28 (20-45)	33 (22-42)	34 (22-47)	31 (20-47)
Female, n (%)	4 (36)	8 (73)	3 (50)	15 (54)
Baseline biomarker levels, mean (range), ng/dL[a,b]				
11-OHA4	828 (142-2185)	1092 (171-3128)	1131 (350-1858)	997 (142-3128)
11-KT	298 (66-1292)	273 (54-680)	367 (141-547)	303 (54-1292)
GC dose, mean (range), mg/d[c]	30 (20-40)	31 (20-40)	23 (20-30)	29 (20-40)

11-KT, 11-ketotestosterone; 11-OHA4, 11β-hydroxyandrostenedione; GC, glucocorticoid.
[a]Reference range values for 11-OHA4 and 11-KT have not been established. Previous reports in healthy women suggest the following circulating levels: 11-OHA4, 121-272 ng/dL; 11-KT, 30 ng/dL.[2]
[b]The conversion factor from conventional units (ng/dL) to SI units (nmol/L) is 0.0331 for 11-OHA4 and 11-KT and 0.0347 for testosterone.
[c]GC dose in hydrocortisone equivalents.

RESULTS

Substantial Reduction in Pre-GC Serum 11-OHA4 Was Rapid and Sustained Through Week 12 in All Atumelnant Dose Cohorts

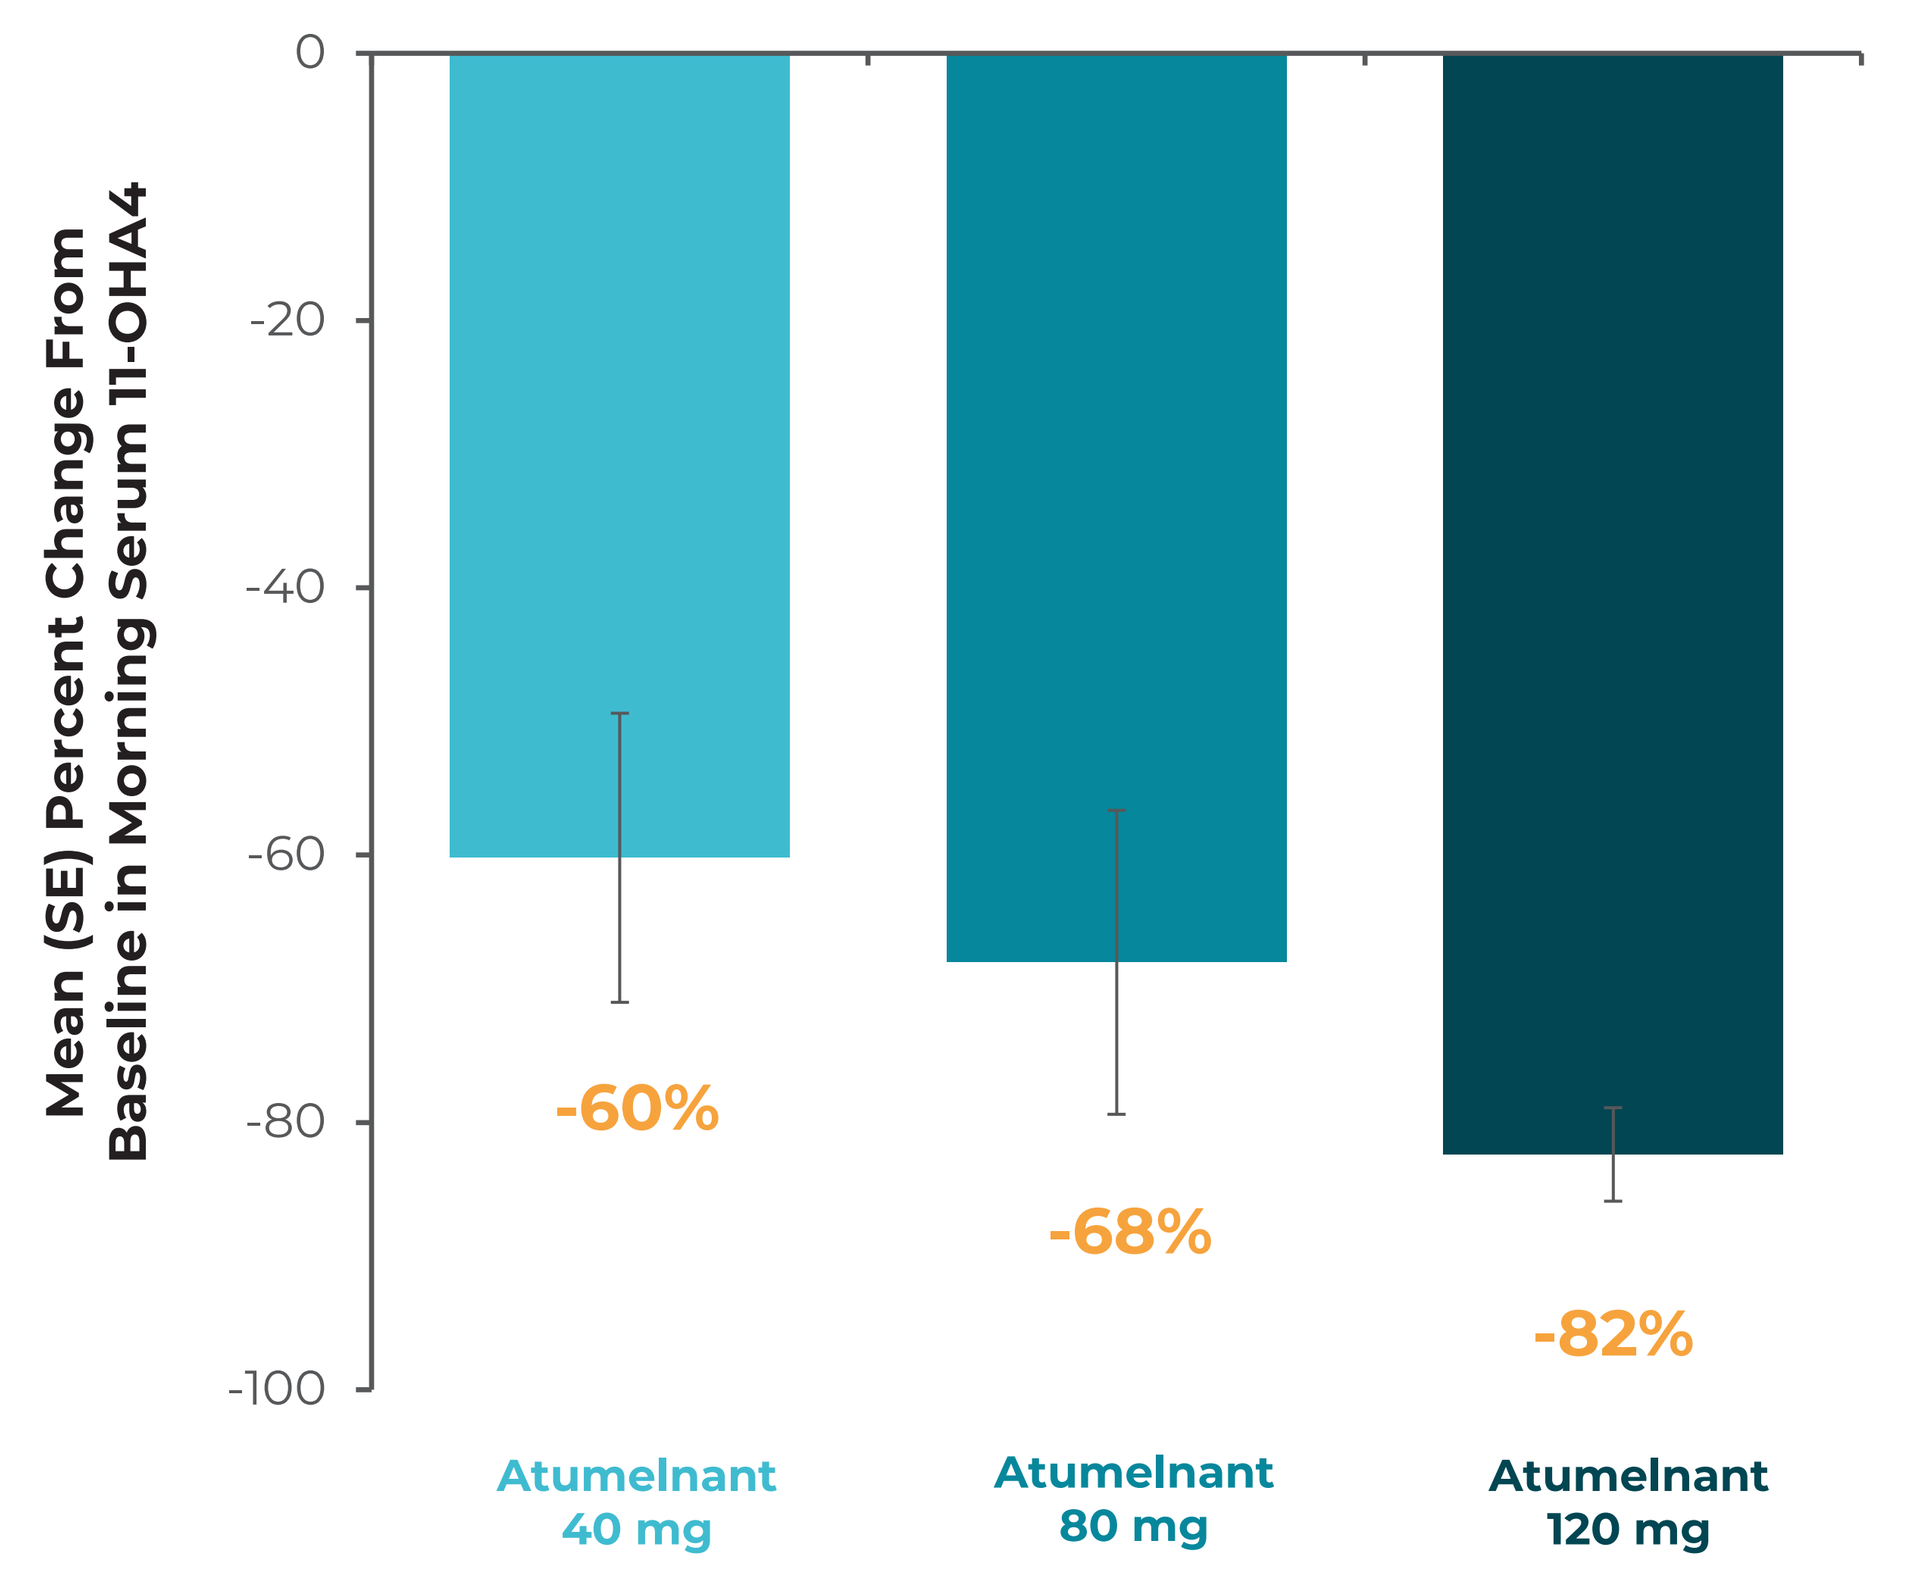

Mean (SE) Percent Change From Baseline in Morning Serum 11-OHA4
0
-20
-40
-60
-80
-100
-60%
-68%
-82%
Atumelnant 40 mg
Atumelnant 80 mg
Atumelnant 120 mg

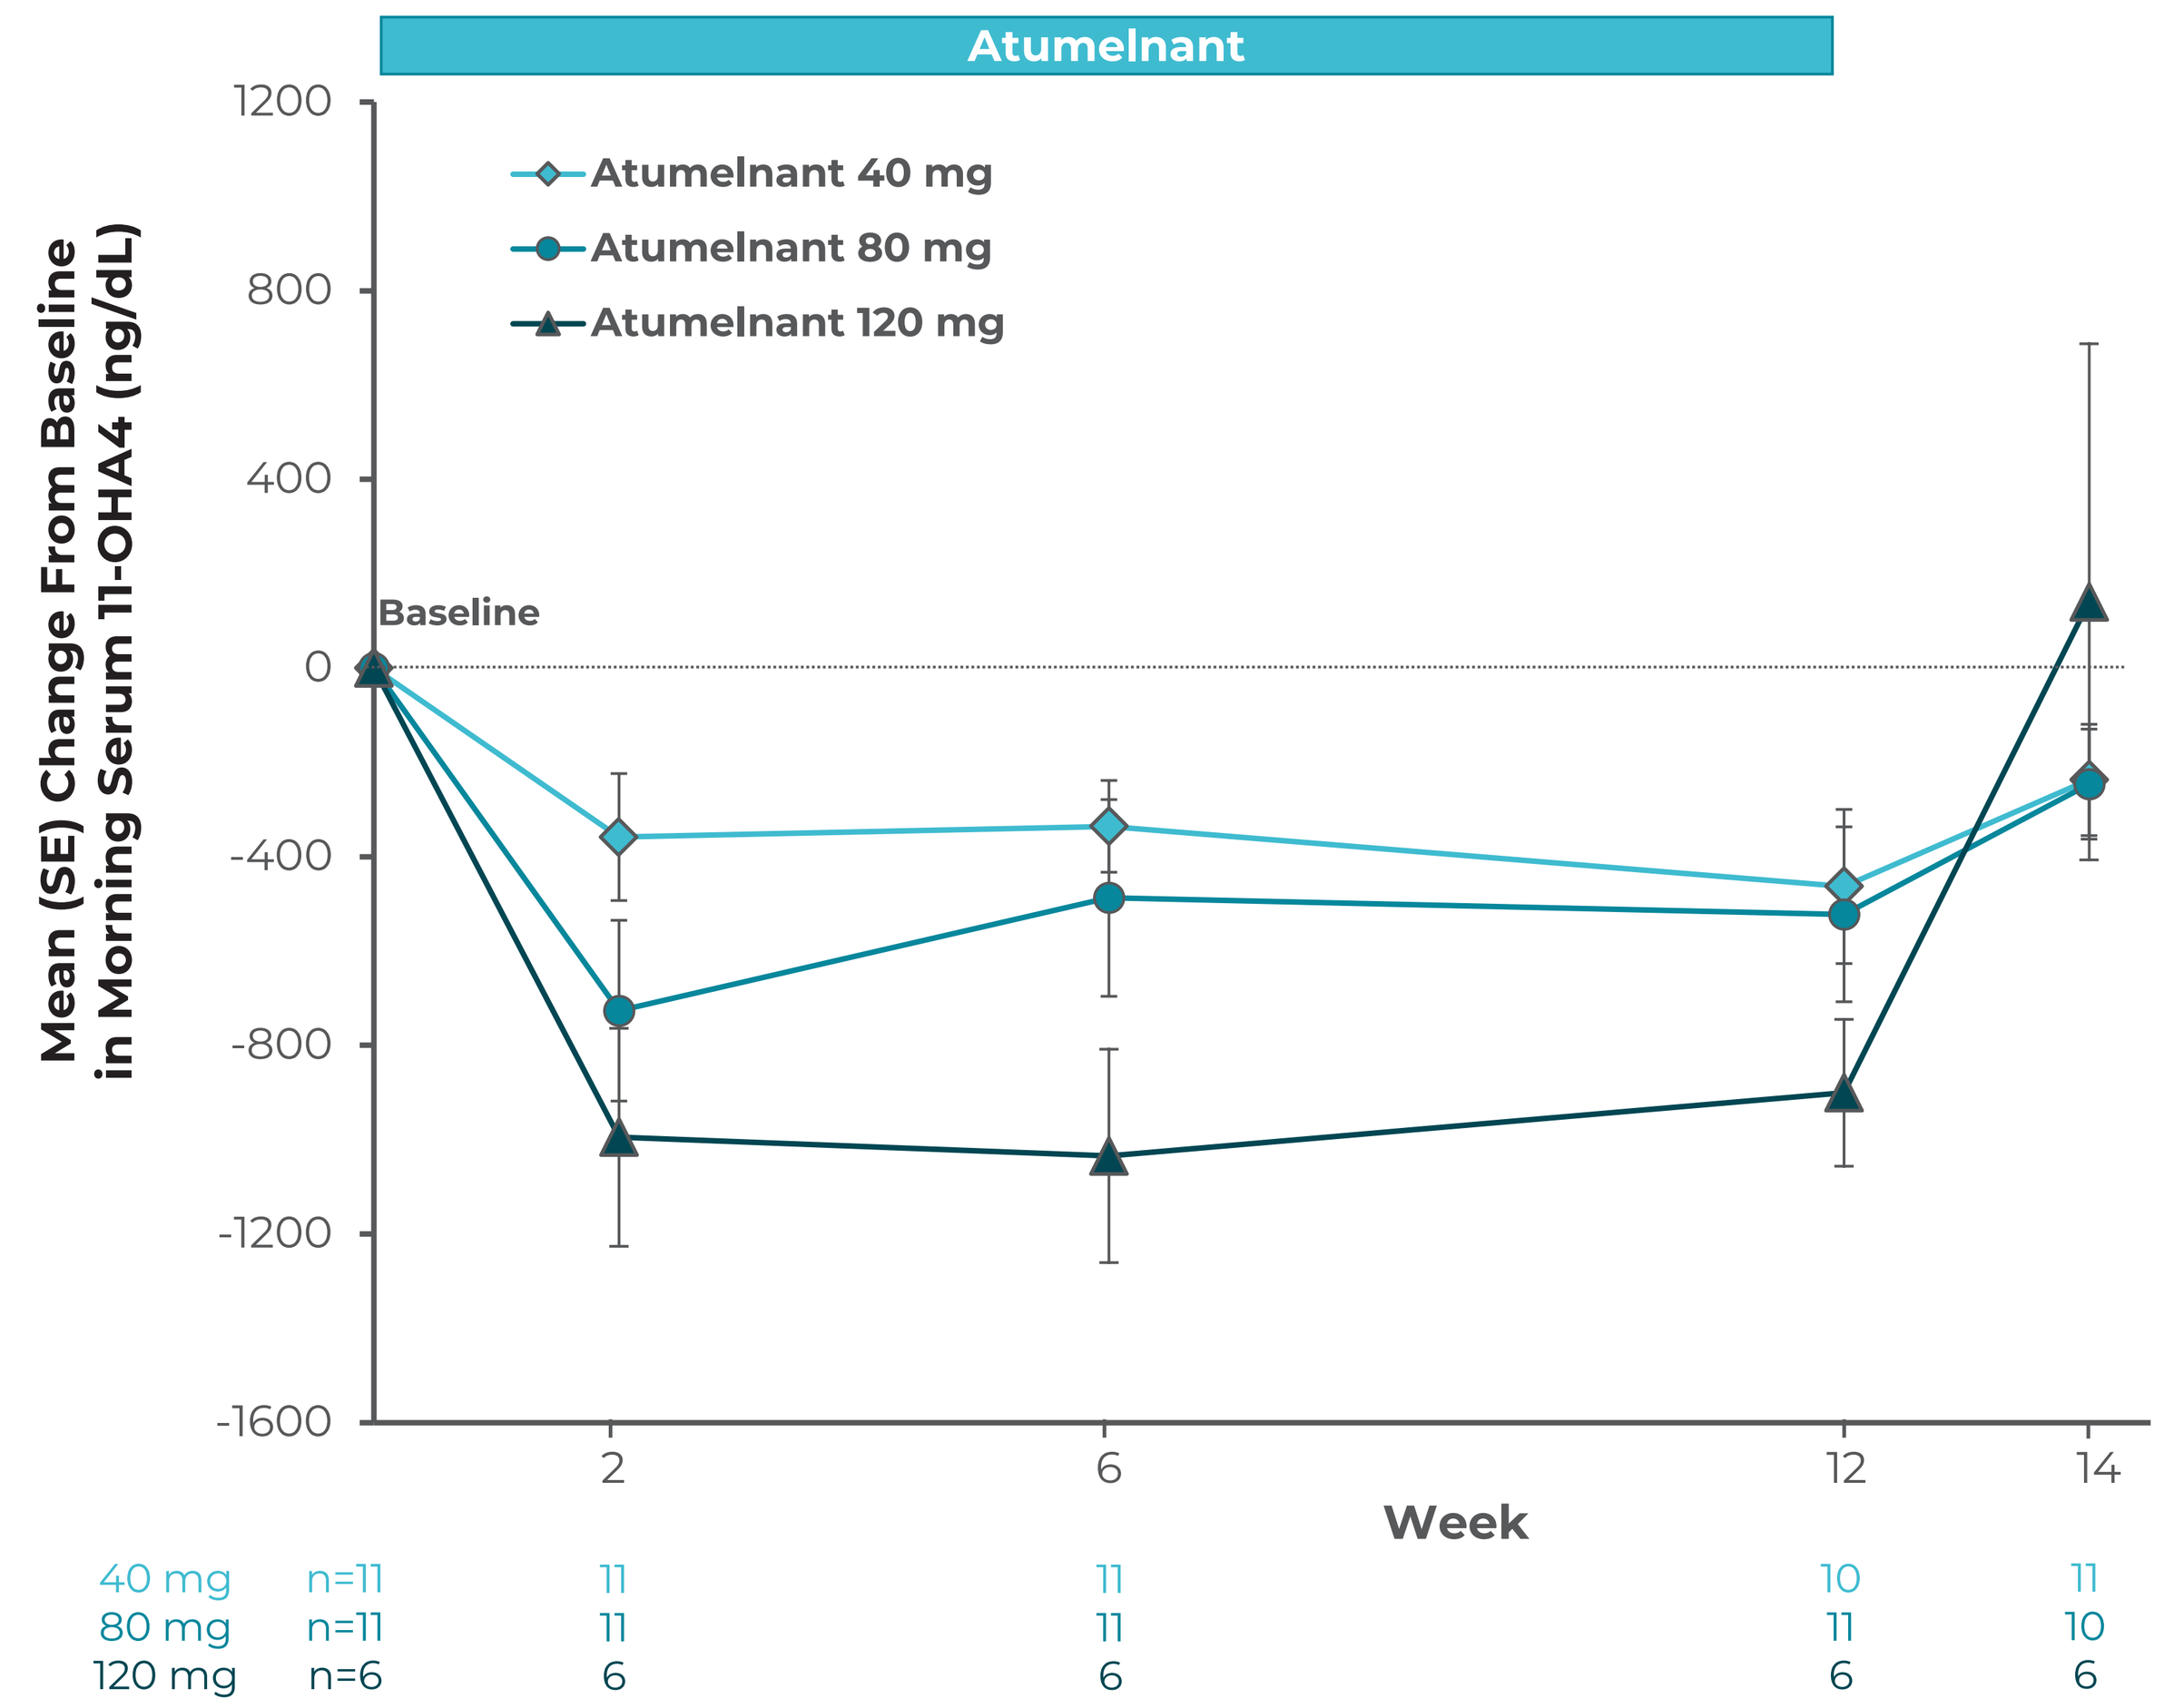

Atumelnant
Atumelnant 40 mg
Atumelnant 80 mg
Atumelnant 120 mg
Mean (SE) Change From Baseline in Morning Serum 11-OHA4 (ng/dL)
1200
800
400
0
-400
-800
-1200
-1600
Baseline
2
6
12
14
Week
40 mg n=11 11 11 10 11
80 mg n=11 11 11 11 10
120 mg n=6 6 6 6 6

11-OHA4, 11β-hydroxyandrostenedione; GC, glucocorticoid.

Substantial Reduction in Pre-GC Serum 11-KT Was Rapid and Sustained Through Week 12 in All Atumelnant Dose Cohorts

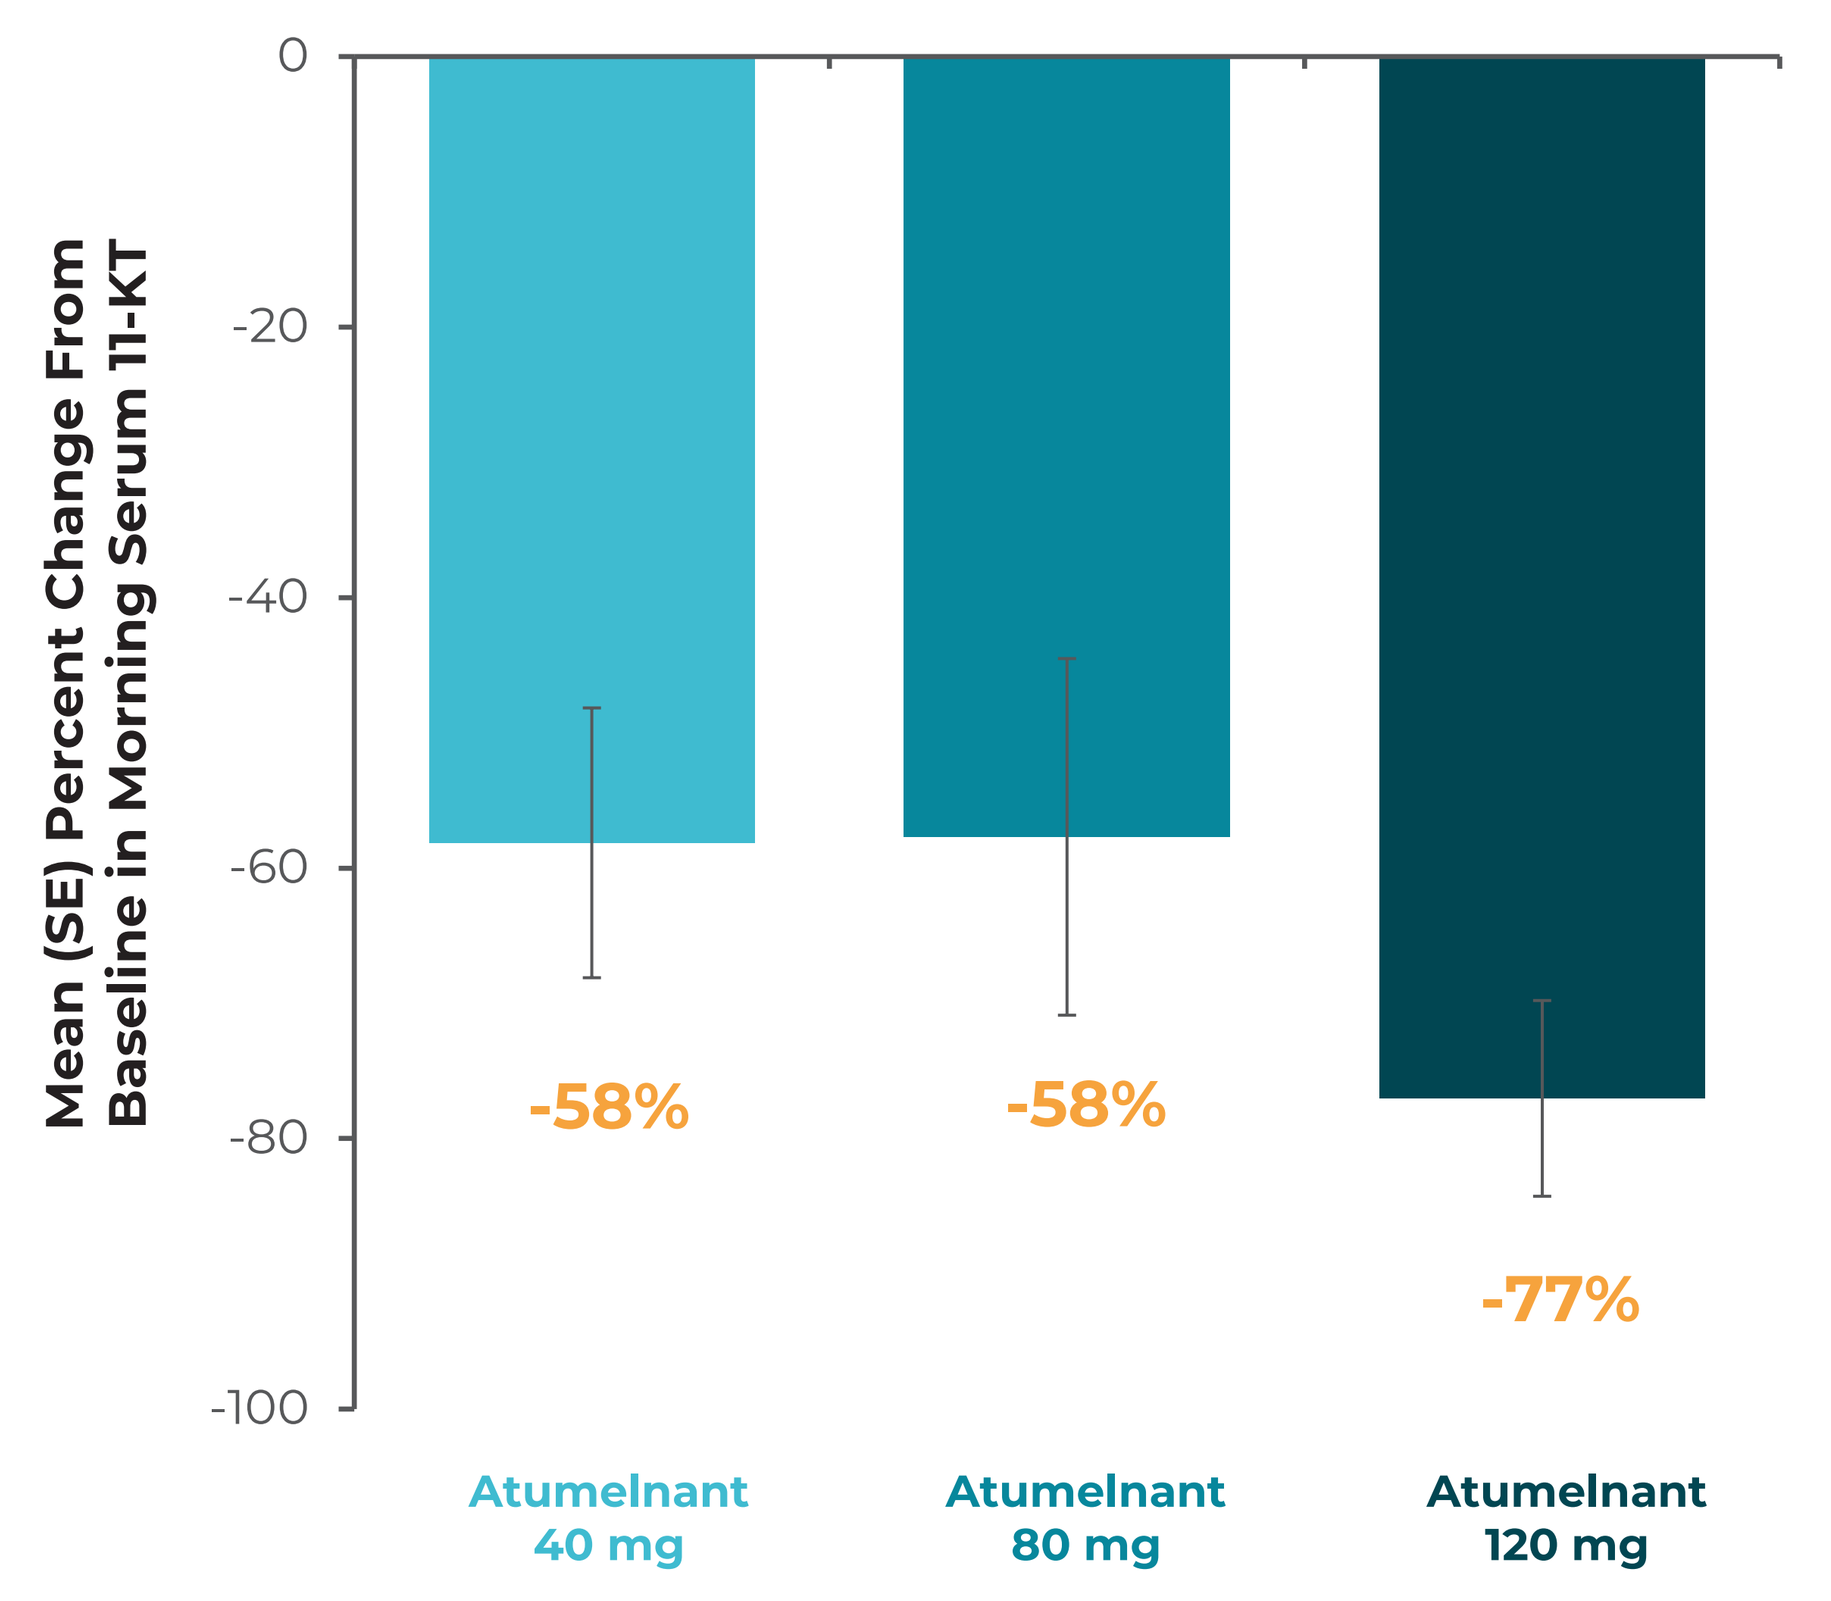

Mean (SE) Percent Change From Baseline in Morning Serum 11-KT
0
-20
-40
-60
-80
-100
-58%
-58%
-77%
Atumelnant 40 mg
Atumelnant 80 mg
Atumelnant 120 mg

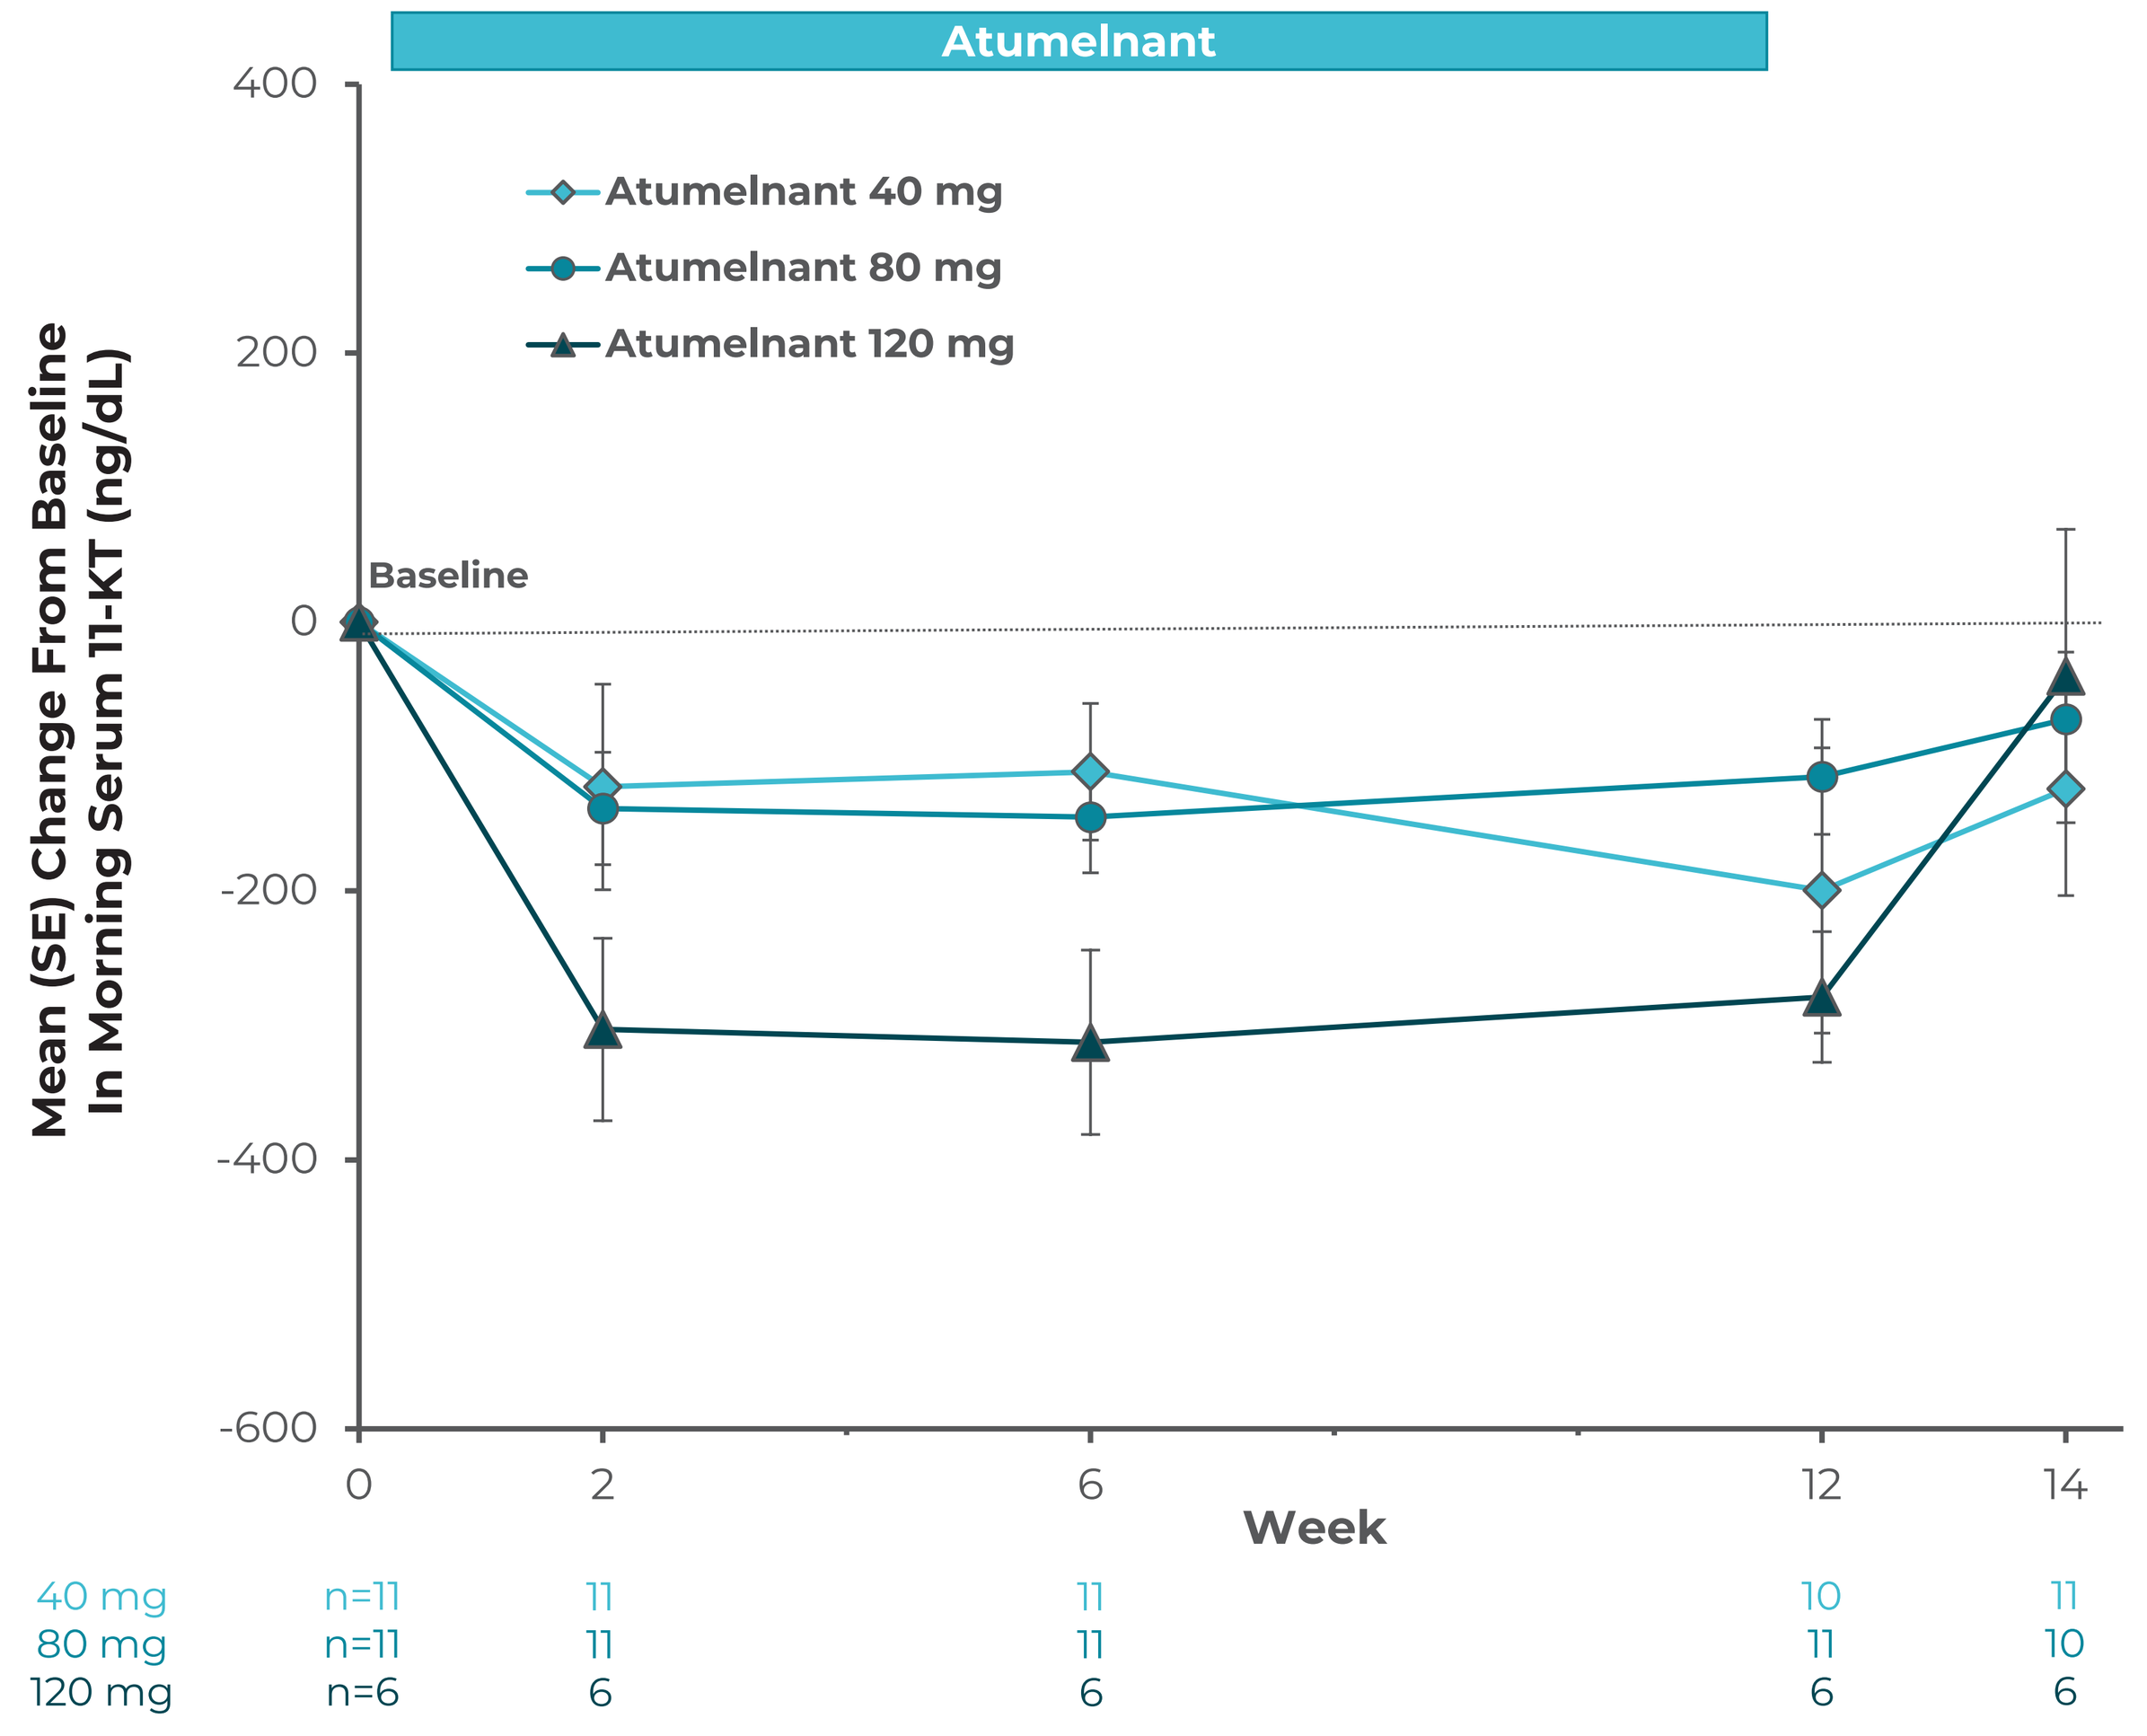

Atumelnant
Atumelnant 40 mg
Atumelnant 80 mg
Atumelnant 120 mg
Mean (SE) Change From Baseline In Morning Serum 11-KT (ng/dL)
400
200
0
-200
-400
-600
Baseline
0
2
6
12
14
Week
40 mg n=11 11 11 10 11
80 mg n=11 11 11 11 10
120 mg n=6 6 6 6 6

11-KT, 11-ketotestosterone; GC, glucocorticoid.

CONCLUSIONS

- Once-daily, oral atumelnant results in rapid and substantial reductions of 11-oxygenated androgens
- Atumelnant treatment for 12 weeks reduced 11-oxygenated androgen levels to circulating levels seen in healthy individuals[2]
- These results are in addition to the reductions in the traditional biomarkers A4 and 17-OHP in participants with classic CAH, with clinical activity observed across all doses[7]
- This reduction in total androgen burden may be linked to improvements in clinical outcomes previously reported within the 12-week time frame of this study, and lends support for assessing 11-oxygenated androgens to accurately quantify total androgen load and consequent treatment response in these patients

REFERENCES

1. Yang M, White PC. *J Clin Endocrinol Metab*. 2025;110(Supplement_1) :S1-S12.
2. Storbeck KH, O'Reilly MW. *Eur J Endocrinol*. 2023;188(4):R98-R109.
3. Turcu AF, et al. *Eur J Endocrinol*. 2016;174(5):601-609.
4. Turcu AF, et al. *J Clin Endocrinol Metab*. 2017;102(8):2701-2710.
5. Kim SH, et al. *ACS Med Chem Lett*. 2024;15(4):478-485.
6. Kusnetzow AK, et al. *J Endocr Soc*. 2020;8(Supplement_1):A133-A134.
7. Auchus RJ, et al. *J Endocr Soc*. 2024;8(Supplement_1):bvae163.249.

ACKNOWLEDGMENTS

Technical editorial and medical writing assistance were provided under the direction of the authors by Ryan Avenatti, PhD, Crinetics Pharmaceuticals, and Heather Caballes, PhD, CMPP, from Citrus Health Group (Chicago, Illinois), and were funded by Crinetics Pharmaceuticals, Inc. (San Diego, California). This study was funded by Crinetics Pharmaceuticals, Inc. (San Diego, CA). This work was supported in part by the Intramural Research Program of the National Institutes of Health (NIH).

For author affiliations and disclosures, please use the QR code.

Crinetics Pharmaceuticals

ENDO 2025 • July 12-15, 2025 • San Francisco, CA

AUTHOR AFFILIATIONS

[1]Medizinische Klinik and Poliklinik IV, LMU Klinikum München, Munich, Germany; [2]Division of Metabolism, Endocrinology, and Diabetes and the Departments of Internal Medicine and Pharmacology, University of Michigan, Ann Arbor, MI, USA; [3]Endocrinology & Diabetes, University College London Hospitals NHS Foundation Trust, London, UK; [4]Laboratorio de Hormonios e Genetica Molecular-LIM 42, da Faculdade de Medicina da Universidade de São Paulo, São Paulo, Brazil; [5]Department of Clinical Biochemistry, Wythenshawe Hospital, Manchester University Hospitals NHS Foundation Trust, Manchester, UK; [6]Crinetics Pharmaceuticals, Inc., San Diego, CA, USA; [7]The National Institutes of Health Clinical Center and Eunice Kennedy Shriver National Institute of Child Health and Human Development, Bethesda, MD, USA

AUTHOR DISCLOSURES

N Reisch has received consulting fees from Crinetics Pharmaceuticals, Diurnal Ltd, Lundbeck A/S, Neurocrine Biosciences, and Spruce Biosciences; and received conference travel support from Recordati Rare Diseases.
RJ Auchus contracted research support and consulting fees from Neurocrine Biosciences, Diurnal Ltd, Corcept Therapeutics, Recordati Rare Diseases, and Crinetics Pharmaceuticals; contracted research support from Adrenas Therapeutics and Spruce Biosciences; and received consulting fees from Quest Diagnostics, Xeris Pharmaceuticals, Novo Nordisk, H Lundbeck A/S, and Sparrow Pharmaceuticals.
U Srirangalingam received consulting fees from Crinetics Pharmaceuticals, Diurnal Ltd, and H Lundbeck A/S.
TASS Bachega is a principal investigator for Crinetics Pharmaceuticals and Spruce Biosciences and has received consulting fees from Novo Nordisk.
B Keevil and **JM Hawley** have nothing to disclose.
A Ayala, **Y Wu**, **EDl Torre Ames**, **A Krasner** and **L Kjems** are employees of Crinetics Pharmaceuticals and own stocks and shares in Crinetics Pharmaceuticals.
DP Merke has nothing to disclose.